Bimekizumab Treatment Results in Improvements in Fatigue and Pain in Biologic DMARD-Naïve or TNFi-IR Patients with Active Psoriatic Arthritis: Pooled 16-Week Results from Two Phase 3 Randomized, Placebo-Controlled Studies

M. E. Husni,[1] P. J. Mease,[2] J. F. Merola,[3] F. Behrens,[4] E. G. Favalli,[5] D. McGonagle,[6] W. Tillett,[7,8] S. Tsuji,[9] B. Ink,[10] D. Assudani,[10] R. Bajracharya,[10] J. Coarse,[11] J. Lambert,[12] L. Gossec[13]

CONTENT PROVIDED FOR SHAREHOLDERS, INVESTORS AND OTHER CAPITAL MARKET PARTICIPANTS ONLY

Presented at ACR Convergence 2022 | Philadelphia, Pennsylvania, USA | November 10–14

2119

Objective

To report the impact of bimekizumab (BKZ) treatment on patient-reported symptoms of fatigue and pain in patients with active psoriatic arthritis (PsA) who are biologic-naïve or had intolerance or inadequate response to TNFi (TNFi-IR).

Background

- Fatigue and pain place a considerable burden of disease on patients with PsA.[1]
- BKZ, a monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A, has demonstrated improvements in patient-reported symptoms up to three years in the phase 2b study BE ACTIVE.[2]

Methods

- BE OPTIMAL (NCT03895203) and BE COMPLETE (NCT03896581) were phase 3 studies assessing BKZ in patients with active PsA who are biologic DMARD (bDMARD)-naïve or TNFi-IR, respectively (**Figure 1**).
- The primary endpoint in both studies was the proportion of patients with ≥50% improvement in American College of Rheumatology criteria response at Week 16.
- We present pooled and individual study data to Week 16 for BKZ and placebo (PBO) treatment arms for:
 - Functional Assessment of Chronic Illness Therapy-Fatigue subscale (FACIT-Fatigue);
 - Proportion of patients reaching the minimum clinically important difference (MCID) of ≥4-point improvement from baseline.
 - Change from baseline.
 - Patient's Assessment of Arthritis Pain
 - Clinically important improvements of ≥30/50/70% from baseline.[3]
 - Change from baseline.
- Non-responder imputation (NRI) and multiple imputation (MI) were used for missing binary and continuous data, respectively.

Results

- A total of 1,073/1,112 (96.5%) patients randomized to BKZ or PBO completed Week 16.
- Baseline characteristics were generally comparable across treatment arms and studies (**Table 1**).
- At Week 16 (pooled and individual analyses), BKZ demonstrated numerically greater and clinically meaningful improvements in patient-reported fatigue (FACIT-Fatigue MCID and change from baseline) compared with PBO (**Figure 2**).
- A higher proportion of BKZ-treated patients achieved greater improvements in patient-reported pain (≥30/50/70% improvements and change from baseline in Patient's Assessment of Arthritis Pain) compared with PBO in both the pooled and individual study populations (**Figure 3**).
- BKZ treatment was associated with a rapid onset of response with improvements in fatigue and pain as early as Week 4 (**Figures 2 and 3**).

Summary

1,073/1,112 (96.5%) patients with active psoriatic arthritis treated with **bimekizumab 160 mg Q4W** or **placebo** in **BE OPTIMAL** or **BE COMPLETE** completed the placebo-controlled phase at Week 16

Greater proportions of bimekizumab-treated patients achieved clinically meaningful improvements in **patient-reported symptoms** at Week 16:

FACIT-Fatigue MCID

53.1%	Bimekizumab
36.3%	Placebo

≥50% improvement in Patient's Assessment of Arthritis Pain

49.6%	Bimekizumab
17.4%	Placebo

Results suggest that **bimekizumab treatment** leads to **improvements in fatigue and pain** irrespective of prior biologic treatment

Figure 1 BE OPTIMAL and BE COMPLETE study designs

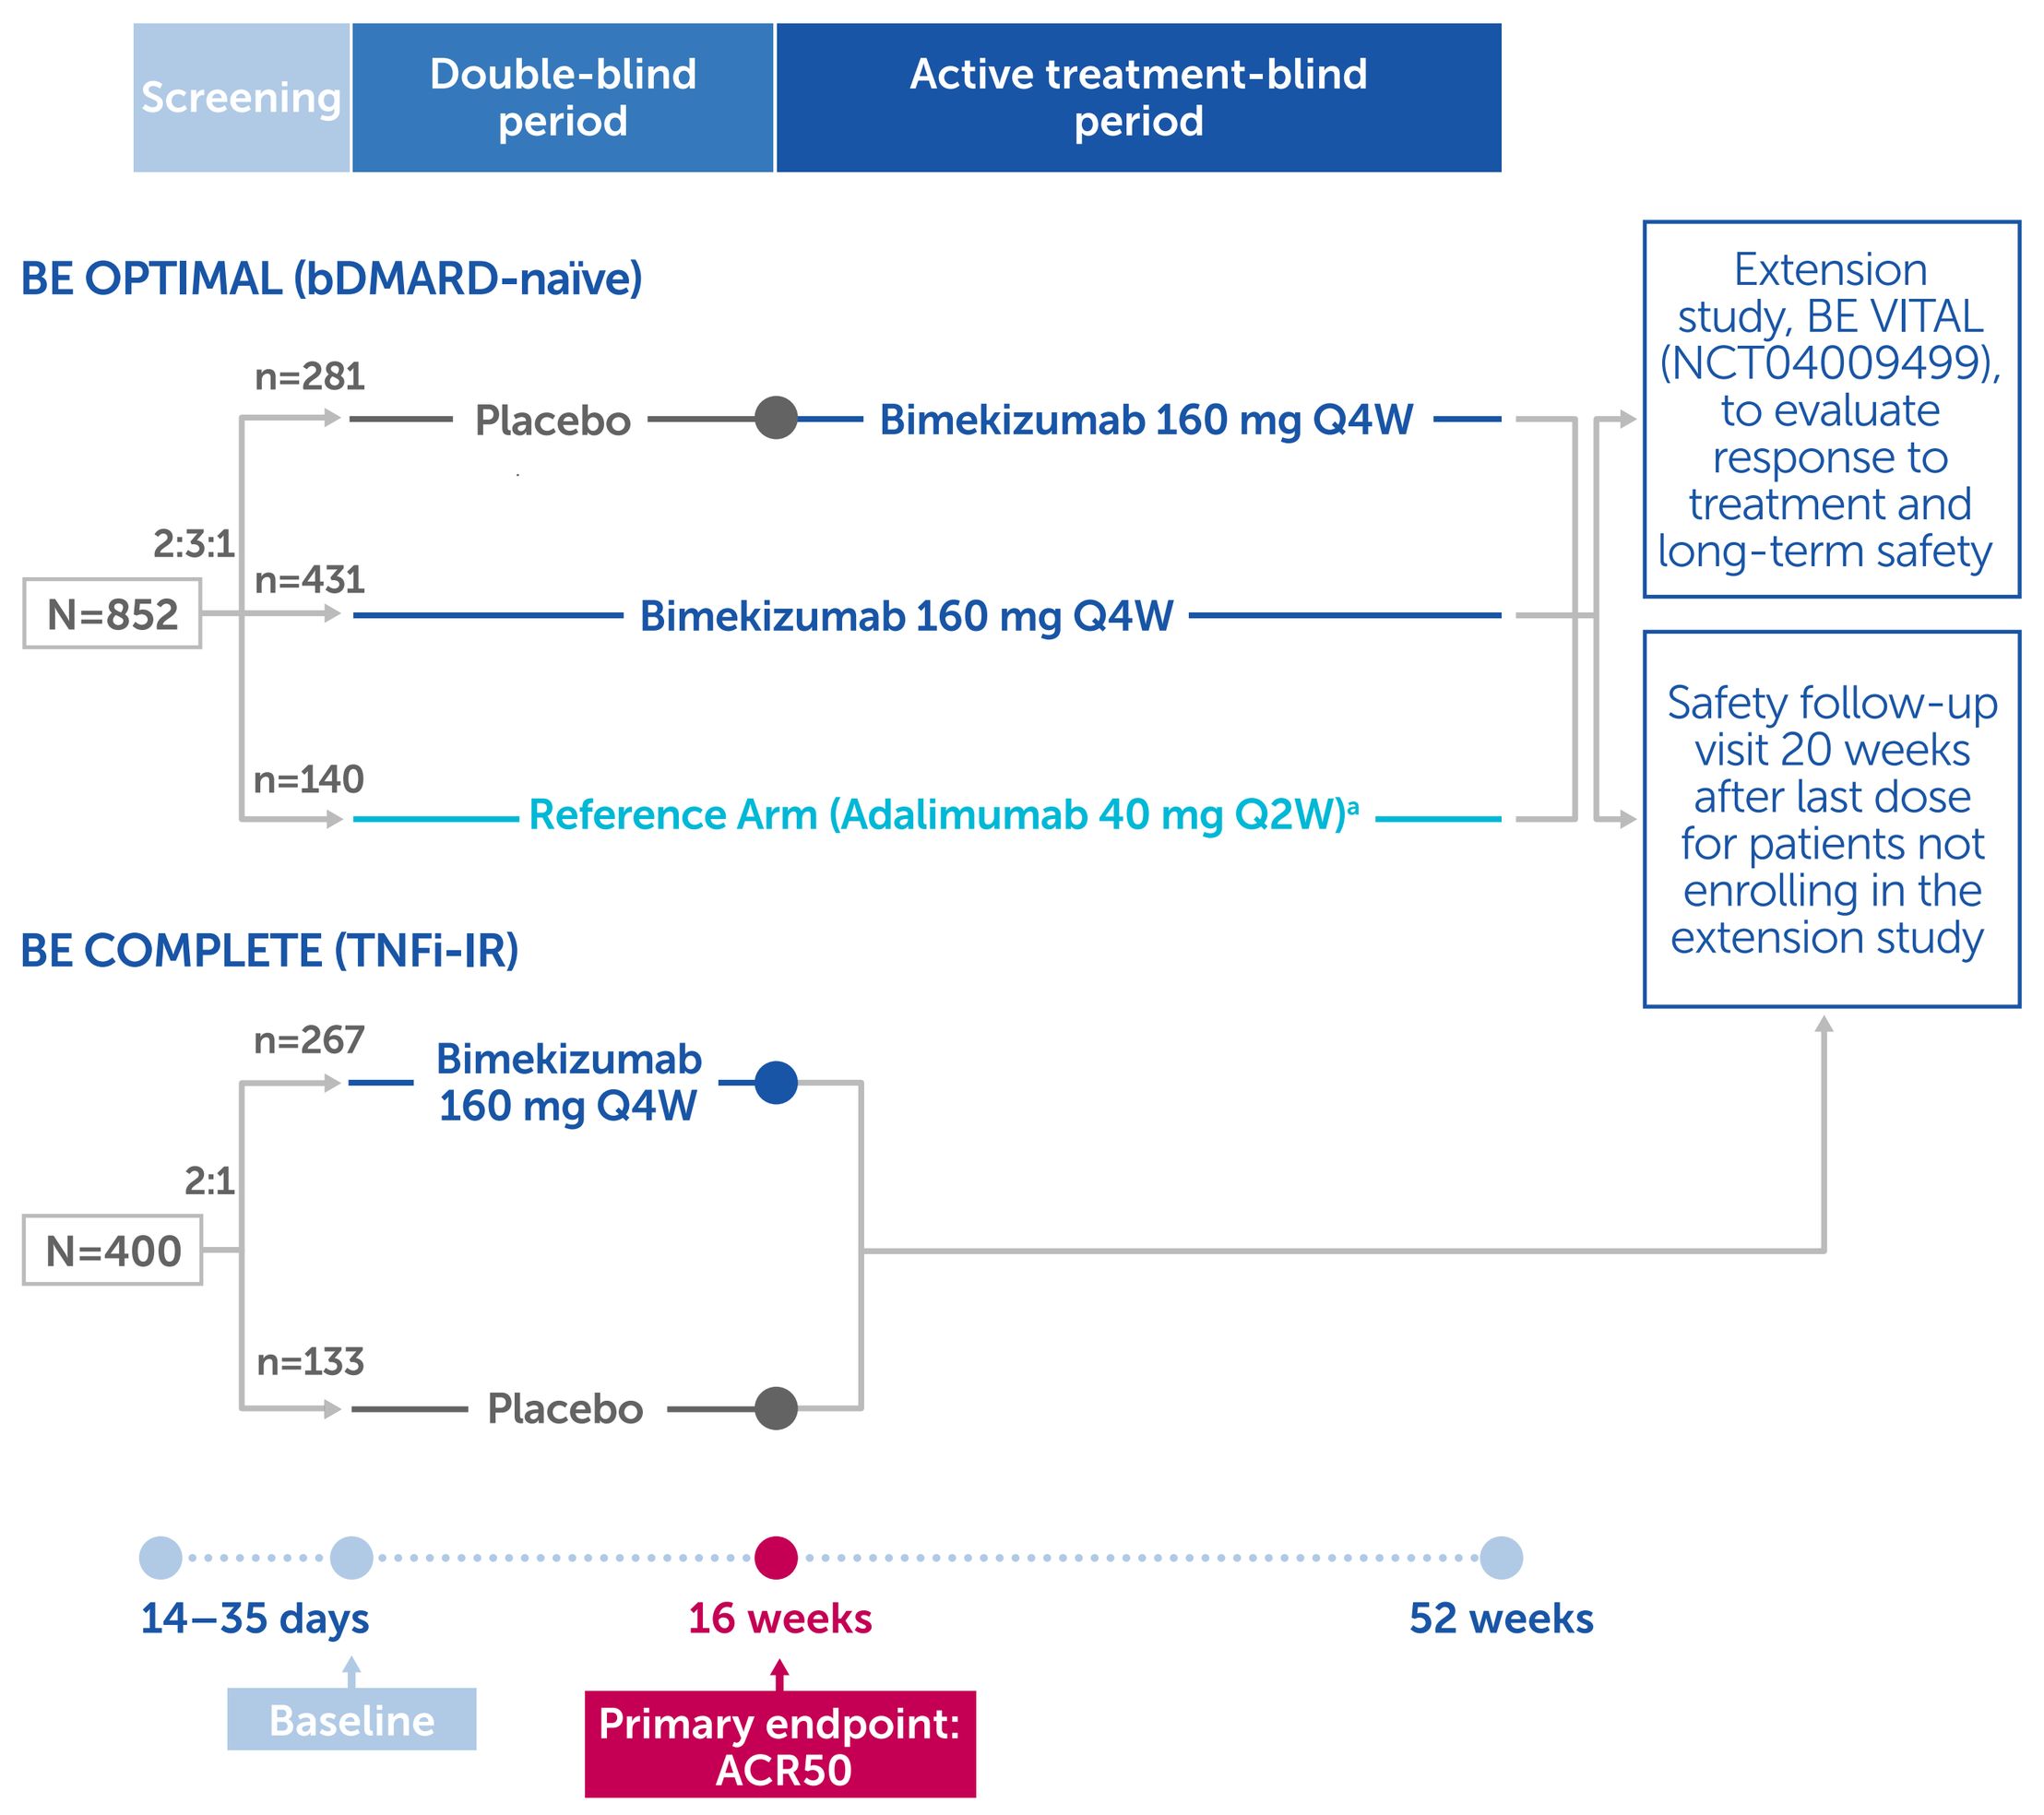
Screening
Double-blind period
Active treatment-blind period
BE OPTIMAL (bDMARD-naïve)
N=852
2:3:1
n=281
Placebo
Bimekizumab 160 mg Q4W
n=431
Bimekizumab 160 mg Q4W
n=140
Reference Arm (Adalimumab 40 mg Q2W)a
Extension study, BE VITAL (NCT04009499), to evaluate response to treatment and long-term safety
Safety follow-up visit 20 weeks after last dose for patients not enrolling in the extension study
BE COMPLETE (TNFi-IR)
N=400
2:1
n=267
Bimekizumab 160 mg Q4W
n=133
Placebo
14–35 days
Baseline
16 weeks
Primary endpoint: ACR50
52 weeks

Study designs that made up the pooled population, BE OPTIMAL (NCT03895203) and BE COMPLETE (NCT03896581). [a]Adalimumab 40 mg Q2W served as an active reference arm. BE OPTIMAL was not powered for comparisons of adalimumab to bimekizumab or adalimumab to placebo. Adalimumab 40 mg Q2W data are not shown.

Figure 2 Improvements in patient-reported fatigue over time (Weeks 0–16)

A) FACIT-Fatigue MCID responders (NRI)

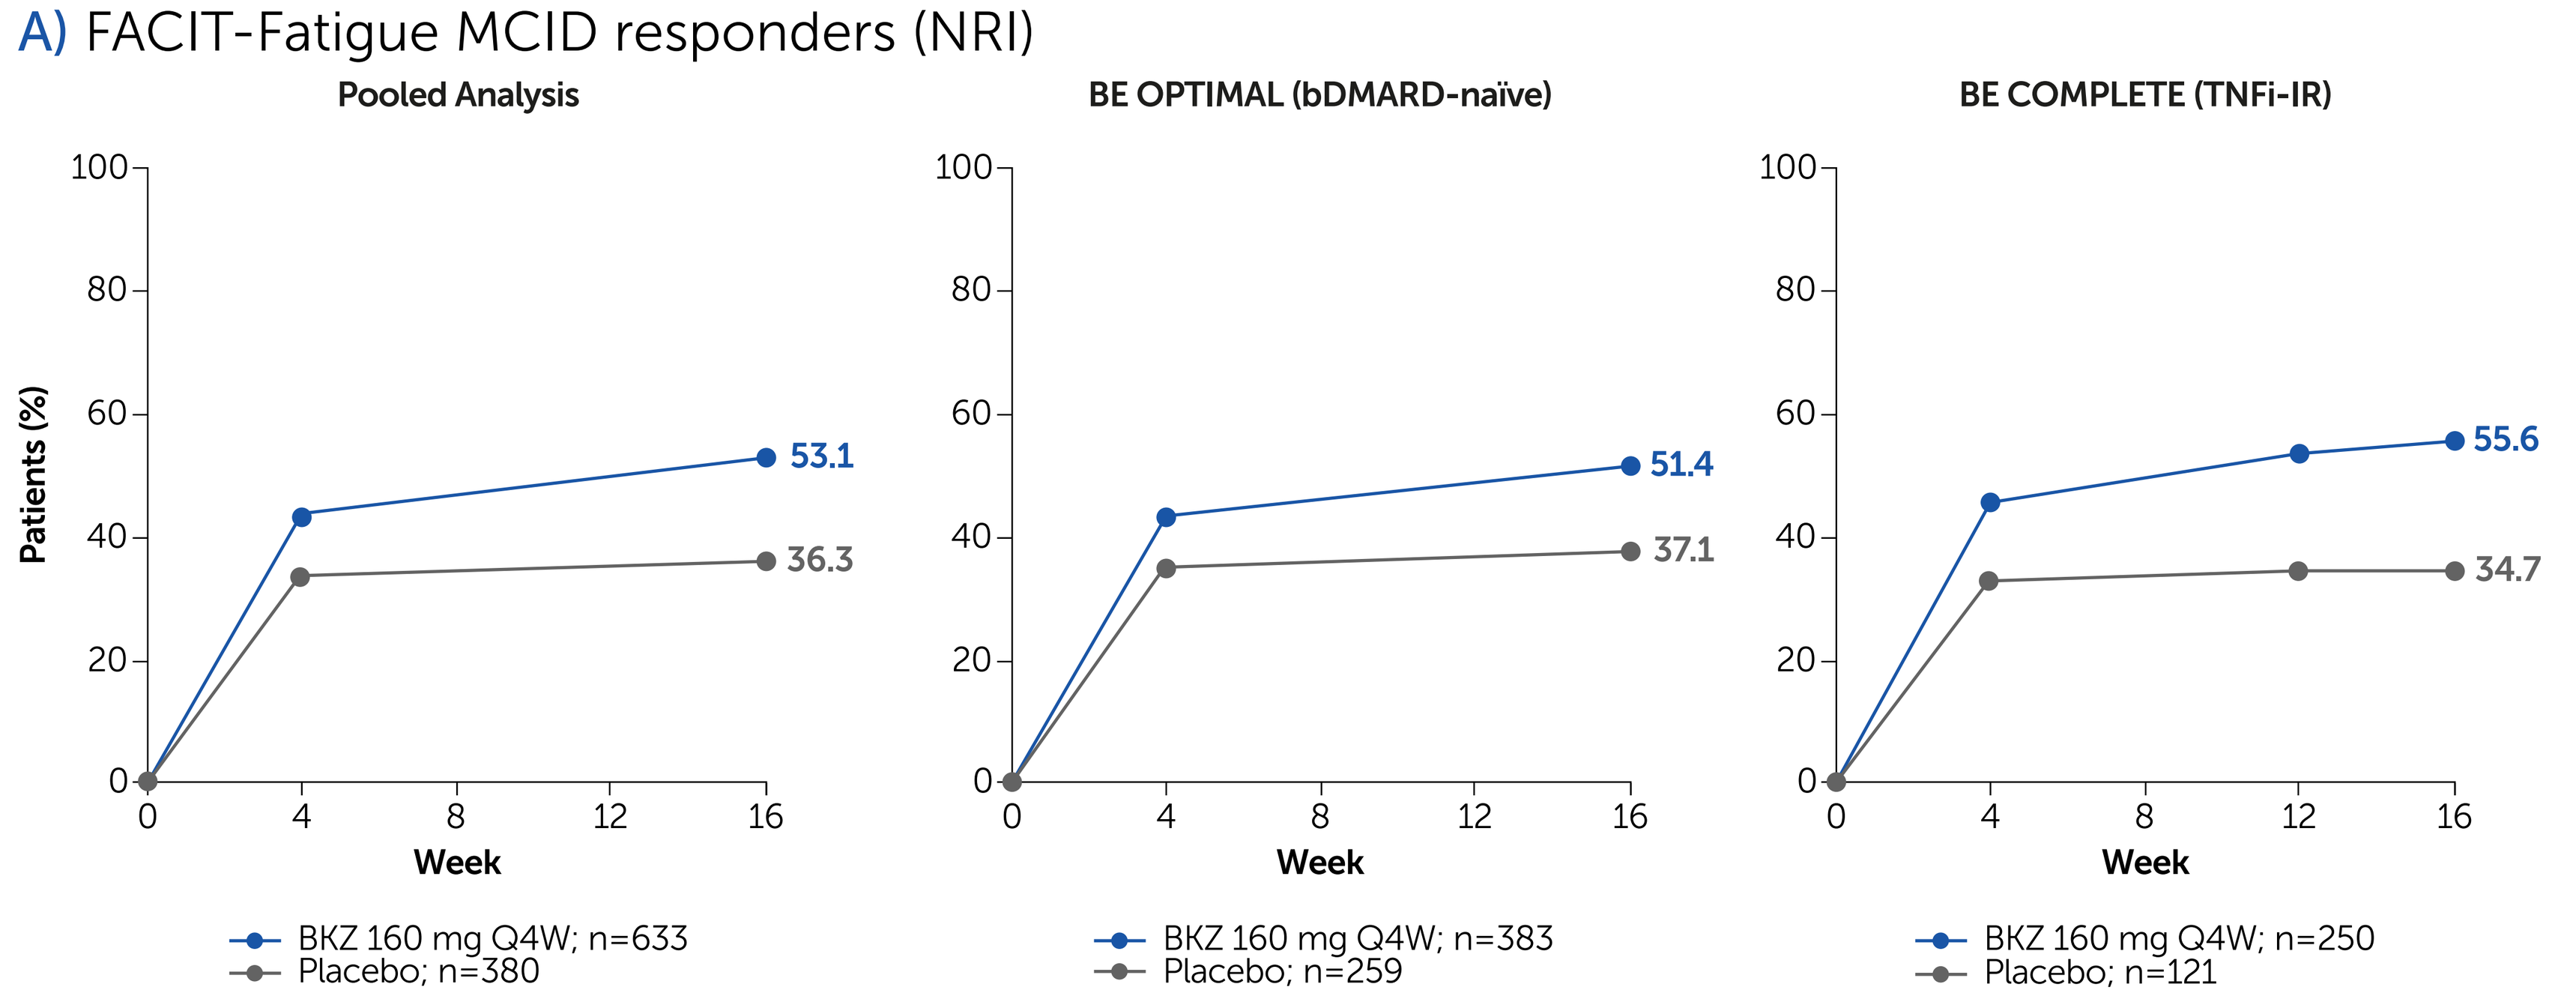
Pooled Analysis
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
Patients (%)
Week
53.1
36.3
51.4
37.1
55.6
34.7
BKZ 160 mg Q4W; n=633
Placebo; n=380
BKZ 160 mg Q4W; n=383
Placebo; n=259
BKZ 160 mg Q4W; n=250
Placebo; n=121

B) FACIT-Fatigue change from baseline (MI)

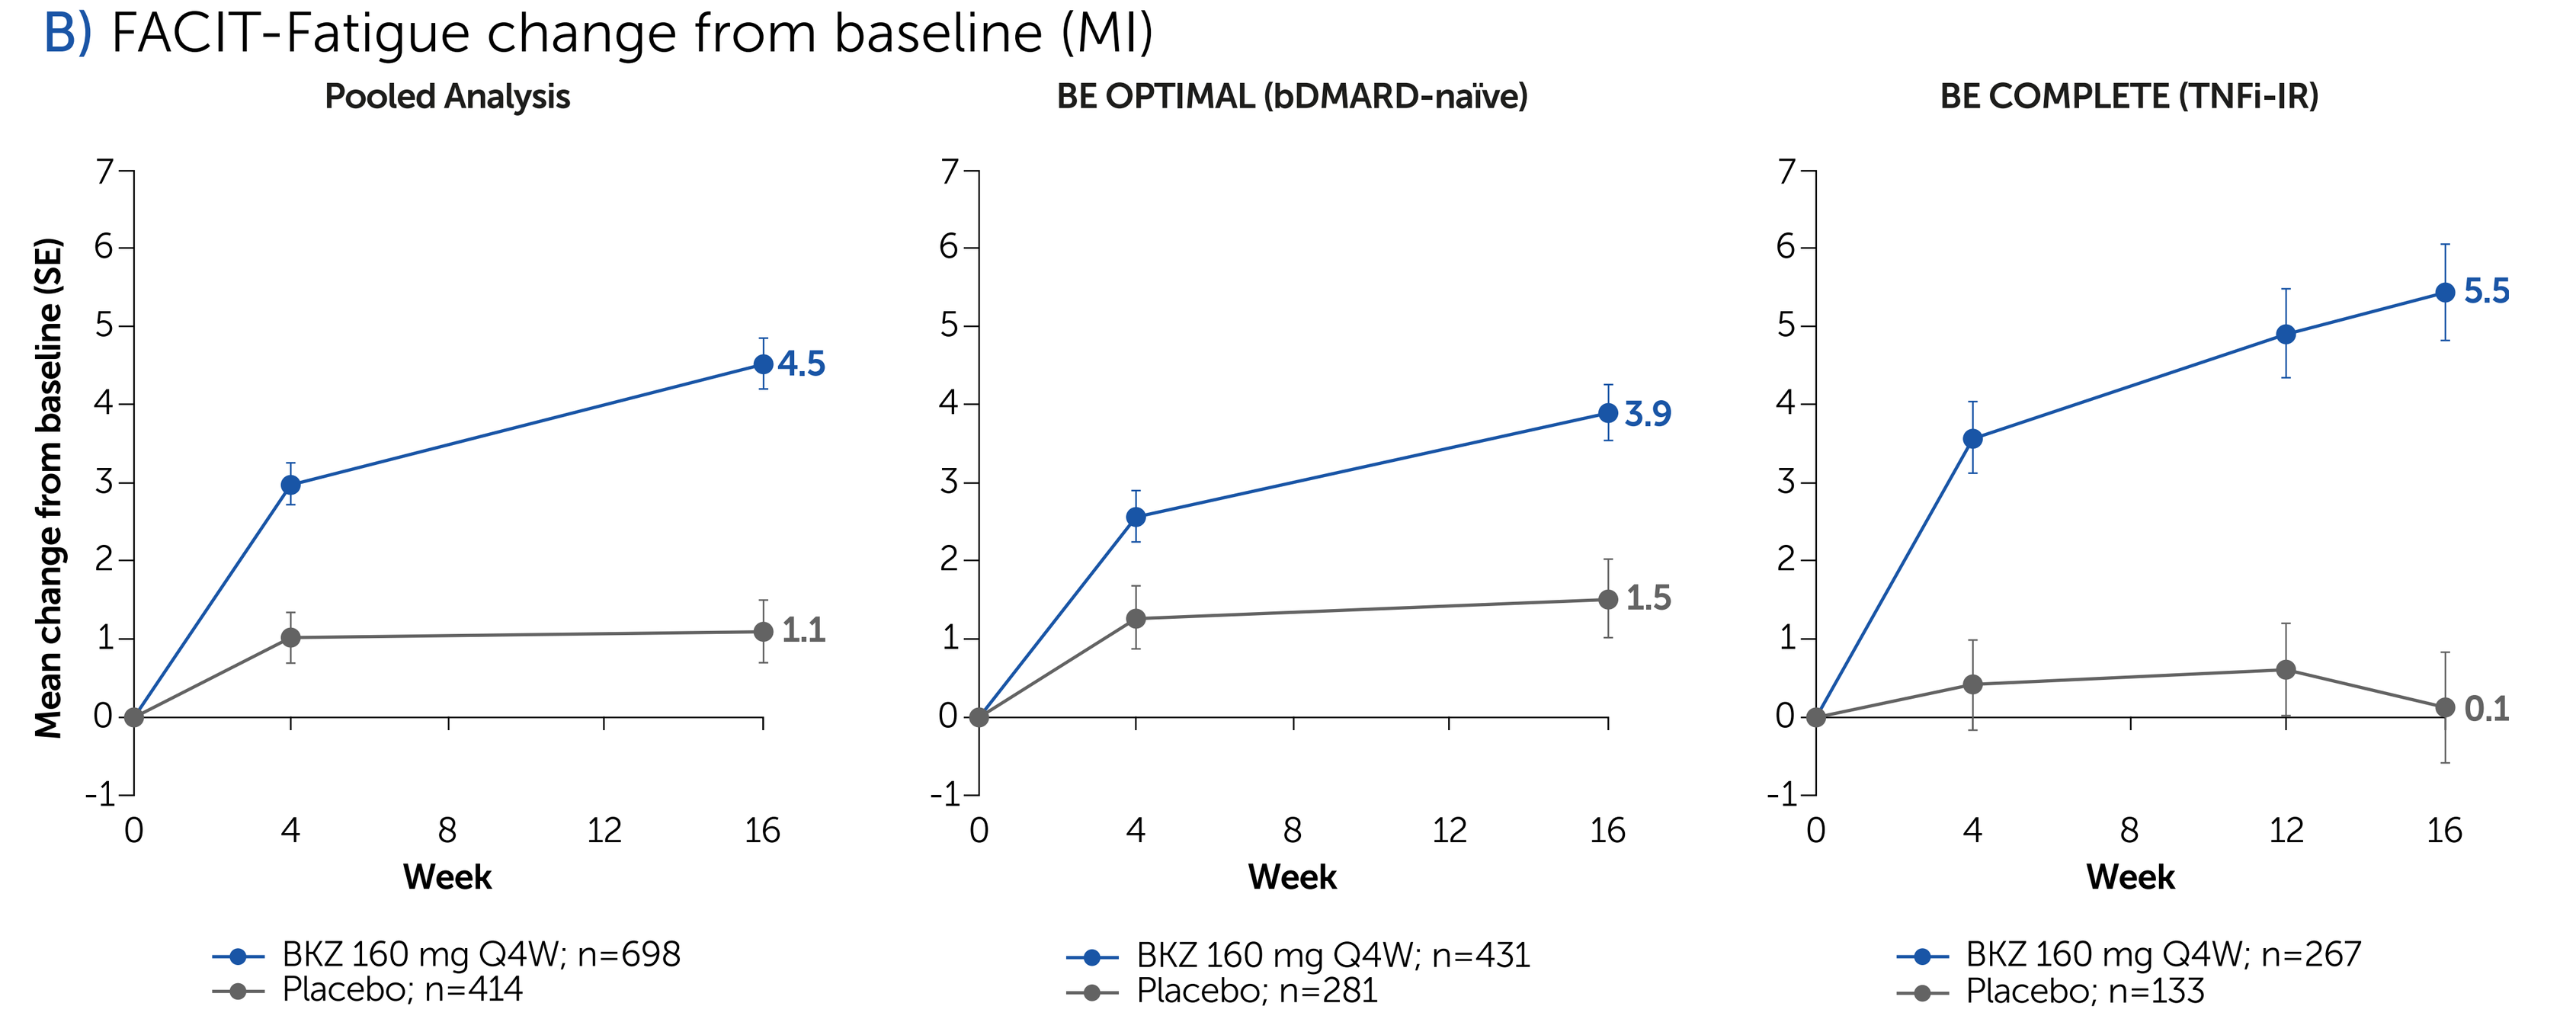
Pooled Analysis
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
Mean change from baseline (SE)
Week
4.5
1.1
3.9
1.5
5.5
0.1
BKZ 160 mg Q4W; n=698
Placebo; n=414
BKZ 160 mg Q4W; n=431
Placebo; n=281
BKZ 160 mg Q4W; n=267
Placebo; n=133

Randomized set. FACIT-Fatigue score ranges from 0 to 52, with 52 being the best possible score. FACIT-Fatigue MCID only measured in patients with FACIT-Fatigue subscale score ≤48 at baseline. FACIT-Fatigue MCID defined as increase in score from baseline ≥4.

Figure 3 Improvements in patient-reported pain over time (Weeks 0–16)

A) Improvements of ≥30/50/70% in Patient's Assessment of Arthritis Pain (NRI)

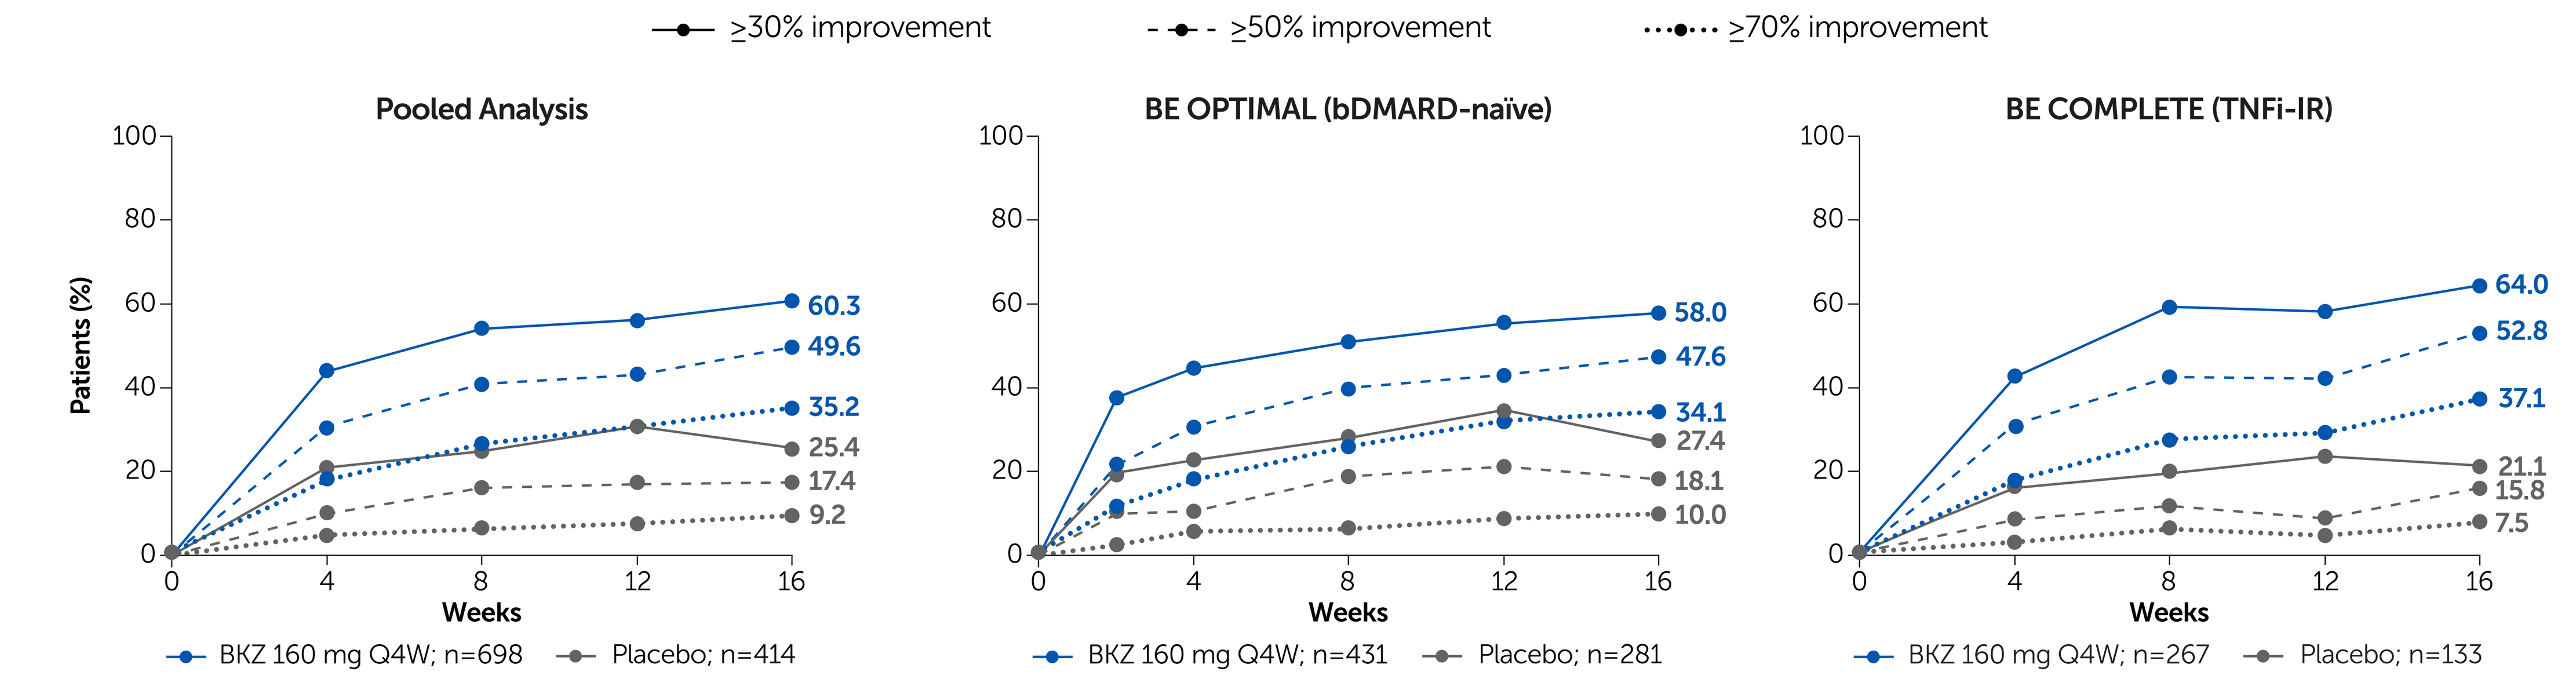
≥30% improvement
≥50% improvement
≥70% improvement
Pooled Analysis
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
Patients (%)
Weeks
60.3
49.6
35.2
25.4
17.4
9.2
58.0
47.6
34.1
27.4
18.1
10.0
64.0
52.8
37.1
21.1
15.8
7.5
BKZ 160 mg Q4W; n=698
Placebo; n=414
BKZ 160 mg Q4W; n=431
Placebo; n=281
BKZ 160 mg Q4W; n=267
Placebo; n=133

B) Patient's Assessment of Arthritis Pain change from baseline (MI)

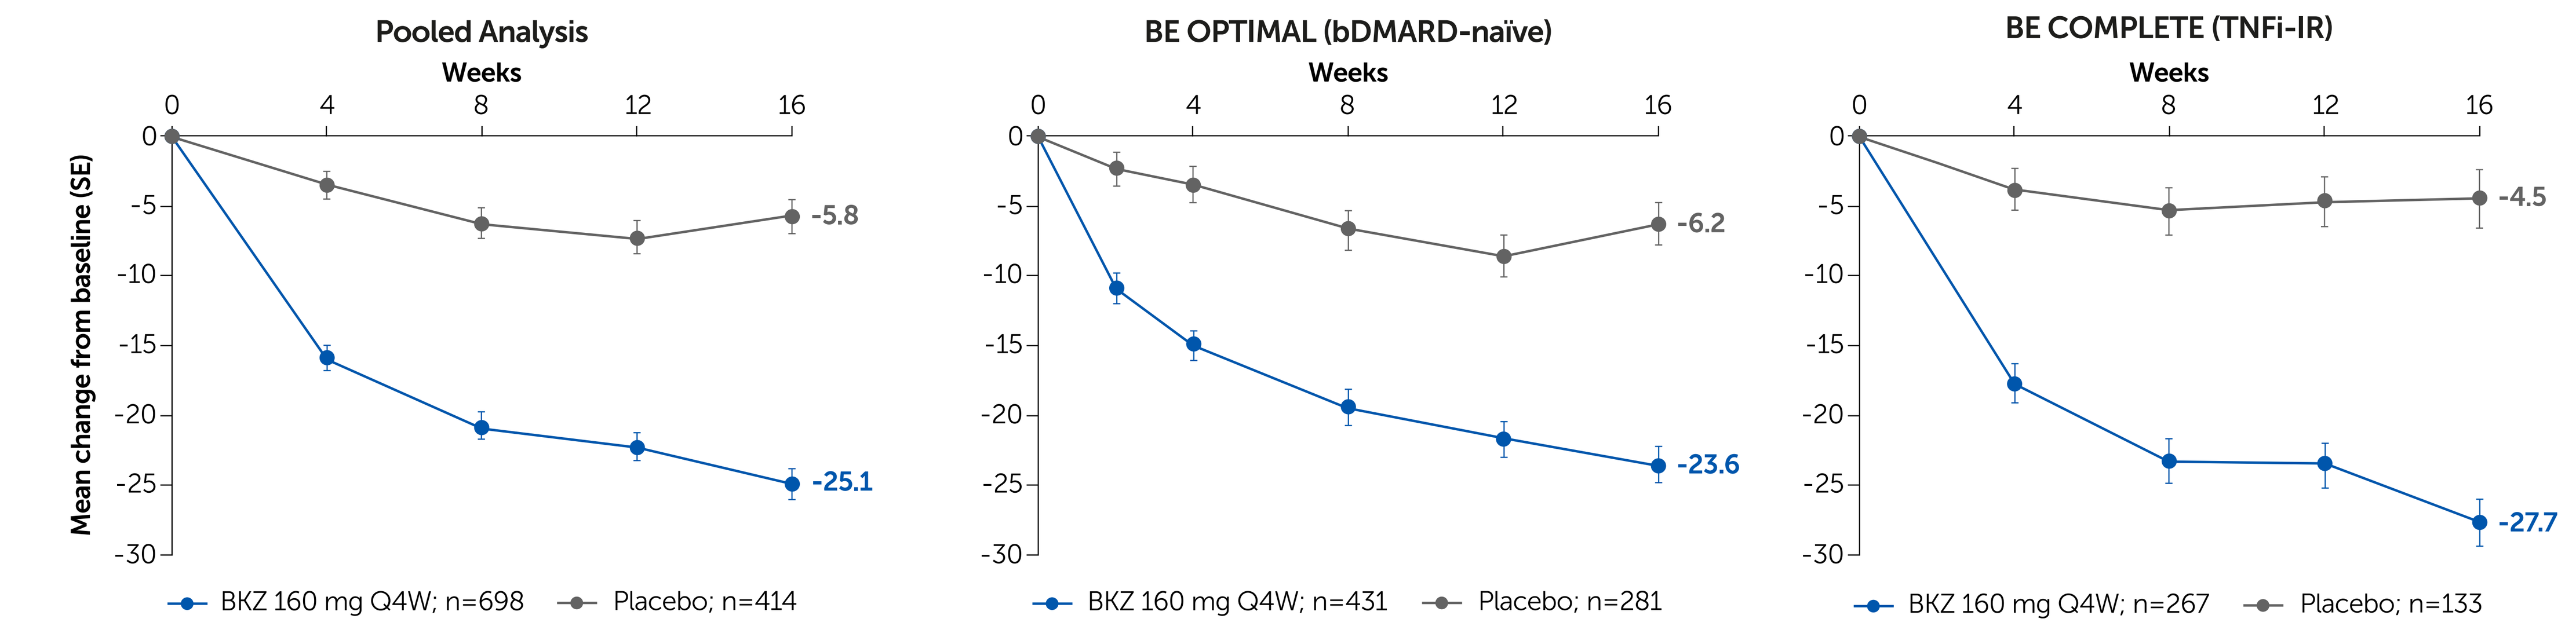
Pooled Analysis
BE OPTIMAL (bDMARD-naïve)
BE COMPLETE (TNFi-IR)
Weeks
Mean change from baseline (SE)
-5.8
-25.1
-6.2
-23.6
-4.5
-27.7
BKZ 160 mg Q4W; n=698
Placebo; n=414
BKZ 160 mg Q4W; n=431
Placebo; n=281
BKZ 160 mg Q4W; n=267
Placebo; n=133

Randomized set. Patient's Assessment of Arthritis Pain 100mm VAS ranges from 0 to 100, with 0 representing "no pain" and 100 "most severe pain". ≥30/50/70% represent clinically important improvements in patient-reported pain.[3]

Table 1 Baseline characteristics

	Pooled Analysis (bDMARD-naïve + TNFi-IR)		BE OPTIMAL (bDMARD-naïve)		BE COMPLETE (TNFi-IR)	
	PBO (n=414)	BKZ 160 mg Q4W (n=698)	PBO (n=281)	BKZ 160 mg Q4W (n=431)	PBO (n=133)	BKZ 160 mg Q4W (n=267)
Age, mean (SD)	49.5 (12.2)	49.1 (12.5)	48.7 (11.7)	48.5 (12.6)	51.3 (12.9)	50.1 (12.4)
Male, n (%)	187 (45.2)	331 (47.4)	127 (45.2)	201 (46.6)	60 (45.1)	130 (48.7)
BMI, kg/m², mean (SD)	29.4 (5.9)	29.6 (6.7)	29.6 (6.1)	29.2 (6.8)	29.0 (5.4)	30.1 (6.5)
BSA affected by psoriasis ≥3%, n (%)	228 (55.0)	393 (56.3)	140 (49.8)	217 (50.3)	88 (66.2)	176 (65.9)
Time since PsA diagnosis (years), mean (SD)	6.8 (7.3)[a]	7.4 (8.6)[b]	5.6 (6.5)[c]	6.0 (7.3)[d]	9.2 (8.1)[e]	9.6 (9.9)[f]
PASI,[g] mean (SD)	8.1 (6.0)	9.1 (8.0)	7.9 (5.6)	8.2 (6.8)	8.5 (6.6)	10.1 (9.1)
HAQ-DI, mean (SD)	0.9 (0.6)	0.9 (0.6)[h]	0.9 (0.6)	0.8 (0.6)[i]	1.0 (0.7)	1.0 (0.6)
hs-CRP ≥6 mg/L, n (%)	180 (43.5)	276 (39.5)	121 (43.1)	158 (36.7)	59 (44.4)	118 (44.2)
FACIT-Fatigue, mean (SD)	36.1 (10.1)	36.8 (10.0)[h]	36.0 (10.2)	37.7 (9.6)[i]	36.3 (9.9)	35.3 (10.5)
Patient's Assessment of Arthritis Pain, mean (SD)	58.4 (23.8)	55.4 (24.3)[h]	56.8 (23.2)	53.6 (24.3)[i]	61.7 (24.6)	58.3 (24.2)

[a]n=411. [b]n=689. [c]n=279. [d]n=423. [e]n=132. [f]n=266. [g]For patients with psoriasis involving at ≥3% BSA at baseline. [h]n=697. [i]n=430.

Conclusions

Bimekizumab treatment resulted in greater improvements compared with placebo in patient-reported symptoms of fatigue and pain. Improvements were consistent between studies, with similar magnitude of response observed in bDMARD-naïve and TNFi-IR patients.

bDMARD: biologic disease-modifying antirheumatic drug; **BKZ:** bimekizumab; **BMI:** body mass index; **BSA:** body surface area; **FACIT-Fatigue:** Functional Assessment of Chronic Illness Therapy-Fatigue; **HAQ-DI:** Health Assessment Questionnaire-Disability Index; **hs-CRP:** high sensitivity C-reactive protein; **IL:** interleukin; **MCID:** minimum clinically important difference; **MI:** multiple imputation; **NRI:** non-responder imputation; **PASI:** Psoriasis Area and Severity Index; **PBO:** placebo; **PsA:** psoriatic arthritis; **Q2W:** every two weeks; **Q4W:** every four weeks; **SD:** standard deviation; **TNFi-IR:** intolerance/inadequate response to tumor necrosis factor inhibitor; **VAS:** visual analog scale.

Institutions: [1]Orthopedic and Rheumatologic Institute, Cleveland Clinic Lerner College of Medicine, Case Western Reserve University, Cleveland, OH, USA; [2]Swedish Medical Center and Providence St. Joseph Health, University of Washington, Seattle, Washington, USA; [3]Harvard Medical School, Brigham and Women's Hospital, Boston, MA, USA; [4]Division of Rheumatology, University Hospital and Fraunhofer Institute for Translational Medicine & Pharmacology ITMP, Fraunhofer Cluster of Excellence Immune-Mediated Diseases CIMD, Goethe University, Frankfurt am Main, Germany; [5]Department of Rheumatology, ASST Gaetano Pini-CTO, University of Milan, Milan, Italy; [6]Academic Unit for the Musculoskeletal Diseases, Leeds Teaching Hospitals NHS Trust, Leeds, UK; [7]Royal National Hospital of Rheumatic Diseases, Bath, UK; [8]Department of Life Sciences, Centre for Therapeutic Innovation, University of Bath, Bath, UK; [9]Department of Orthopaedics and Rheumatology, Nippon Life Hospital, Osaka, Japan; [10]UCB Pharma, Slough, UK; [11]UCB Pharma, Morrisville, NC, USA; [12]UCB Pharma, Colombes, France; [13]Sorbonne Université, INSERM, Institut Pierre Louis d'Epidémiologie et de Santé Publique, Paris, France, and AP-HP, Pitié-Salpêtrière Hospital, Rheumatology Department, Paris, France.

References: [1]Ogdie A. RMD Open 2020;6(3):e001321; [2]Mease PJ. Rheumatology (Oxford) 2022;keac353; [3]Dworkin RH. J Pain 2008;9(2):105–21.

Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **MEH, PJM, JFM, FB, EGF, DM, WT, ST, BI, DA, RB, JC, JL, LG;** Drafting of the publication, or revising it critically for important intellectual content: **MEH, PJM, JFM, FB, EGF, DM, WT, ST, BI, DA, RB, JC, JL, LG;** Final approval of the publication: **MEH, PJM, JFM, FB, EGF, DM, WT, ST, BI, DA, RB, JC, JL, LG**. **Author Disclosures: MEH:** Advisory board member and consultant for AbbVie, Amgen, BMS, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma. **PJM:** Research grants from AbbVie, Amgen, BMS, Eli Lilly, Gilead, Janssen, Novartis, Pfizer, Sun Pharma, and UCB Pharma; consultancy fees from AbbVie, Acelyrin, Aclaris, Amgen, BMS, Boehringer Ingelheim, Eli Lilly, Galapagos, Gilead, GSK, Janssen, MoonLake, Novartis, Pfizer, Sun Pharma, and UCB Pharma; speakers bureau from AbbVie, Amgen, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma. **JFM:** Consultant and/or investigator for AbbVie, Amgen, Biogen, BMS, Dermavant, Eli Lilly, Janssen, LEO Pharma, Novartis, Pfizer, Sanofi, Sun Pharma, and UCB Pharma. **FB:** Consultant and/or speaker and/or investigator for AbbVie, Affibody, Amgen, Boehringer Ingelheim, Celgene, Chugai, Eli Lilly, Genzyme, GSK, Janssen, MoonLake, MSD, Novartis, Pfizer, Roche, Sandoz, and Sanofi. **EGF:** Consultancy/speaker fees from AbbVie, BMS, Celltrion, Eli Lilly, Galapagos, Janssen, MSD, Novartis, Pfizer, and UCB Pharma. **DM:** Grants/research support from AbbVie, Celgene, Janssen, Merck, Novartis, and Pfizer; consulting fees and honoraria from AbbVie, Celgene, Janssen, Merck, Novartis, Pfizer, and UCB Pharma; speakers bureau for AbbVie, Celgene, Janssen, Merck, Novartis, Pfizer, and UCB Pharma. **WT:** Research grants, consulting fees, speaking fees, and/or honoraria from AbbVie, Amgen, Celgene, Eli Lilly, GSK, Janssen, MSD, Novartis, Pfizer, and UCB Pharma. **ST:** Consulting and/or speaker fees from AbbVie, Eisai, Eli Lilly, Janssen, Kyowa Kirin, Novartis, and UCB Pharma. **BI:** Employee and stockholder of UCB Pharma; stockholder of AbbVie and GSK. **DA, RB, JC, JL:** Employees and stockholders of UCB Pharma. **LG:** Research grants from Amgen, Galapagos, Eli Lilly, Pfizer, Sandoz, and UCB Pharma; consulting fees from AbbVie, Amgen, BMS, Celltrion, Eli Lilly, Galapagos, Gilead, GSK, Janssen, Novartis, Pfizer, and UCB Pharma. **Acknowledgments:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to these studies. The authors acknowledge Heather Edens, PhD, UCB Pharma, for publication coordination, Jessica A. Buttress, BSc, Costello Medical, Cambridge, UK, for medical writing and editorial assistance, and the Costello Medical Design Team for design support. These studies were funded by UCB Pharma. All costs associated with development of this presentation were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit the website below.
Website: UCBposters.com/ACR2022
Poster ID: 2119
Link expiration: 26 February 2023