Bimekizumab Reduced MRI Inflammatory Lesions in Patients with Axial Spondyloarthritis: Week 52 Results from the BE MOBILE 1 and 2 Phase 3 Studies

POS0246

Xenofon Baraliakos,[1] Victoria Navarro-Compán,[2] Denis Poddubnyy,[3] Maureen Dubreuil,[4] Alex Bennett,[5] Lennart Jans,[6] Ute Massow,[7] Carmen Fleurinck,[8] Thomas Vaux,[9] Alicia M. Ellis,[10] Natasha de Peyrecave,[8] Walter P. Maksymowych[11]

Objective

To evaluate the impact of bimekizumab on MRI inflammatory lesions of the sacroiliac joints and spine to Week 52 in two phase 3 studies.

Background

- Inflammatory lesions of the sacroiliac joints (SIJ) and spine are a key feature of axial spondyloarthritis (axSpA).[1,2]
- Bimekizumab (BKZ), a monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A, has demonstrated consistent and sustained efficacy up to Week 52 across the full disease spectrum of axSpA (non-radiographic axSpA [nr-axSpA] and radiographic axSpA [r-axSpA, i.e. ankylosing spondylitis]).[3–5]

Methods

- BE MOBILE 1 (NCT03928704) and BE MOBILE 2 (NCT03928743) study designs have been reported previously; both comprised a 16-week double-blind placebo (PBO)-controlled period followed by a 36-week maintenance period.[5] From Week 16, all patients received subcutaneous BKZ 160 mg every 4 weeks (Q4W) to Week 52.
- MRIs were performed in the subset of patients in the MRI sub-studies and assessed via central reading by two independent expert readers (scores averaged), with a third adjudicator in cases of disagreement.
- MRI endpoints (Spondyloarthritis Research Consortium of Canada [SPARCC] SIJ and ASspiMRI-a Berlin modification ['Berlin spine'] inflammation scores) and the proportion of patients achieving MRI remission (SPARCC SIJ <2; Berlin spine ≤2)[6] are reported at baseline, Weeks 16 and 52.

Results

Patients

- At baseline, 59.8% of patients with nr-axSpA (152/254) and 41.6% of patients with r-axSpA (138/332) had SPARCC SIJ assessments as part of the MRI sub-studies; 57.4% (146/254) and 41.3% (137/332) of patients had Berlin spine assessments.
- Baseline characteristics in the MRI sub-studies were largely comparable between treatment arms and reflected those of the overall patient population (**Table 1**).

SPARCC SIJ and Berlin Spine Scores

- Improvements in mean MRI scores observed at Week 16 were maintained to Week 52 for patients initially randomised to BKZ. Patients who switched from PBO to BKZ at Week 16 reached similar levels of improvement to BKZ-randomised patients at Week 52 (**Figure 1**).
- Similar patterns of improvement were observed for change from baseline (CfB) in SPARCC SIJ and Berlin spine scores, with patients with higher baseline inflammation scores showing the greatest improvements with BKZ (**Figure 2**).

Achievement of MRI Remission

- Among patients with SIJ inflammation at baseline, a greater proportion of BKZ- vs PBO-randomised patients achieved SPARCC SIJ remission at Week 16. A similar pattern was seen for patients with spine inflammation at baseline and Berlin spine remission at Week 16 (**Figure 3**).
- The proportion of patients achieving MRI remission largely increased to Week 52 (**Figure 3**).

Conclusions

Across the full disease spectrum of axSpA, dual inhibition of IL-17A and IL-17F with bimekizumab resulted in a reduction in MRI inflammatory lesions of the SIJ and spine, with greater improvements observed for patients with higher levels of baseline inflammation, and large proportions of patients achieving MRI remission at Week 52.

Summary

Axial spondyloarthritis is characterised by MRI inflammatory lesions of the sacroiliac joints (SIJ) and spine

To Week 52, bimekizumab treatment led to improvements in inflammation scores, with patients with higher baseline inflammation scores showing greatest improvements

Continuous bimekizumab treatment led to MRI remission at Week 52 in:

76–80% of patients for MRI SPARCC SIJ

40–77% of patients for MRI Berlin spine

Table 1 Baseline characteristics for MRI sub-study and overall patient populations

n (%), unless stated otherwise	nr-axSpA (BE MOBILE 1) PBO n=70[a]	nr-axSpA (BE MOBILE 1) BKZ 160 mg Q4W n=82[a]	nr-axSpA (BE MOBILE 1) Overall study population N=254[b]	r-axSpA (BE MOBILE 2) PBO n=48[a]	r-axSpA (BE MOBILE 2) BKZ 160 mg Q4W n=91[a]	r-axSpA (BE MOBILE 2) Overall study population N=332[b]
Age, years, mean (SD)	40.0 (12.5)	38.9 (11.6)	39.4 (11.5)	39.7 (12.9)	40.1 (12.2)	40.4 (12.3)
Male	31 (44.3)	50 (61.0)	138 (54.3)	31 (64.6)	68 (74.7)	240 (72.3)
HLA-B27 positive	47 (67.1)	65 (79.3)	197 (77.6)	39 (81.3)	80 (87.9)	284 (85.5)
Symptom duration, years, mean (SD)	8.7 (9.2)	9.0 (8.5)	9.0 (8.8)	12.7 (9.0)	13.7 (11.0)	13.5 (10.3)
ASDAS, mean (SD)	3.6 (0.7)	3.6 (0.7)	3.7 (0.7)	3.6 (0.8)	3.9 (0.8)	3.7 (0.8)[c]
BASDAI, mean (SD)	6.4 (1.3)	6.9 (1.2)	6.8 (1.3)	6.4 (1.4)	6.6 (1.4)	6.5 (1.3)
hs-CRP, mg/L, geometric mean (geometric CV, %)	4.9 (207.5)	3.8 (276.1)	4.8 (261.8)	6.1 (195.1)	7.6 (244.7)	6.6 (246.3)
Prior TNFi exposure	8 (11.4)	5 (6.1)	27 (10.6)	7 (14.6)	13 (14.3)	54 (16.3)
MRI SPARCC SIJ ≥2[d]	46 (65.7)	50 (61.0)	–	21 (43.8)	42 (46.7)	–
MRI Berlin spine >2[e]	13 (19.4)	17 (21.5)	–	20 (41.7)	36 (40.4)	–

Randomised set. SPARCC SIJ and Berlin spine scores range from 0–72 and 0–69, respectively, with lower scores indicating less inflammation; [a]Includes only patients in the MRI sub-study; [b]All patients in the overall study were pooled, regardless of treatment arm; [c]n=331; [d]In patients with SPARCC SIJ assessments: nr-axSpA: BKZ: n=82, PBO: n=70; r-axSpA: BKZ: n=90, PBO: n=48; [e]In patients with Berlin spine assessments: nr-axSpA: BKZ: n=79; PBO: n=67; r-axSpA: BKZ: n=89; PBO: n=48.

Figure 1 Mean MRI inflammation scores to Week 52 (OC)

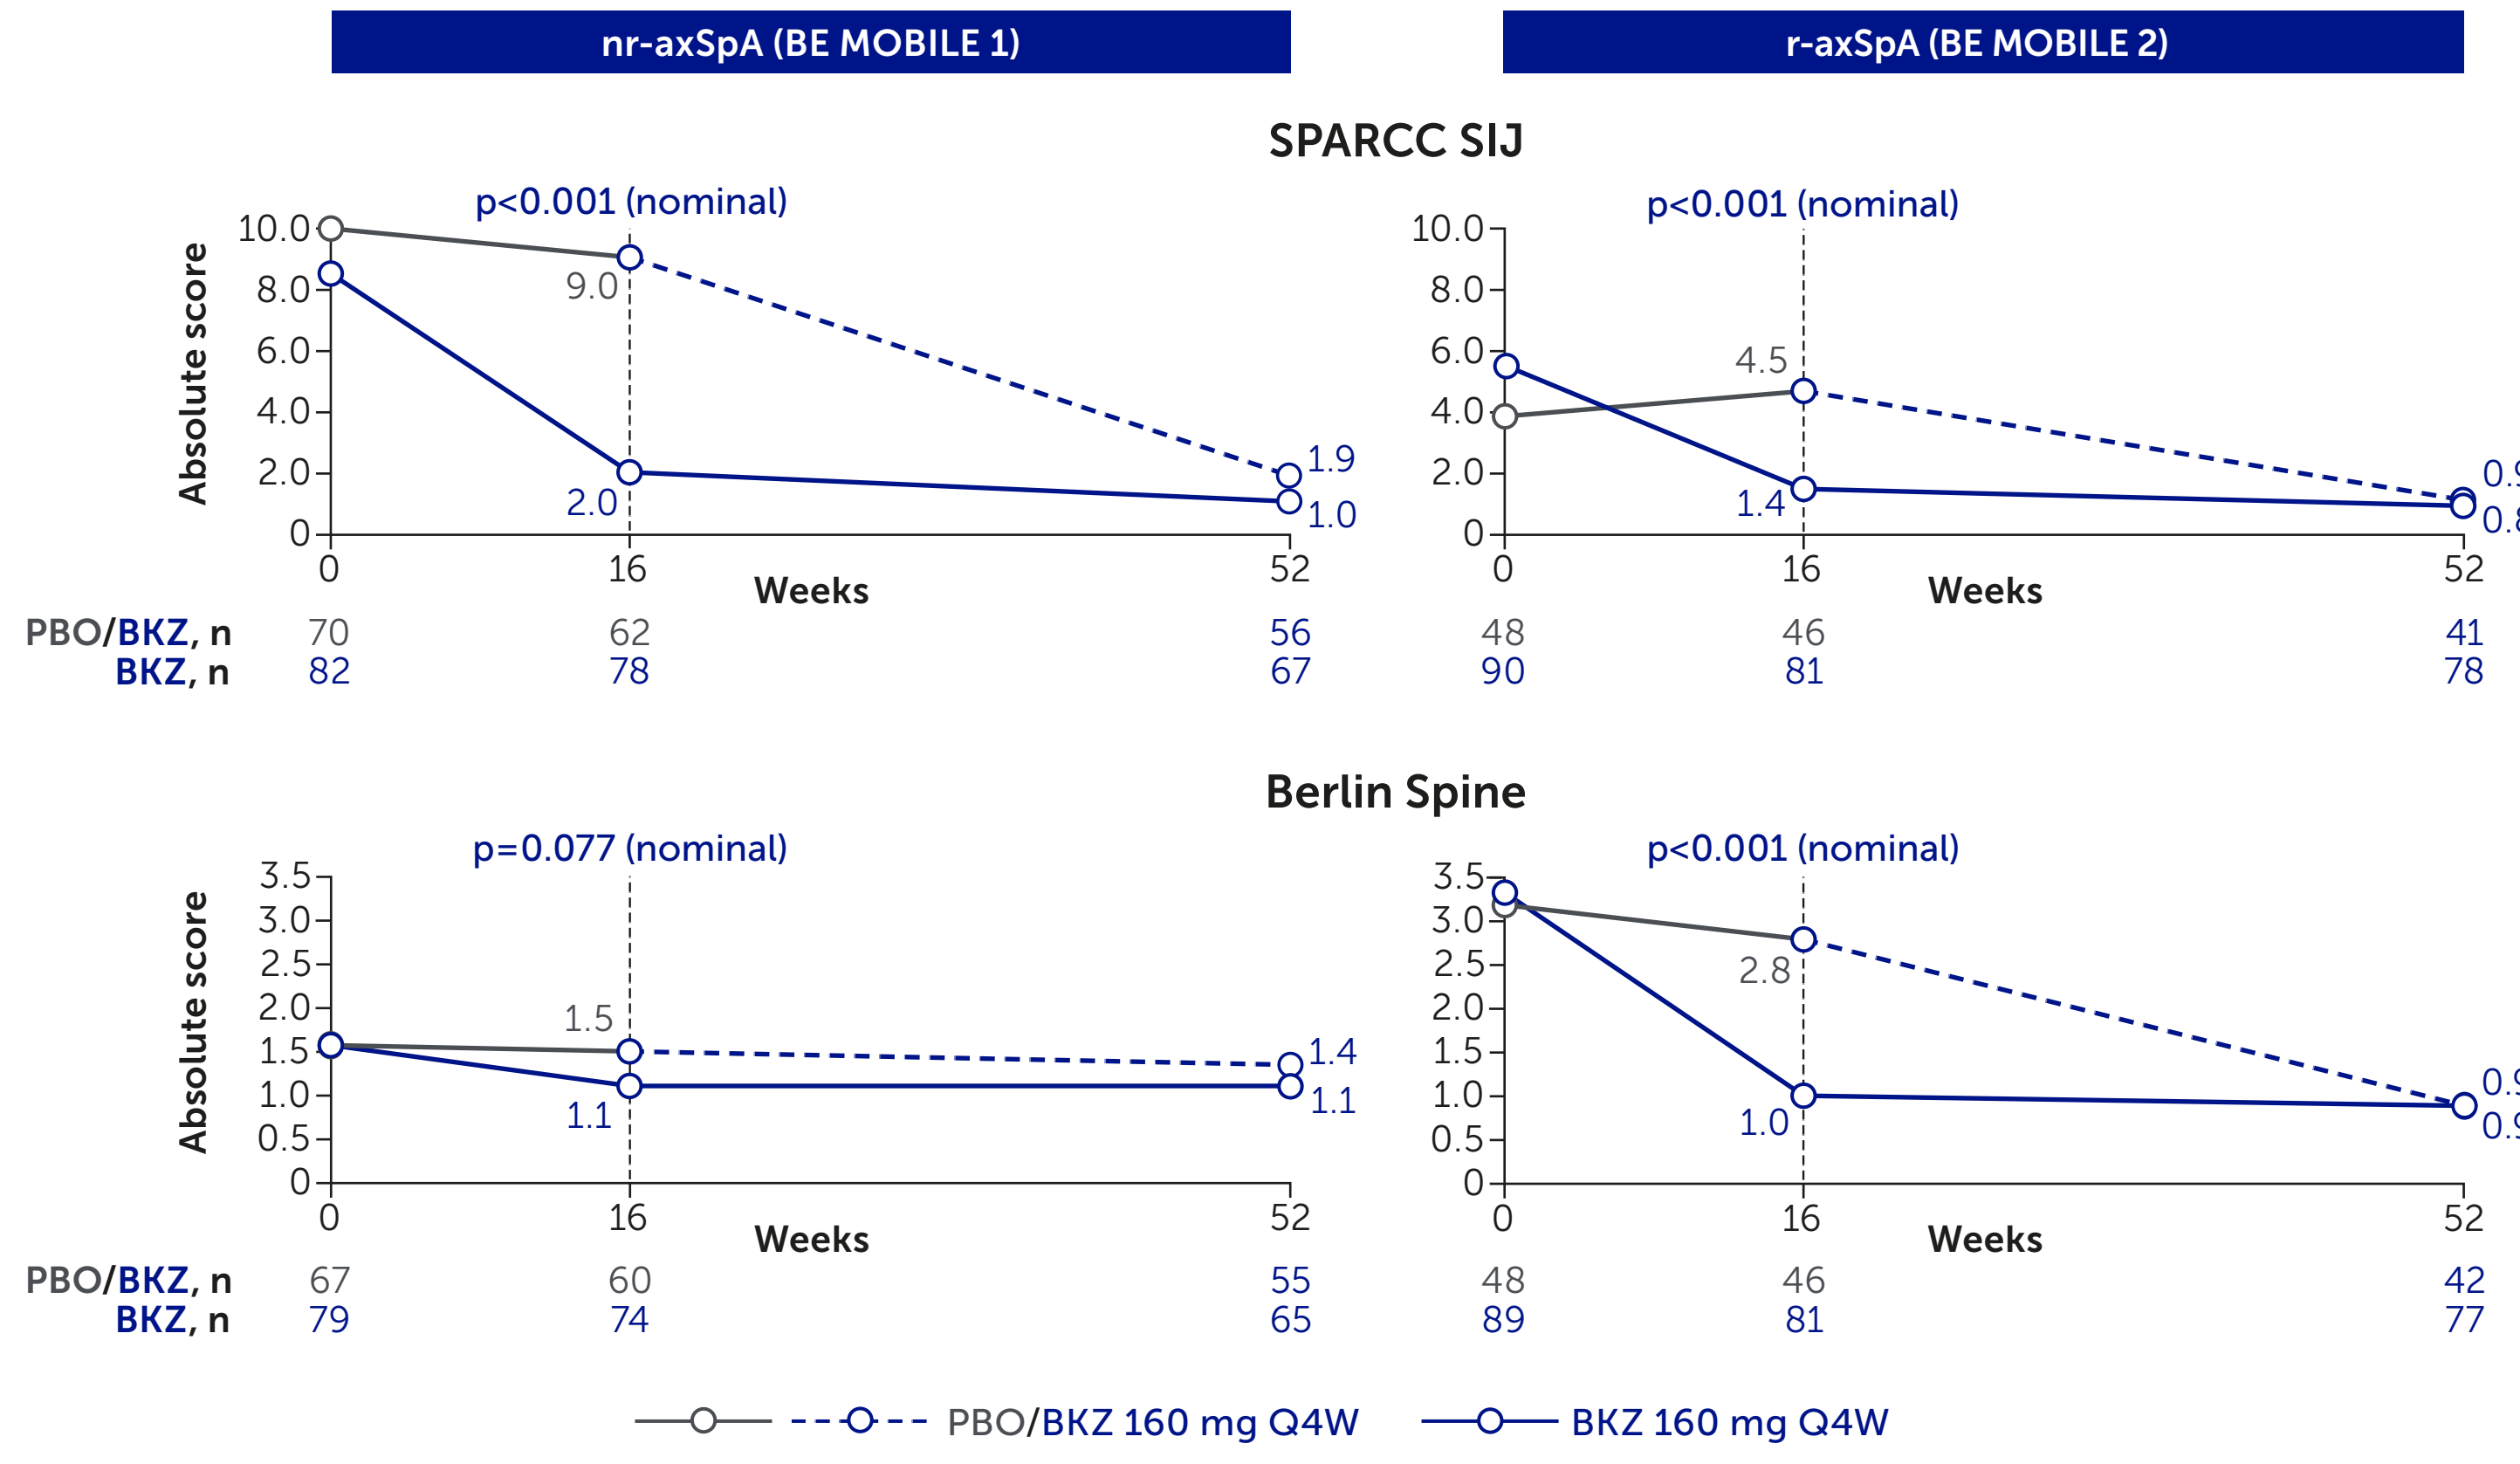
nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
SPARCC SIJ
p<0.001 (nominal)
p<0.001 (nominal)
Absolute score
Weeks
PBO/BKZ, n 70 62 56
BKZ, n 82 78 67
PBO/BKZ, n 48 46 41
BKZ, n 90 81 78
Berlin Spine
p=0.077 (nominal)
p<0.001 (nominal)
PBO/BKZ, n 67 60 55
BKZ, n 79 74 65
PBO/BKZ, n 48 46 42
BKZ, n 89 81 77
PBO/BKZ 160 mg Q4W
BKZ 160 mg Q4W

Randomised set. Includes only patients in the MRI sub-studies. SPARCC SIJ and Berlin spine scores range from 0–72 and 0–69, respectively, with lower scores indicating less inflammation.

Figure 3 Proportion of patients achieving MRI remission (OC)

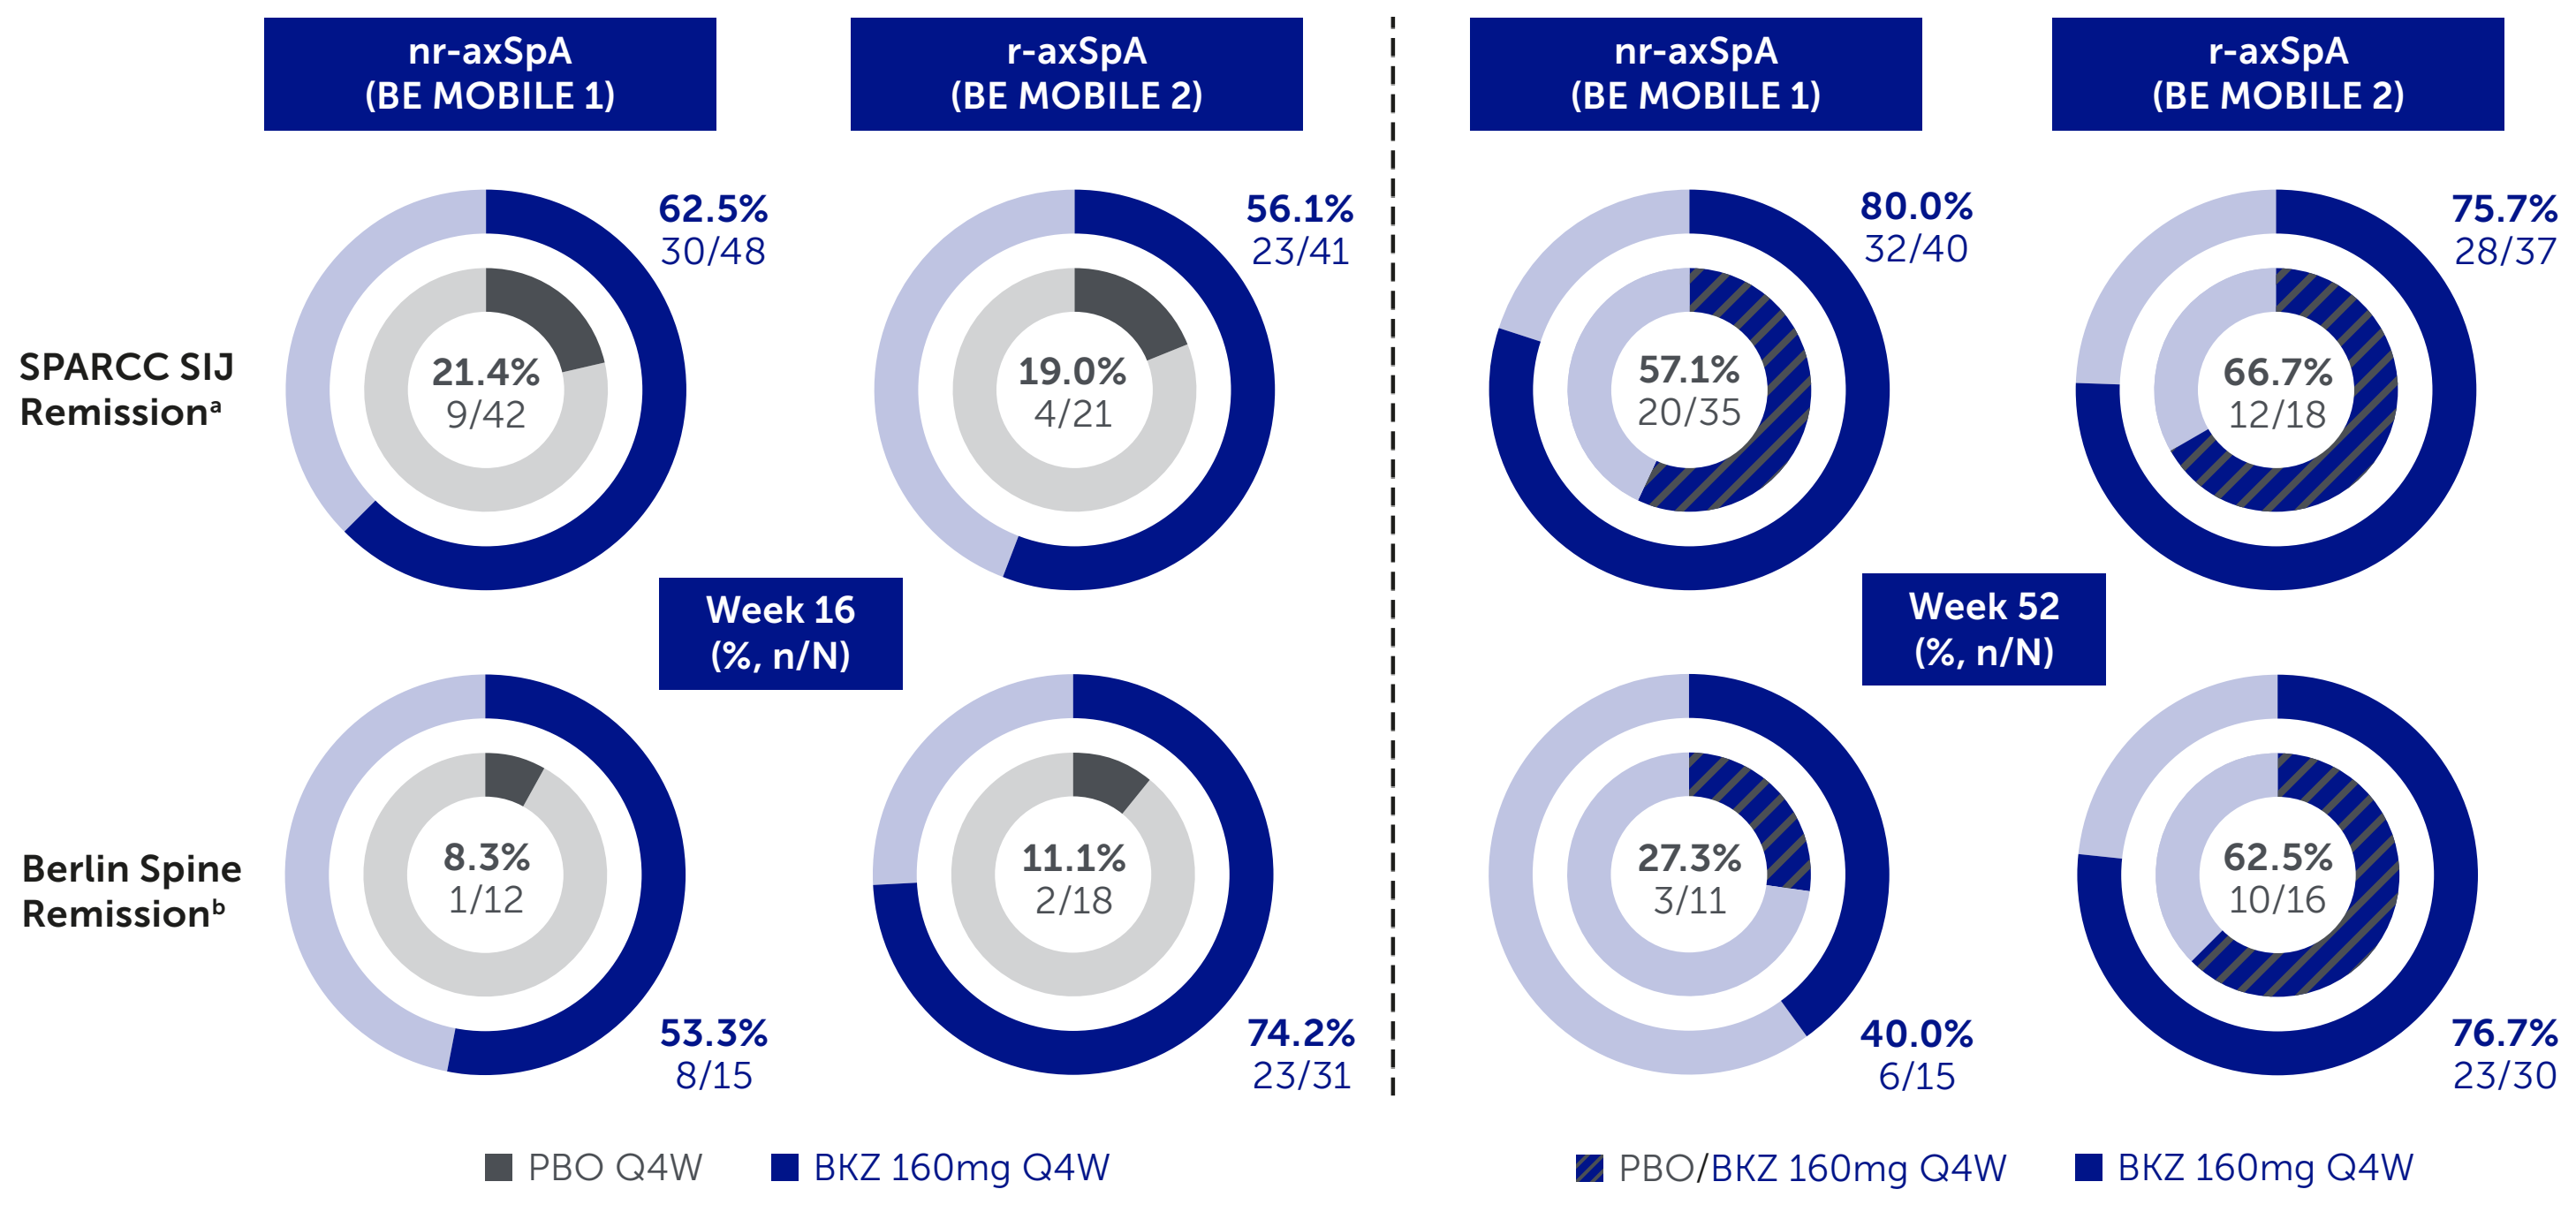
nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
SPARCC SIJ Remission[a]
Berlin Spine Remission[b]
Week 16 (%, n/N)
Week 52 (%, n/N)
62.5% 30/48
21.4% 9/42
56.1% 23/41
19.0% 4/21
80.0% 32/40
57.1% 20/35
75.7% 28/37
66.7% 12/18
53.3% 8/15
8.3% 1/12
74.2% 23/31
11.1% 2/18
40.0% 6/15
27.3% 3/11
76.7% 23/30
62.5% 10/16
PBO Q4W
BKZ 160mg Q4W
PBO/BKZ 160mg Q4W
BKZ 160mg Q4W

Randomised set. Includes only patients in the MRI sub-studies. [a]SPARCC SIJ <2, assessed in the subgroup of patients with SPARCC SIJ ≥2 at baseline; [b]Berlin spine ≤2, assessed in the subgroup of patients with Berlin spine >2 at baseline.

Figure 2 Mirror plots for baseline and CfB in MRI inflammation scores at Week 16 (OC)

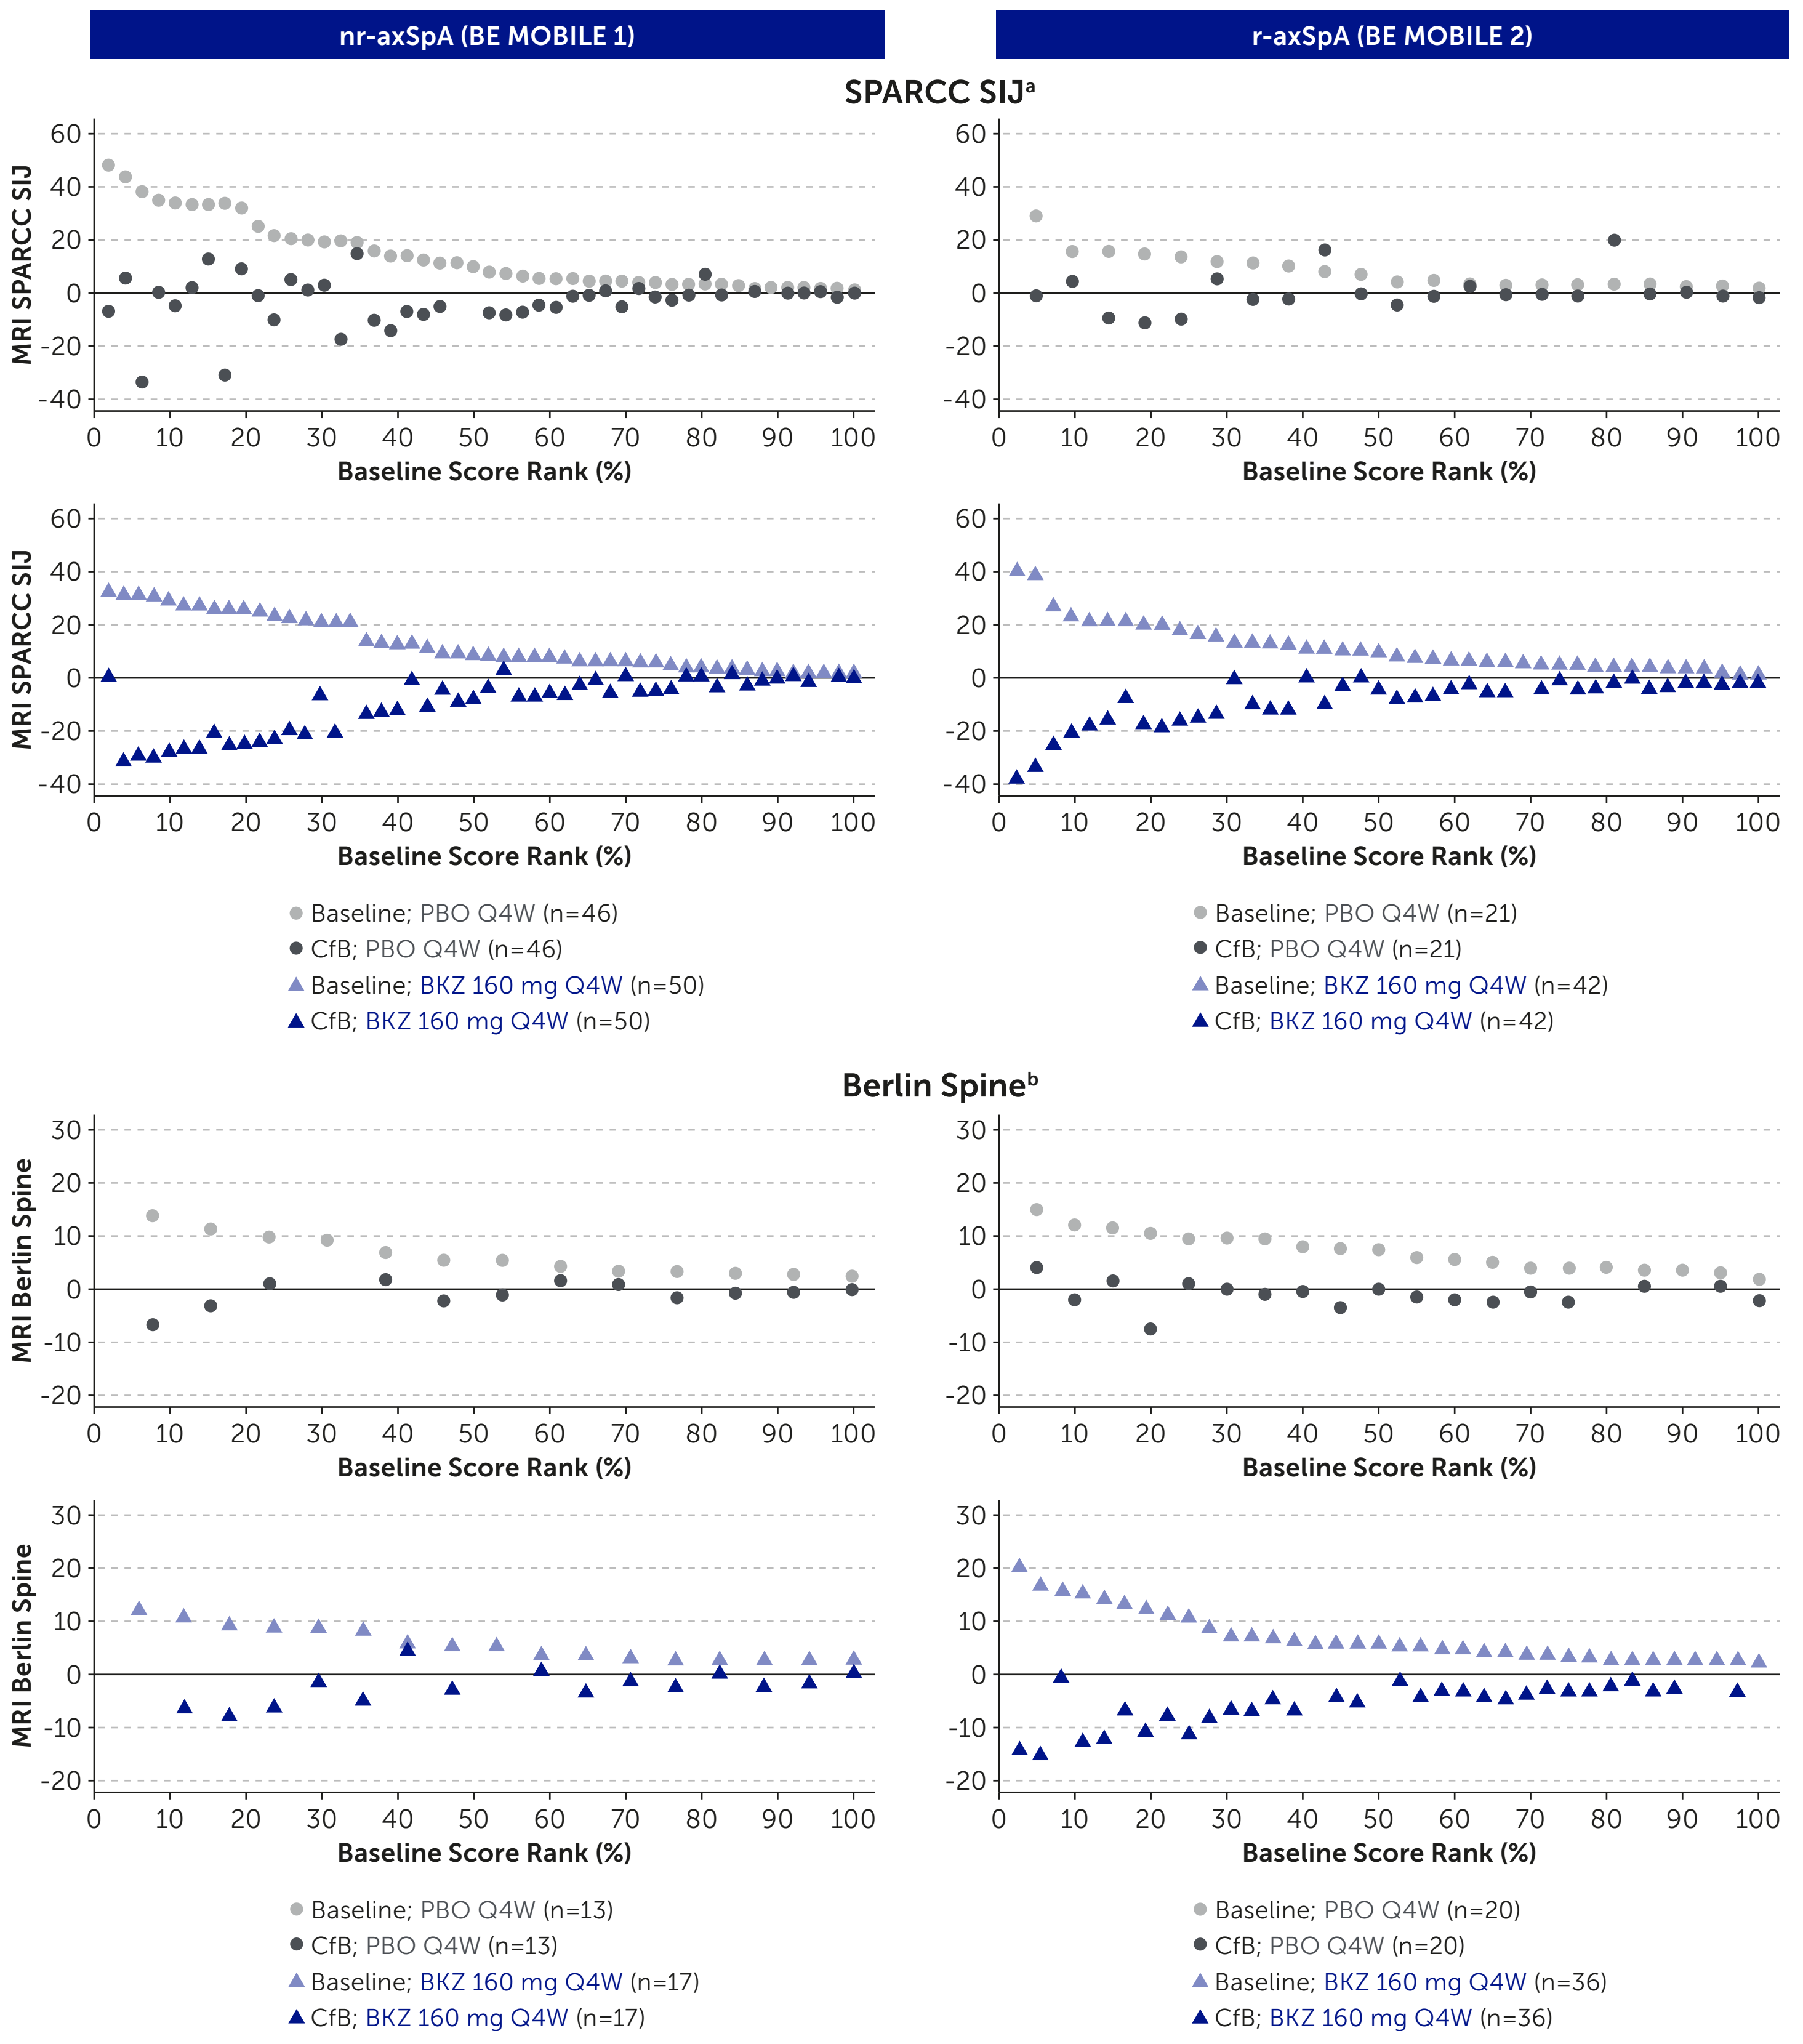
nr-axSpA (BE MOBILE 1)
r-axSpA (BE MOBILE 2)
SPARCC SIJ[a]
MRI SPARCC SIJ
Baseline Score Rank (%)
Baseline; PBO Q4W (n=46)
CfB; PBO Q4W (n=46)
Baseline; BKZ 160 mg Q4W (n=50)
CfB; BKZ 160 mg Q4W (n=50)
Baseline; PBO Q4W (n=21)
CfB; PBO Q4W (n=21)
Baseline; BKZ 160 mg Q4W (n=42)
CfB; BKZ 160 mg Q4W (n=42)
Berlin Spine[b]
MRI Berlin Spine
Baseline; PBO Q4W (n=13)
CfB; PBO Q4W (n=13)
Baseline; BKZ 160 mg Q4W (n=17)
CfB; BKZ 160 mg Q4W (n=17)
Baseline; PBO Q4W (n=20)
CfB; PBO Q4W (n=20)
Baseline; BKZ 160 mg Q4W (n=36)
CfB; BKZ 160 mg Q4W (n=36)

Randomised set. Includes only patients in the MRI sub-studies. [a]SPARCC SIJ assessed in the subgroup of patients with SPARCC SIJ ≥2 at baseline; [b]Berlin spine assessed in the subgroup of patients with Berlin spine >2 at baseline.

ASDAS: Ankylosing Spondylitis Disease Activity Score; **ASspiMRI-a:** Ankylosing Spondylitis spine MRI–activity; **BASDAI:** Bath Ankylosing Spondylitis Disease Activity Index; **BKZ:** bimekizumab; **CfB:** change from baseline; **CV:** coefficient of variation; **HLA-B27:** human leukocyte antigen-B27; **hs-CRP:** high-sensitivity C-reactive protein; **IL:** interleukin; **MRI:** magnetic resonance imaging; **n/N:** number of patients achieving the threshold/number of participants with a non-missing measurement for that timepoint; **nr-axSpA:** non-radiographic axial spondyloarthritis; **OC:** observed case; **PBO:** placebo; **Q4W:** every four weeks; **r-axSpA:** radiographic axial spondyloarthritis; **SD:** standard deviation; **SIJ:** sacroiliac joints; **SPARCC:** Spondyloarthritis Research Consortium of Canada; **TNFi:** tumour necrosis factor inhibitor.

Affiliations: [1]Ruhr-University Bochum, Rheumazentrum Ruhrgebiet Herne, Bochum, Germany; [2]La Paz University Hospital, Department of Rheumatology, IdiPaz, Madrid, Spain; [3]Charité – Universitätsmedizin Berlin, Department of Gastroenterology, Infectious Diseases and Rheumatology, Berlin, Germany; [4]Boston University School of Medicine, Department of Rheumatology, Massachusetts, USA; [5]Defence Medical Rehabilitation Centre, Academic Department of Military Rehabilitation, Stanford Hall, Loughborough, UK; [6]Ghent University Hospital, Department of Radiology, Ghent, Belgium; [7]UCB Pharma, Monheim am Rhein, Germany; [8]UCB Pharma, Brussels, Belgium; [9]UCB Pharma, Slough, UK; [10]UCB Pharma, Raleigh, North Carolina, USA; [11]University of Alberta, Department of Medicine, 568 Heritage Building, Edmonton, Alberta, Canada.

References: [1]Sieper J. Lancet 2017;390:73–84; [2]Navarro-Compán V. Ann Rheum Dis 2021;80:1511–21; [3]Baraliakos X. Arthritis Rheumatol 2022;74 (suppl 9); [4]Boel A. Ann Rheum Dis 2019;78:1545–9; [5]van der Heijde D. Ann Rheum Dis 2023;0:1–12; [6]van der Heijde D. Ann Rheum Dis 2018;77;699–705.
Author contributions: Substantial contributions to study conception/design or acquisition/analysis/interpretation of data: **XB**, **VNC**, **DP**, **MD**, **AB**, **LJ**, **UM**. **CF**, **TV**, **AME**, **NdP**, **WPM**; drafting of the publication or revising it critically for important intellectual content: **XB**, **VNC**, **DP**, **MD**, **AB**, **LJ**, **UM**. **CF**, **TV**, **AME**, **NdP**, **WPM**; final approval of the publication: **XB**, **VNC**, **DP**, **MD**, **AB**, **LJ**, **UM**. **CF**, **TV**, **AME**, **NdP**, **WPM**. **Disclosures: XB:** Speakers bureau for AbbVie, BMS, Chugai, Eli Lilly, Galapagos, Gilead, MSD, Novartis, Pfizer and UCB Pharma; paid instructor for AbbVie, BMS, Chugai, Eli Lilly, Galapagos, Gilead, MSD, Novartis, Pfizer and UCB Pharma; consultant for AbbVie, BMS, Chugai, Eli Lilly, Galapagos, Gilead, MSD, Novartis, Pfizer and UCB Pharma; **VNC:** Speakers bureau for AbbVie, Eli Lilly, Janssen, MSD, Novartis, Pfizer and UCB Pharma; consultant for AbbVie, Eli Lilly, MSD, Novartis, Pfizer and UCB Pharma; research grants from AbbVie and Novartis; **DP:** Speaker for AbbVie, BMS, Eli Lilly, MSD, Novartis, Pfizer and UCB Pharma; consultant for AbbVie, Biocad, Eli Lilly, Gilead, GSK, Novartis, Pfizer, Samsung Bioepis and UCB Pharma; grant/research support from AbbVie, Eli Lilly, MSD, Novartis and Pfizer; **MD:** Educational and research grants from Pfizer paid to institution; consultant for Amgen and UCB Pharma; **AB:** Teaching honorarium from AbbVie, Biogen, Novartis, Pfizer and UCB Pharma; research grants from Pfizer; advisory board fees from AbbVie, Eli Lilly, MSD, Novartis, Pfizer and UCB Pharma; **LJ:** Speaker for AbbVie, Eli Lilly, Novartis and UCB Pharma; **UM, CF, TV, AME, NdP:** Employees of UCB Pharma; **WPM:** Honoraria/consulting fees from AbbVie, BMS, Boehringer-Ingelheim, Celgene, Eli Lilly, Galapagos, Janssen, Novartis, Pfizer and UCB Pharma; research grants from AbbVie, Pfizer and UCB Pharma; educational grants from AbbVie, Janssen, Novartis and Pfizer; Chief Medical Officer for CARE ARTHRITIS. **Acknowledgements:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Celia Menckeberg, UCB Pharma, Breda, The Netherlands, for publication coordination, Faye Bolan, PhD, Costello Medical, Manchester, UK for medical writing and editorial assistance, and the Costello Medical Creative team for design support. These studies were funded by UCB Pharma. All costs associated with development of this presentation were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit:
https://ucbposters.com/?PosterID=EULAR2023_POS0246

Poster ID: POS0246

Link expiration: 17 June 2023

Presented at EULAR 2023 | Milan, Italy | 31 May–3 June 2023