Bimekizumab Improvements in Efficacy on Disease Activity Assessed via Composite Endpoints in Biologic DMARD-Naïve and TNFi-IR Patients with Active PsA: Pooled 16-Week Results from Phase 3 Randomized, Placebo-Controlled Studies

CONTENT PROVIDED FOR SHAREHOLDERS, INVESTORS AND OTHER CAPITAL MARKET PARTICIPANTS ONLY

Philip J. Mease,[1] Laura C. Coates,[2] Robert Landewé,[3] Iain B. McInnes,[4] Christopher T. Ritchlin,[5] Tatsuya Atsumi,[6] Frank Behrens,[7] Dafna D. Gladman,[8] Laure Gossec,[9,10] Peter Nash,[11] Barbara Ink,[12] Deepak Assudani,[12] Rajan Bajracharya,[12] Jason Coarse,[13] Adam R. Prickett,[12] Alice B. Gottlieb[14]

Presented at ACR Convergence 2022 | Philadelphia, Pennsylvania, USA | November 10–14

2117

Objective

To assess the efficacy of bimekizumab (BKZ) treatment versus placebo (PBO) on disease activity using composite outcome measures in patients with active psoriatic arthritis (PsA) using pooled data from two phase 3 trials.

Background

- PsA is a disease with multiple manifestations; it is important that the efficacy of new interventions is assessed across the spectrum of the disease using composite endpoints.[1]
- BKZ, a monoclonal IgG1 antibody that selectively inhibits IL-17F in addition to interleukin (IL)-17A, has shown efficacy and tolerability up to 16 weeks in patients with active PsA.[2,3]

Methods

- BE OPTIMAL (NCT03895203) and BE COMPLETE (NCT03896581) are phase 3 studies assessing BKZ in patients with PsA who are biologic DMARD (bDMARD)-naïve or had intolerance or inadequate response to TNF inhibitors (TNFi-IR), respectively (**Figure 1**).
- The primary endpoint in both studies was the proportion of patients with ≥50% improvement in American College of Rheumatology criteria (ACR50) response at Week 16; minimal disease activity (MDA) at Week 16 was a secondary endpoint.
- We present pooled and individual study data for BKZ and PBO treatment arms for composite endpoints at Week 16: MDA and very low disease activity (VLDA), Disease Activity in Psoriatic Arthritis (DAPSA), Psoriatic Arthritis Disease Activity Score (PASDAS), and ACR50+Psoriasis Area and Severity Index (PASI)100.
- Non-responder imputation (NRI) and worst-category imputation (WCI) were used for missing binary and multi-category data, respectively.

Results

- A total of 1,073/1,112 (96.5%) patients randomized to BKZ or PBO completed Week 16.
- Baseline characteristics were generally comparable between studies, with numerical differences between BE OPTIMAL and BE COMPLETE for time since PsA diagnosis, concomitant methotrexate, and body surface area (BSA) affected by psoriasis ≥3% (**Table 1**).
- At Week 16 (pooled and individual analyses), a higher proportion of BKZ-treated patients achieved MDA and VLDA versus PBO-treated patients (**Figure 2**).
- Compared with the PBO group, a higher proportion of BKZ-treated patients achieved low disease activity (LDA) and remission (REM) at Week 16, as measured by DAPSA and PASDAS (**Figure 3A–B**).
- Numerically higher proportions of BKZ- versus PBO-treated patients achieved ACR50+PASI100 (**Figure 3C**).

Conclusions

Pooled data demonstrate that numerically higher proportions of bimekizumab-treated patients achieved clinically relevant disease activity thresholds, assessed by composite endpoints, compared with placebo.

Results were consistent between studies, with similar magnitude of response observed, suggesting that bimekizumab treatment leads to improvements in overall disease activity irrespective of prior bDMARD use.

Summary

Composite endpoints are key in assessing the efficacy of new treatments across multiple domains of PsA.[1]

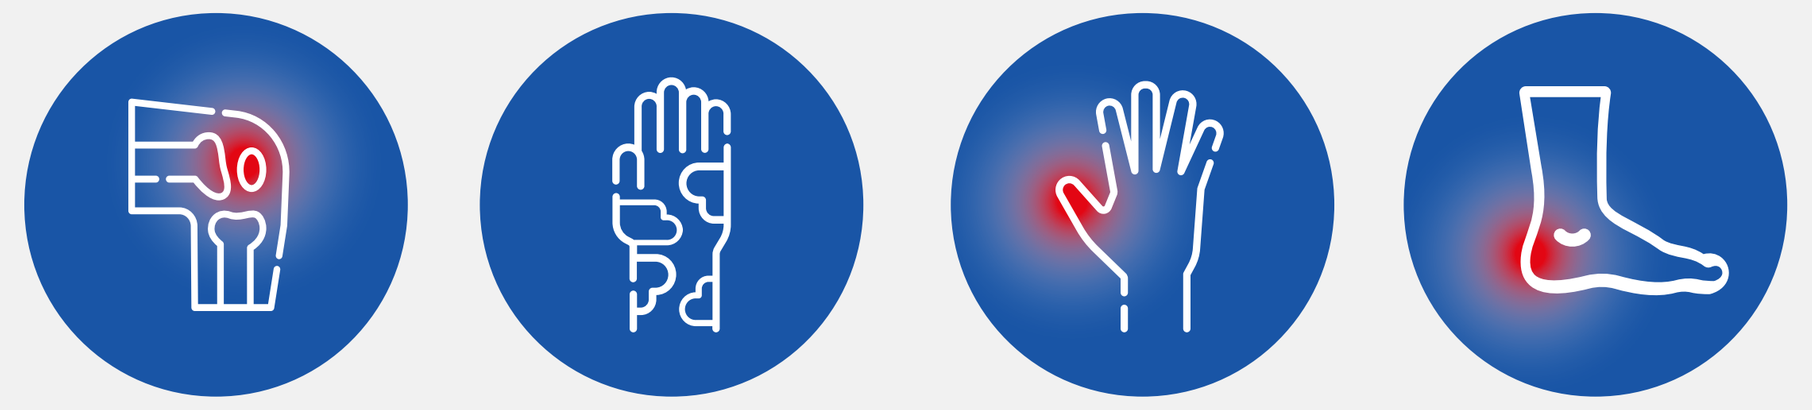
Joints
Skin
Dactylitis
Enthesitis

Numerically higher proportions of BKZ-treated patients achieved clinically relevant disease activity thresholds at Week 16, irrespective of prior bDMARD use:

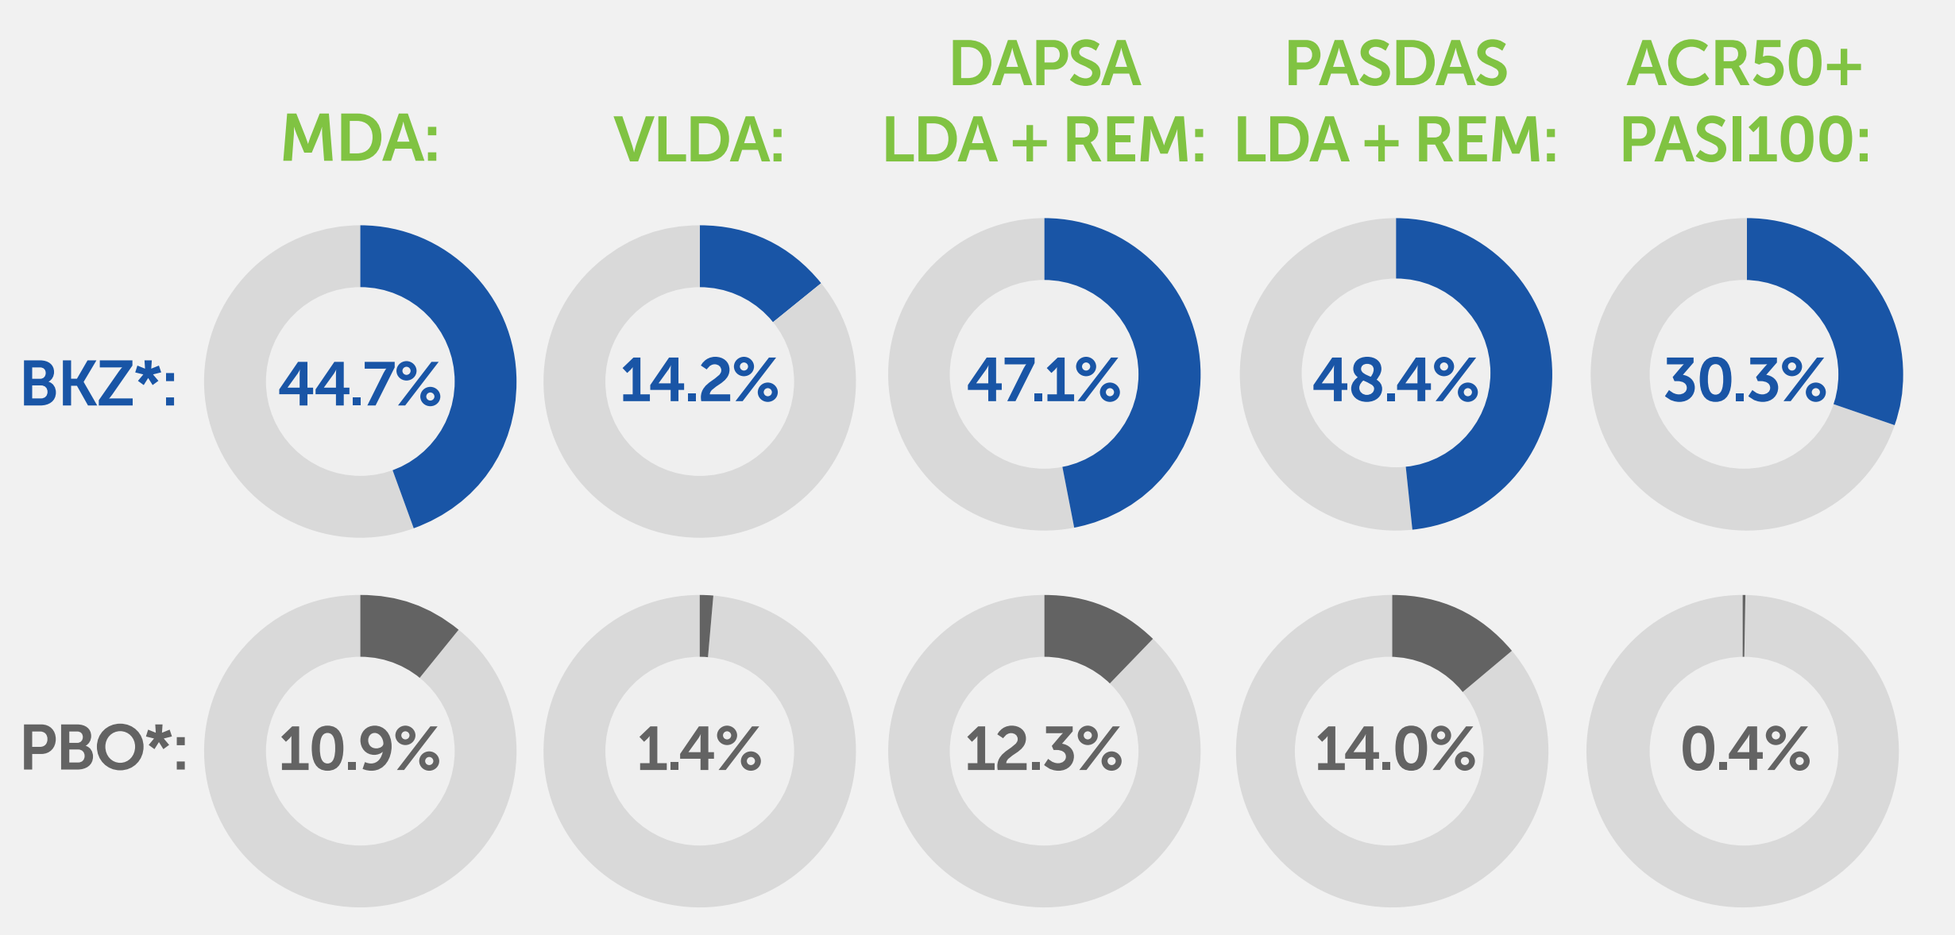
MDA:
VLDA:
DAPSA LDA + REM:
PASDAS LDA + REM:
ACR50+ PASI100:
BKZ*:
44.7%
14.2%
47.1%
48.4%
30.3%
PBO*:
10.9%
1.4%
12.3%
14.0%
0.4%

Results suggest that bimekizumab treatment leads to improvement in overall disease activity. Similar magnitude of response was observed in bDMARD-naïve and TNFi-IR patients.

*Based on pooled data for composite endpoints at Week 16.

Figure 1 BE OPTIMAL and BE COMPLETE study designs

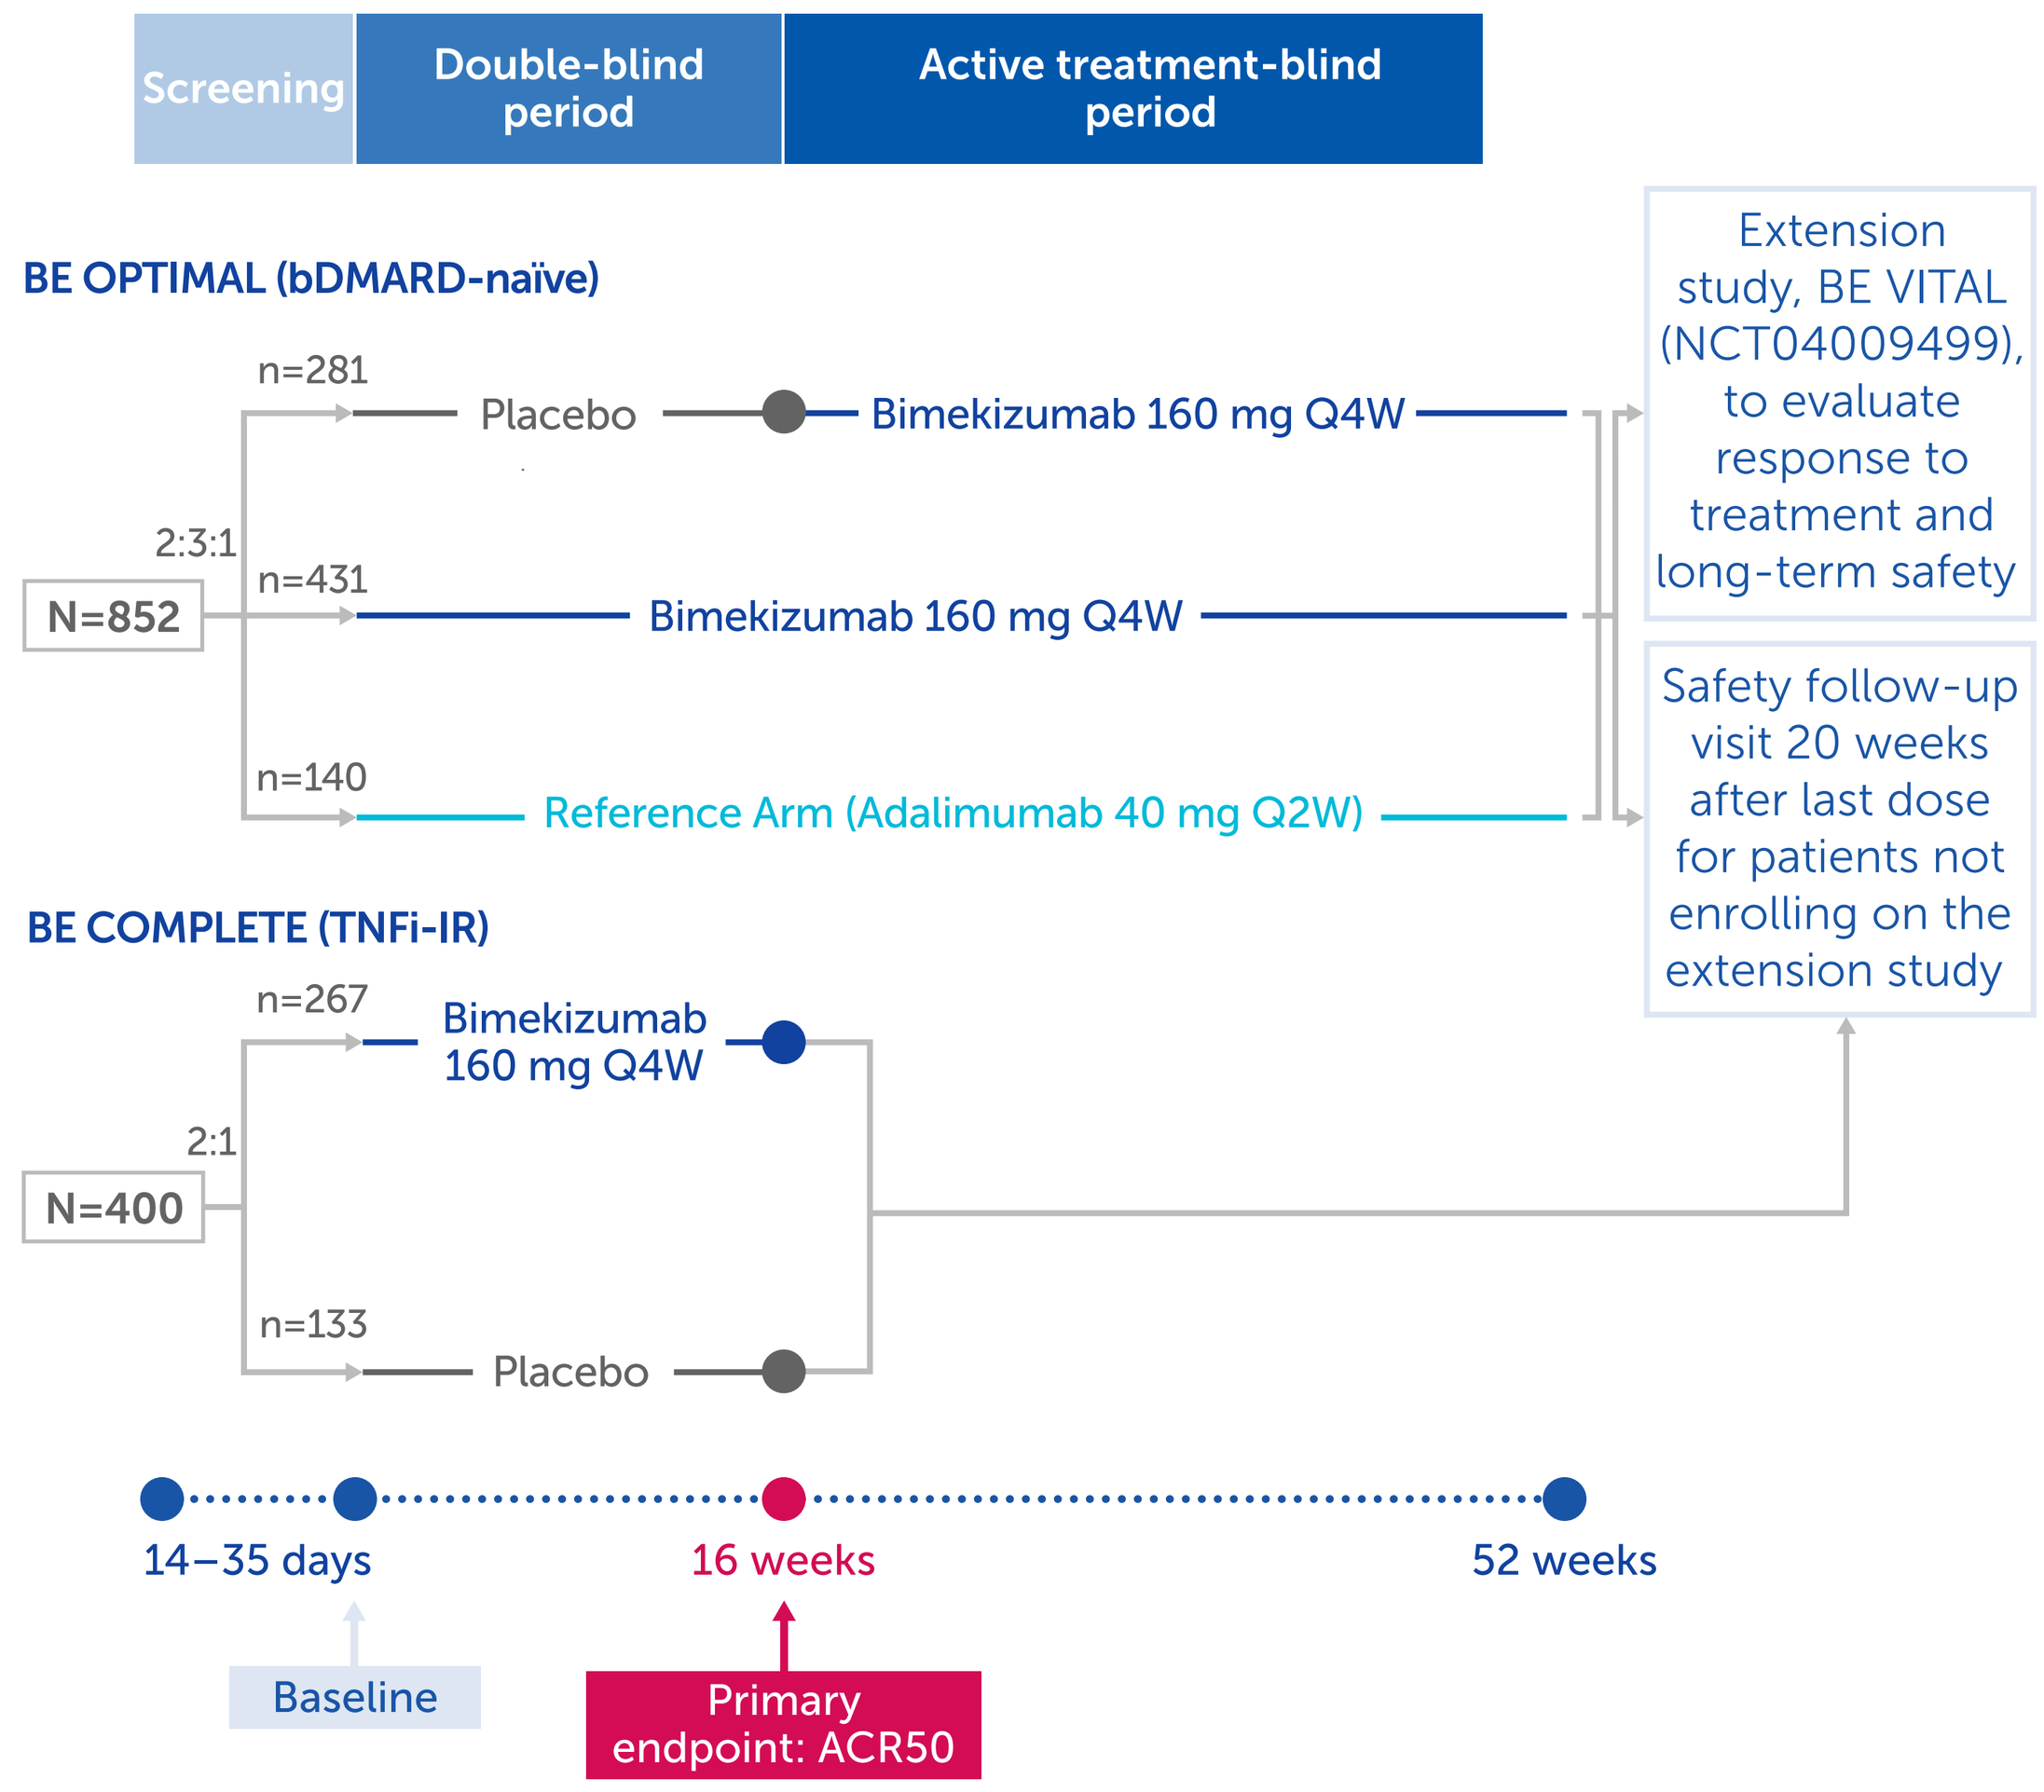
Screening
Double-blind period
Active treatment-blind period
BE OPTIMAL (bDMARD-naïve)
n=281
Placebo
Bimekizumab 160 mg Q4W
2:3:1
N=852
n=431
Bimekizumab 160 mg Q4W
n=140
Reference Arm (Adalimumab 40 mg Q2W)
Extension study, BE VITAL (NCT04009499), to evaluate response to treatment and long-term safety
Safety follow-up visit 20 weeks after last dose for patients not enrolling on the extension study
BE COMPLETE (TNFi-IR)
n=267
Bimekizumab 160 mg Q4W
2:1
N=400
n=133
Placebo
14–35 days
16 weeks
52 weeks
Baseline
Primary endpoint: ACR50

Study designs that made up the pooled population, BE OPTIMAL (NCT03895203) and BE COMPLETE (NCT03896581). Adalimumab 40 mg Q2W served as an active reference arm. BE OPTIMAL was not powered for comparisons of adalimumab to bimekizumab or adalimumab to placebo. Adalimumab 40 mg Q2W data are not shown in this poster.

Table 1 Baseline characteristics

	Pooled Analysis (bDMARD-naïve + TNFi-IR)		BE OPTIMAL (bDMARD-naïve)		BE COMPLETE (TNFi-IR)	
	PBO (n=414)	BKZ 160 mg Q4W (n=698)	PBO (n=281)	BKZ 160 mg Q4W (n=431)	PBO (n=133)	BKZ 160 mg Q4W (n=267)
Age, mean (SD)	49.5 (12.2)	49.1 (12.5)	48.7 (11.7)	48.5 (12.6)	51.3 (12.9)	50.1 (12.4)
Male, n (%)	187 (45.2)	331 (47.4)	127 (45.2)	201 (46.6)	60 (45.1)	130 (48.7)
BMI, kg/m², mean (SD)	29.4 (5.9)	29.6 (6.7)	29.6 (6.1)	29.2 (6.8)	29.0 (5.4)	30.1 (6.5)
Time since PsA diagnosis (years), mean (SD)	6.8 (7.3)[a]	7.4 (8.6)[b]	5.6 (6.5)[c]	6.0 (7.3)[d]	9.2 (8.1)[e]	9.6 (9.9)[f]
MTX at baseline, n (%)	213 (51.4)	371 (53.2)	162 (57.7)	252 (58.5)	51 (38.3)	119 (44.6)
BSA affected by psoriasis ≥3%, n (%)	228 (55.1)	393 (56.3)	140 (49.8)	217 (50.3)	88 (66.2)	176 (65.9)
PASI,[g] mean (SD)	8.1 (6.0)	9.1 (8.0)	7.9 (5.6)	8.2 (6.8)	8.5 (6.6)	10.1 (9.1)
TJC (of 68 joints), mean (SD)	17.8 (13.1)	17.4 (12.5)	17.1 (12.5)	16.8 (11.8)	19.3 (14.2)	18.4 (13.5)
SJC (of 66 joints), mean (SD)	9.7 (7.6)	9.2 (6.7)	9.5 (7.3)	9.0 (6.2)	10.3 (8.2)	9.7 (7.5)
Enthesitis,[h] n (%)	106 (25.6)[i]	249 (35.7)[j]	70 (24.9)	143 (33.2)[j]	36 (27.1)[i]	106 (39.7)
Dactylitis, n (%)	47 (11.4)[k]	90 (12.9)[l]	33 (11.7)[i]	56 (13.0)[l]	14 (10.5)[i]	34 (12.7)
hs-CRP ≥6 mg/L, n (%)	180 (43.5)	276 (39.5)	121 (43.1)	158 (36.7)	59 (44.4)	118 (44.2)
HAQ-DI, mean (SD)	0.9 (0.6)	0.9 (0.6)[i]	0.9 (0.6)	0.8 (0.6)[i]	1.0 (0.7)	1.0 (0.6)
PtAAP, mean (SD)	58.4 (23.8)	55.4 (24.3)[i]	56.8 (23.2)	53.6 (24.3)[i]	61.7 (24.6)	58.3 (24.2)

[a]n=411; [b]n=689; [c]n=279; [d]n=423; [e]n=132; [f]n=266; [g]For patients with psoriasis involving at least 3% of BSA at baseline; [h]As per the Leeds Enthesitis Index; [i]Data missing for 1 patient; [j]Data missing for 6 patients; [k]Data missing for 2 patients; [l]Data missing for 7 patients.

Figure 2 Patients achieving MDA and VLDA to Week 16 (NRI)

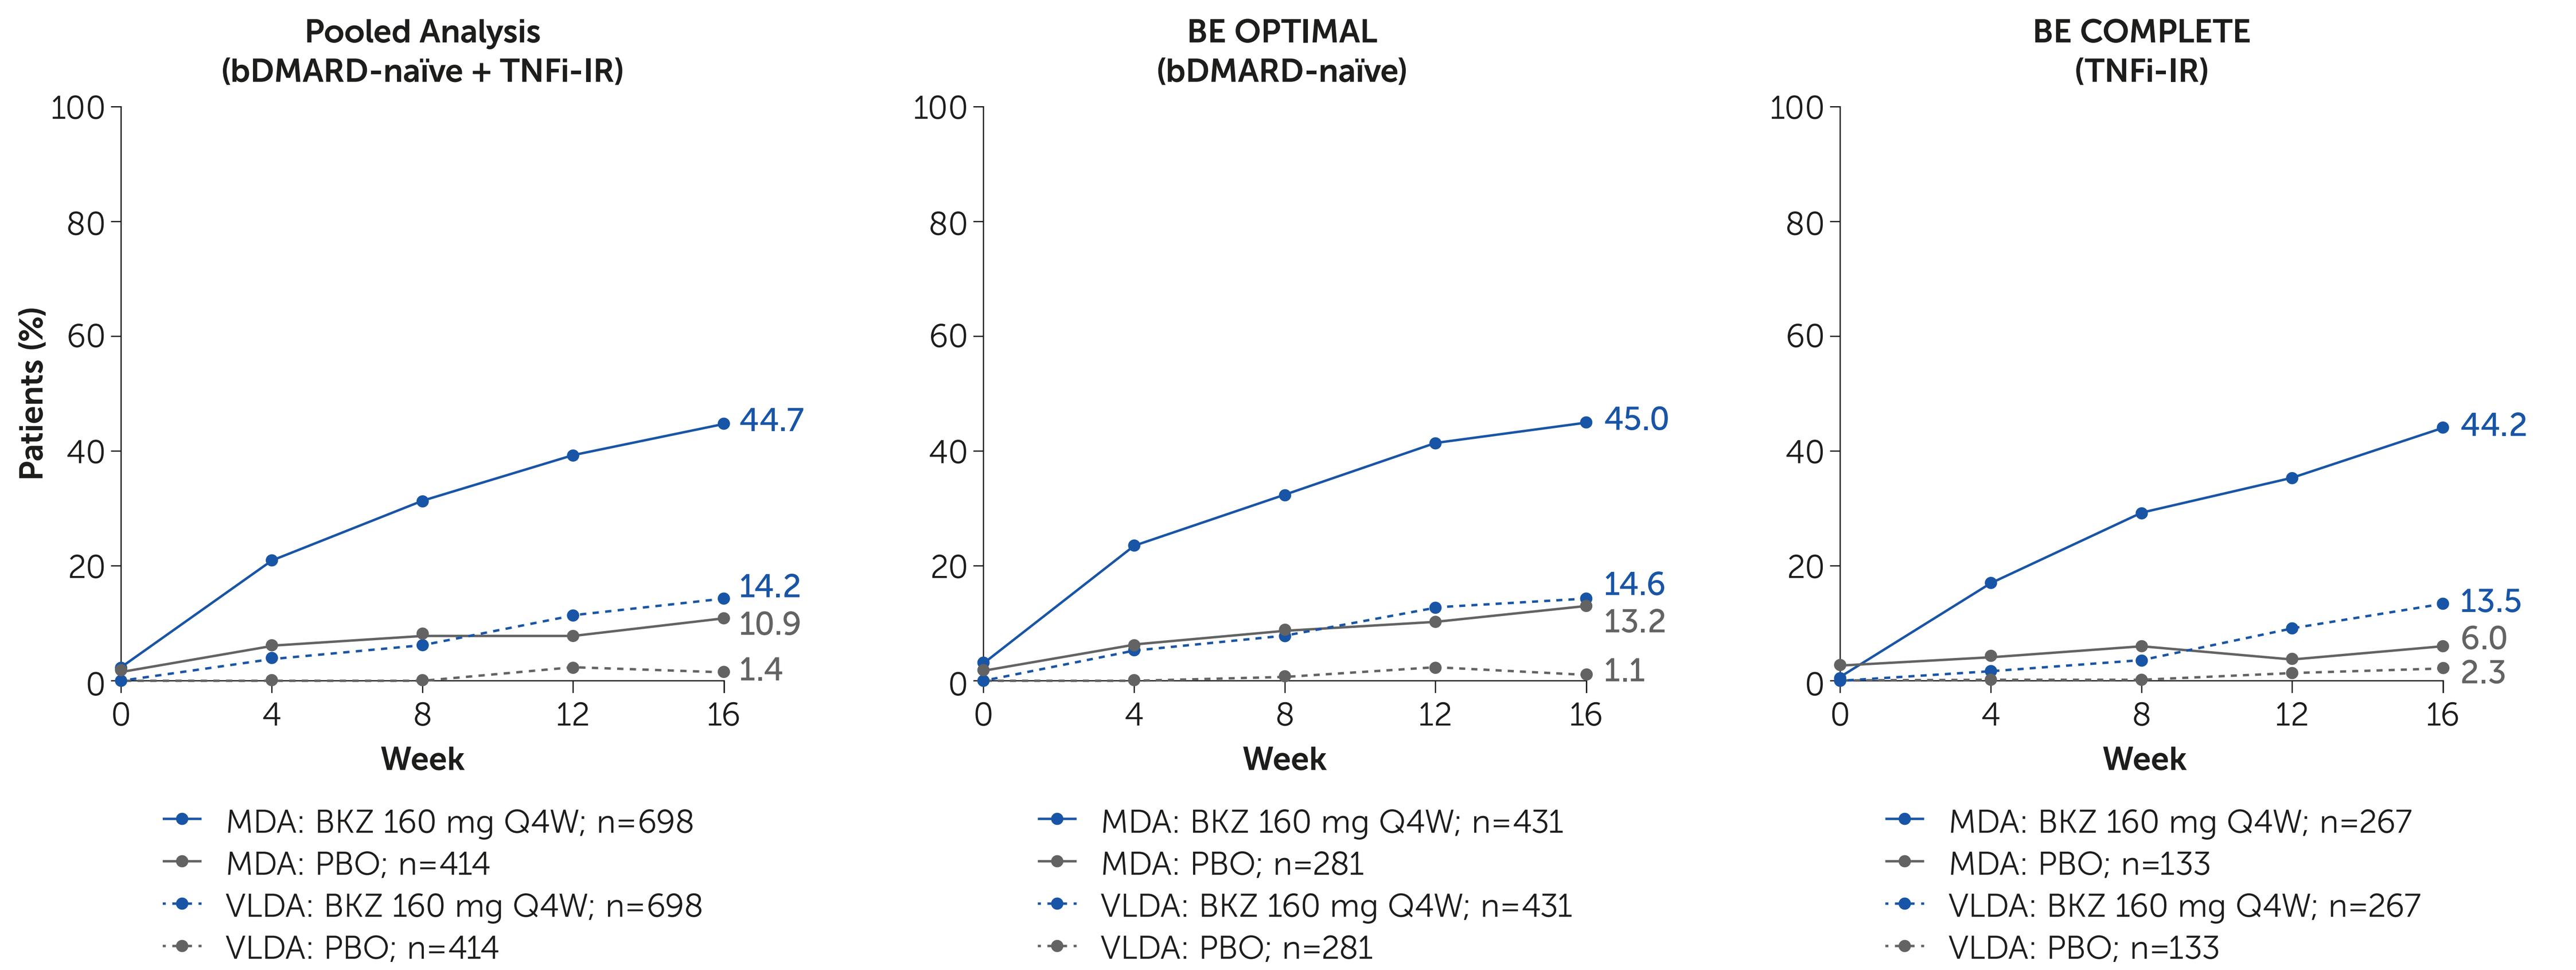
Pooled Analysis (bDMARD-naïve + TNFi-IR)
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
Patients (%)
Week
44.7
14.2
10.9
1.4
45.0
14.6
13.2
1.1
44.2
13.5
6.0
2.3
MDA: BKZ 160 mg Q4W; n=698
MDA: PBO; n=414
VLDA: BKZ 160 mg Q4W; n=698
VLDA: PBO; n=414
MDA: BKZ 160 mg Q4W; n=431
MDA: PBO; n=281
VLDA: BKZ 160 mg Q4W; n=431
VLDA: PBO; n=281
MDA: BKZ 160 mg Q4W; n=267
MDA: PBO; n=133
VLDA: BKZ 160 mg Q4W; n=267
VLDA: PBO; n=133

Randomized set. NRI. Percentage of MDA and VLDA responders from Baseline (Week 0) to Week 16 shown for the pooled population (N=1,112), BE OPTIMAL (N=712) and BE COMPLETE (N=400). The pooled population consists of the BE OPTIMAL and BE COMPLETE populations. A patient has achieved MDA if 5/7 key outcome measures are achieved, and VLDA if 7/7 key outcome measures are achieved. Outcome measures are TJC ≤1, SJC ≤1, PASI ≤1 or BSA ≤3, patient pain VAS ≤15 mm, patient global disease activity VAS ≤20 mm, HAQ-DI ≤0.5, tender entheseal joints ≤1.

Figure 3 Patients achieving other composite endpoints at Week 16

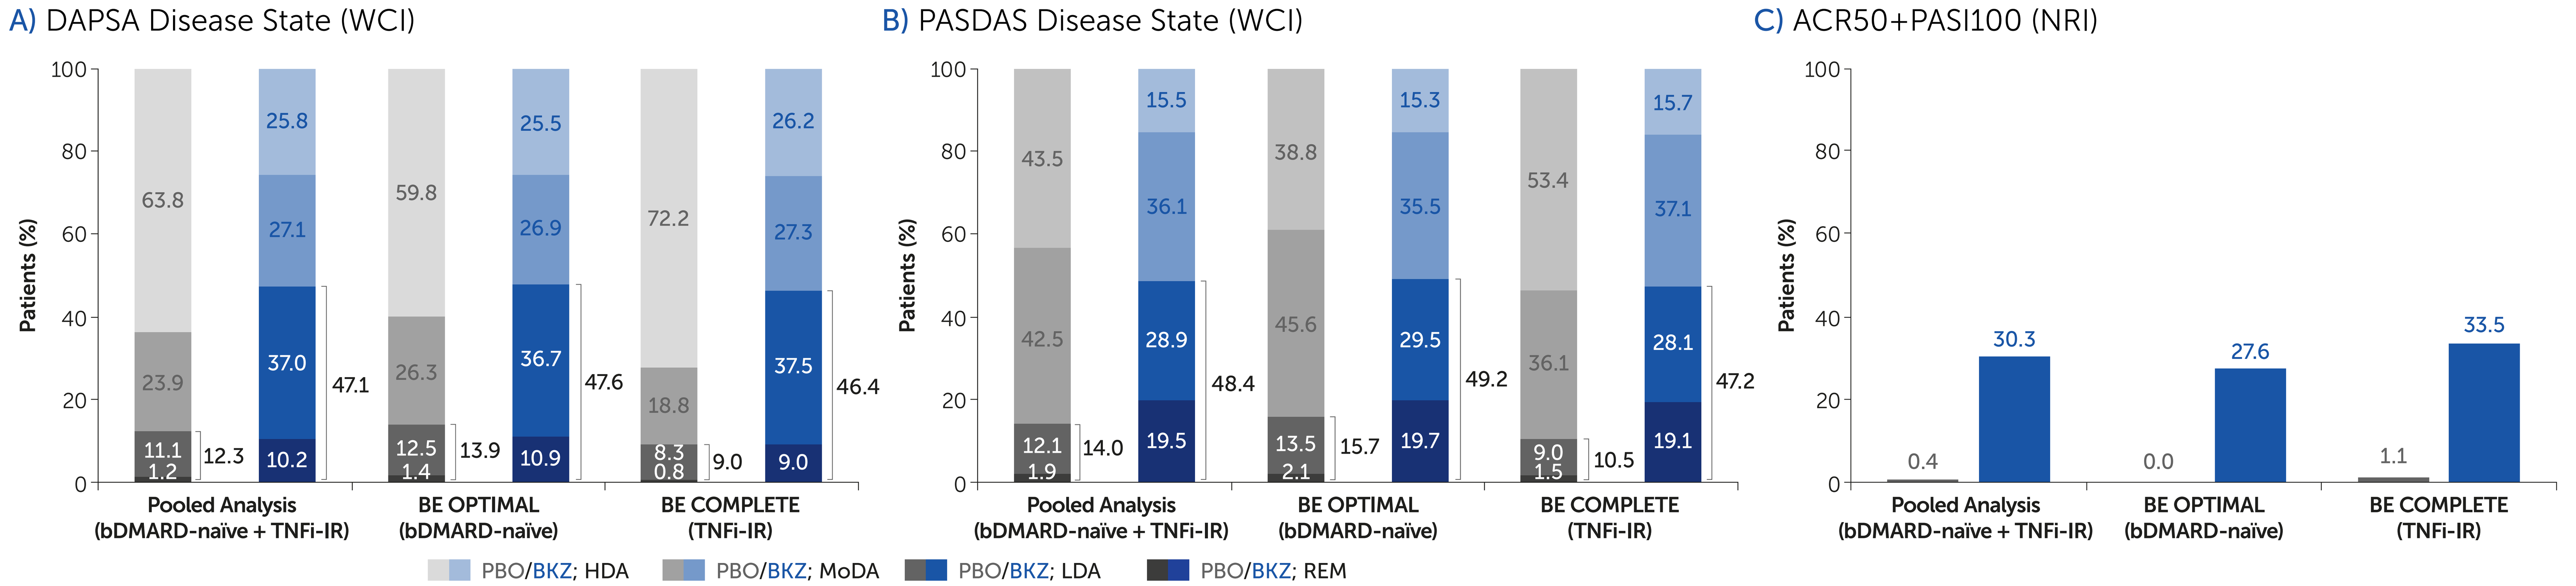
A) DAPSA Disease State (WCI)
B) PASDAS Disease State (WCI)
C) ACR50+PASI100 (NRI)
Patients (%)
Pooled Analysis (bDMARD-naïve + TNFi-IR)
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
63.8
23.9
11.1
1.2
12.3
25.8
27.1
37.0
10.2
47.1
59.8
26.3
12.5
1.4
13.9
25.5
26.9
36.7
10.9
47.6
72.2
18.8
8.3
0.8
9.0
26.2
27.3
37.5
9.0
46.4
43.5
42.5
12.1
1.9
14.0
15.5
36.1
28.9
19.5
48.4
38.8
45.6
13.5
2.1
15.7
15.3
35.5
29.5
19.7
49.2
53.4
36.1
9.0
1.5
10.5
15.7
37.1
28.1
19.1
47.2
0.4
30.3
0.0
27.6
1.1
33.5
PBO/BKZ; HDA
PBO/BKZ; MoDA
PBO/BKZ; LDA
PBO/BKZ; REM

Randomized set. DAPSA state, PASDAS category, and ACR50+PASI100 responders at Week 16 in patients treated with BKZ and PBO. DAPSA is a composite score that includes SJC, TJC, patient global VAS, PtAAP, and hs-CRP (mg/L). Patients achieve HDA if DAPSA value is >28, MoDA if DAPSA value is >14 and ≤28, LDA if DAPSA value is >4 and ≤14, and REM if DAPSA ≤4. PASDAS is a composite score that includes physician and patient global VAS, SJC, TJC, LEI, tender dactylitis count, SF-36 PCS, and hs-CRP. Patients achieve HDA if PASDAS score is ≥5.4, MoDA if PASDAS score is ≥3.2 and ≤5.4, LDA if PASDAS score is >1.9 and <3.2, and REM if PASDAS ≤1.9. Patients achieve ACR50+PASI100 if they had psoriasis involving at least 3% BSA at baseline, and achieved both a 50% improvement in ACR criteria and complete skin clearance.

ACR50: ≥50% improvement in American College of Rheumatology criteria; **BKZ:** bimekizumab; **bDMARD:** biologic disease-modifying antirheumatic drug; **BMI:** body mass index; **BSA:** body surface area; **DAPSA:** Disease Activity in Psoriatic Arthritis; **HAQ-DI:** Health Assessment Questionnaire-Disability Index; **HDA:** high disease activity; **hs-CRP:** high-sensitivity C-Reactive Protein; **IL:** interleukin; **LDA:** low disease activity; **LEI:** Leeds Enthesitis Index; **MDA:** minimal disease activity; **MoDA:** moderate disease activity; **MTX:** methotrexate; **NRI:** non-responder imputation; **PASI:** Psoriasis Area and Severity Index; **PASI100:** 100% improvement in PASI; **PASDAS:** Psoriatic Arthritis Disease Activity Score; **PBO:** placebo; **PCS:** Physical Component Summary; **PsA:** psoriatic arthritis; **PtAAP:** Patient's Assessment of Arthritis Pain; **Q2W:** every two weeks; **Q4W:** every four weeks; **REM:** remission; **SD:** standard deviation; **SF-36:** Short-Form 36-item Health Survey; **SJC:** swollen joint count; **TJC:** tender joint count; **TNFi-IR:** intolerance or inadequate response to tumor necrosis factor inhibitors; **VAS:** visual analog scale; **VLDA:** very low disease activity; **WCI:** worst-category imputation.

Institutions: [1]Swedish Medical Center/Providence St. Joseph Health and University of Washington, Seattle, WA, USA; [2]Nuffield Department of Orthopaedics, Rheumatology and Musculoskeletal Diseases, University of Oxford and Oxford Biomedical Research Centre, Oxford University Hospitals NHS Trust, Oxford, UK; [3]Amsterdam Rheumatology & Clinical Immunology Center, Amsterdam and Zuyderland MC, Heerlen, The Netherlands; [4]College of Medical Veterinary and Life Sciences, University of Glasgow, Glasgow, UK; [5]Department of Medicine, University of Rochester, Rochester, NY, USA; [6]Department of Rheumatology and Nephrology, Hokkaido University Hospital, Sapporo, Japan; [7]Division of Rheumatology, University Hospital and Fraunhofer Institute for Translational Medicine & Pharmacology ITMP, Fraunhofer Cluster of Excellence Immune-Mediated Diseases CIMD, Frankfurt, Goethe University, Frankfurt am Main, Germany; [8]Schroeder Arthritis Institute, Krembil Research Institute, University Health Network, Institute of Medical Science, University of Toronto, Ontario, Canada; [9]Sorbonne Université, INSERM, Institut Pierre Louis d'Epidémiologie et de Santé Publique, Paris, France; [10]Pitié-Salpêtrière hospital, AP-HP, Rheumatology Department, Paris, France; [11]School of Medicine, Griffith University, Brisbane, Australia; [12]UCB Pharma, Slough, UK; [13]UCB Pharma, Morrisville, NC, USA; [14]Department of Dermatology, The Icahn School of Medicine at Mount Sinai, New York, NY, USA.

References: [1]Gladman DD. Rheum Dis Clin North Am 2015;41(4):569–79; [2]McInnes IB. Ann Rheum Dis 2022;81:206–7; [3]Merola JF. Ann Rheum Dis 2022;81:167–9.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **PJM, LCC, RL, IBM, CTR, TA, FB, DDG, LG, PN, BI, DA, RB, JC, ARP, ABG;** Drafting of the publication, or revising it critically for important intellectual content: **PJM, LCC, RL, IBM, CTR, TA, FB, DDG, LG, PN, BI, DA, RB, JC, ARP, ABG;** Final approval of the publication: **PJM, LCC, RL, IBM, CTR, TA, FB, DDG, LG, PN, BI, DA, RB, JC, ARP, ABG.** **Author Disclosures:** **PJM:** Research grants from AbbVie, Amgen, BMS, Eli Lilly, Gilead, Janssen, Novartis, Pfizer, Sun Pharma, and UCB Pharma; consultancy fees from AbbVie, Acelyrin, Aclaris, Amgen, BMS, Boehringer Ingelheim, Eli Lilly, Galapagos, Gilead, GSK, Janssen, Moonlake, Novartis, Pfizer, Sun Pharma, and UCB Pharma; speakers' bureau from AbbVie, Amgen, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma. **LCC:** Grants/research support from AbbVie, Amgen, Celgene, Eli Lilly, Gilead, Janssen, Novartis, Pfizer, and UCB Pharma; Paid consultant for AbbVie, Amgen, BMS, Boehringer Ingelheim, Celgene, Domain, Eli Lilly, Gilead, Galapagos, Janssen, Moonlake, Novartis, Pfizer, and UCB Pharma; Paid as a speaker for AbbVie, Amgen, Biogen, Celgene, Eli Lilly, Galapagos, Gilead, GSK, Janssen, Medac, Novartis, Pfizer, and UCB Pharma. **RL:** Consultancy fees from AbbVie, AstraZeneca, BMS, Eli Lilly, Novartis, Pfizer, and UCB Pharma; research grants from AbbVie, Pfizer, Novartis, and UCB Pharma; owner of Rheumatology Consultancy BV, an AMS company under Dutch law. **IBM:** Consulting fees and honoraria from AbbVie, AstraZeneca, BMS, Boehringer Ingelheim, Cabaletta, Causeway Therapeutics, Celgene, Eli Lilly, Evelo, Janssen, Moonlake, Novartis, and UCB Pharma; Research support from Boehringer Ingelheim, BMS, Celgene, Janssen, Novartis, and UCB Pharma. **CTR:** Grants from AbbVie, Amgen, and UCB Pharma; consultant for AbbVie, Amgen, Eli Lilly, Gilead, Janssen, Novartis, Pfizer, and UCB Pharma. **TA:** Research grants from Alexion, Chugai Pharmaceutical, Daiichi Sankyo, Mitsubishi Tanabe Pharma, Otsuka Pharmaceutical, Pfizer, and Teijin Pharma; speaker fees from AbbVie, BMS, Chugai Pharmaceutical, Daiichi Sankyo, Eisai, Eli Lilly Japan K.K., Kyowa Kirin, Mitsubishi Tanabe Pharma, Novartis, Pfizer, Taiho Pharmaceutical, Takeda Pharmaceutical, and UCB Pharma; fees for consultancies from AbbVie, AstraZeneca, Medical & Biological Laboratories, Pfizer, Nippon Boehringer Ingelheim, Novartis, and Ono Pharmaceutical, outside the submitted work. **FB:** Consultant and/or speaker and/or investigator for AbbVie, Affibody, Amgen, Boehringer Ingelheim, Celgene, Chugai, Eli Lilly, Genzyme, GSK, Janssen, MSD, MoonLake, Novartis, Pfizer, Roche, Sandoz, and Sanofi. **DDG:** Grants from AbbVie, Amgen, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma; consulting fees from AbbVie, Amgen, BMS, Eli Lilly, Galapagos, Gilead, Janssen, Novartis, Pfizer, and UCB Pharma. **LG:** Research grants from Amgen, Eli Lilly, Galapagos, Pfizer, Sandoz, and UCB Pharma; consulting fees from AbbVie, Amgen, BMS, Celltrion, Eli Lilly, Galapagos, Gilead, GSK, Janssen, Novartis, Pfizer, and UCB Pharma. **PN:** Grants for research, clinical trials and honoraria for advice and lectures on behalf of AbbVie, BMS, Boehringer Ingelheim, Eli Lilly, Gilead/Galapagos, GSK, Janssen, Novartis, Pfizer, Samsung, Sanofi, and UCB Pharma. **BI:** Employee of UCB Pharma and shareholder of AbbVie, GSK and UCB Pharma. **DA, RB, JC:** Employees and shareholders of UCB Pharma. **ARP:** Employee of UCB Pharma, stockholder of GSK and UCB Pharma. **ABG:** Honoraria as an advisory board member, non-promotional speaker or consultant for Amgen, AnaptysBio, Avotres Therapeutics, BMS, Boehringer Ingelheim, Dermavant, Eli Lilly, Janssen, Novartis, Pfizer, Sanofi, Sun Pharma, UCB Pharma, and Xbiotech (stock options for an RA project); research/educational grants from AnaptysBio, BMS, Janssen, Novartis, Ortho Dermatologics, Sun Pharma, and UCB Pharma; all funds go to the Icahn School of Medicine at Mount Sinai. **Acknowledgments:** We would like to thank the patients and their caregivers in addition to the investigators and their teams who contributed to these studies. The authors acknowledge Heather Edens, PhD, UCB Pharma, Smyrna, GA, USA for publication coordination, Laura Mawdsley, MSc, and Luke Green, PhD, Costello Medical, Cambridge, UK for medical writing and editorial assistance, and the Costello Medical Design Team for design support. These studies were funded by UCB Pharma. All costs associated with development of this poster were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit the website below.

Website: UCBposters.com/ACR2022; Poster ID: 2117

Link expiration: 26 February 2023