CONTENT PROVIDED FOR SHAREHOLDERS, INVESTORS AND OTHER CAPITAL MARKET PARTICIPANTS ONLY

A decentralized, prospective, observational study to collect real-world data from patients with myasthenia gravis using smartphones

Sandra Steyaert[1], Meelis Lootus[1], Chethan Sarabu[1], Zeenia Framroze[1], Harriet Dickinson[2], Emily Kunka[3], Jean-Christophe Steels[3], Nirav R. Shah[1], Francesca Rinaldo[1]

[1]Sharecare Inc. Palo Alto, CA, USA; [2]Global Real World Evidence, UCB Pharma, Slough, UK; [3]UCB Pharma, Brussels, Belgium

14th MGFA International Conference on Myasthenia and Related Disorders, 10–12 May 2022, Miami, FL, USA

P84

Introduction

- Effective treatment for rare diseases is often limited by:
 - The low prevalence of the condition
 - The variety of settings and specialties in which patients are treated leading to fragmented care
 - Potential heterogeneity in symptom presentation and clinical manifestations of the disease and how they change over time.
- The use of real-world data and digital phenotyping offer a promising solution to these challenges, allowing collection of multidimensional data that capture the unmet needs of patients.
- The ubiquity of smartphones and wearables allows data to be collected more frequently and in a decentralized manner, potentially
 - providing a more-complete picture of the lived experience of the disease
 - making participation in research more accessible and convenient for patients.
- Here we describe a 3-month prospective observational study in adults with myasthenia gravis (MG) using fully decentralized methods to assess the feasibility of participant-driven, real-world data collection from smartphones and wearable devices.

Study objectives

- The objectives of this study were to:
 - Collect self-reported outcomes and passively generated health data from patients with MG, covering exacerbation and non-exacerbation periods
 - Determine concordance between self-reported exacerbation status and MG-Activities of Daily Living (MG-ADL) scores
 - Characterize symptom involvement during exacerbations, and identify subtypes of exacerbations.

Methods

- The study was conducted using Smart Omix (Sharecare, Inc), a platform for decentralized real-world research that allows for multidimensional data collection directly from participant smartphones.
- Participants were recruited in a fully decentralized manner from across the United States, using social media channels to direct interested individuals to an online pre-screener. Approved participants joined the study by downloading the study app (available on iOS and Android) and completing a CFR part 11 compliant eConsent process.
- The inclusion criteria were:
 1) Documented diagnosis of MG (uploaded via app)
 2) Experienced ocular or bulbar symptoms
 3) Aged 18+
 4) US resident for the duration of the study
 5) Able to read, understand and write in English
 6) Own a smartphone that could support the research application.
- Onboarding:
 - Upload MG diagnosis documentation to app
 - Survey on demographic and MG disease characteristics, comorbid conditions and active medications.
- During the 3-month study:
 - Reported daily symptoms, symptom severity, and exacerbation status using a digital version of the MG-ADL assessment
 - Optional connections to contribute secondary, passive data streams, such as daily step count.
- Participants were compensated with a $250 Amazon gift card if they completed the study with a preset adherence of 60%. The study was reviewed and approved by a central institutional review board (Salus IRB), protocol number DOC-005-2020.

Results

Study population characteristics

- The study enrolled and onboarded 113 participants across 37 US states, between October 2020 and July 2021; 73% (n=82) completed the study.
- Age, gender and race distributions are shown in **Figures 1A** and **1B**. Participants were representative of clinically observed age and gender distributions for MG[1].
- The study population represented an MG phenotype with a high burden of exacerbations: the majority of patients reported experiencing multiple exacerbations per year (**Figure 1C**), with median MG-ADL scores of 5 (during periods with low symptom burden) and 14 (during periods of high symptom burden) reported in the onboarding survey. For participants who completed the study, the majority (76%) reported at least one active MG medication, with 28% reporting treatment for refractory disease (**Figure 1D**).
- The most frequently reported comorbid conditions were hypertension (n=26), depression (n=12), and type 2 diabetes (n=11). In total, the study collected 5101 self-reported daily check-ins for symptoms, symptom severity and exacerbation status from study participants.

Concordance between MG-ADL scores and self-reported exacerbation status

- Median MG-ADL scores during self-reported exacerbation and non-exacerbation periods were 5 (interquartile range 2–8, range 0–24) and 0 (interquartile range 0–0, range 0–15), respectively (**Figure 2A**). 45 participants (39.8%) reported MG exacerbations, with an average of 6.3 exacerbation days per participant over the 90-day study period. A significant association between average MG-ADL scores and exacerbation status was observed for this sub-cohort (Wilcoxon signed-rank p value <0.0001) (**Figure 2B**).

Concordance between daily step counts, MG-ADL score and exacerbation status

- Daily step count data were provided by 29 participants (26%). Overall participants with lower MG-ADL score took more steps (correlation coefficient r=−0.15, **Figure 3A**) and those patients who also reported an exacerbation (n=14) took fewer steps on exacerbation days than non-exacerbation days (Kruskal–Wallis p=0.03, **Figure 3B**).

Clusters of specific symptom signatures during exacerbations

- Unsupervised machine learning methods were applied to the data of participants who reported exacerbations (n=45) to examine symptom signatures and clustering during exacerbation and non-exacerbation periods.
 - Two methodologies were combined: principal component analysis (PCA), followed by an unsupervised clustering method, K-means[2]. While PCA aims to find a low-dimensional representation of the observed data, unsupervised clustering methods such as K-means are focused on finding homogeneous subgroups among the observations.
- Four symptom clusters were identified (**Figure 4A**). A feature analysis was performed to evaluate what combination of symptoms distinguished each unique cluster from the other clusters (**Figure 4B**).

Limitations

- The study may be limited by the representativeness of the study population, and thus the generalizability of the findings to the broader MG patient population.
 - The decentralized format and digital methods are likely to have been selective for participants with a moderate-to-high level of digital literacy.
 - While the study population was appropriately diverse with regards to age, gender and geographic distribution, the ethnic diversity of participants was limited.
- The majority of participants who withdrew from the study did so because they felt they were too ill or functionally impaired to continue, which may represent a source of attrition bias.

Figure 1

A

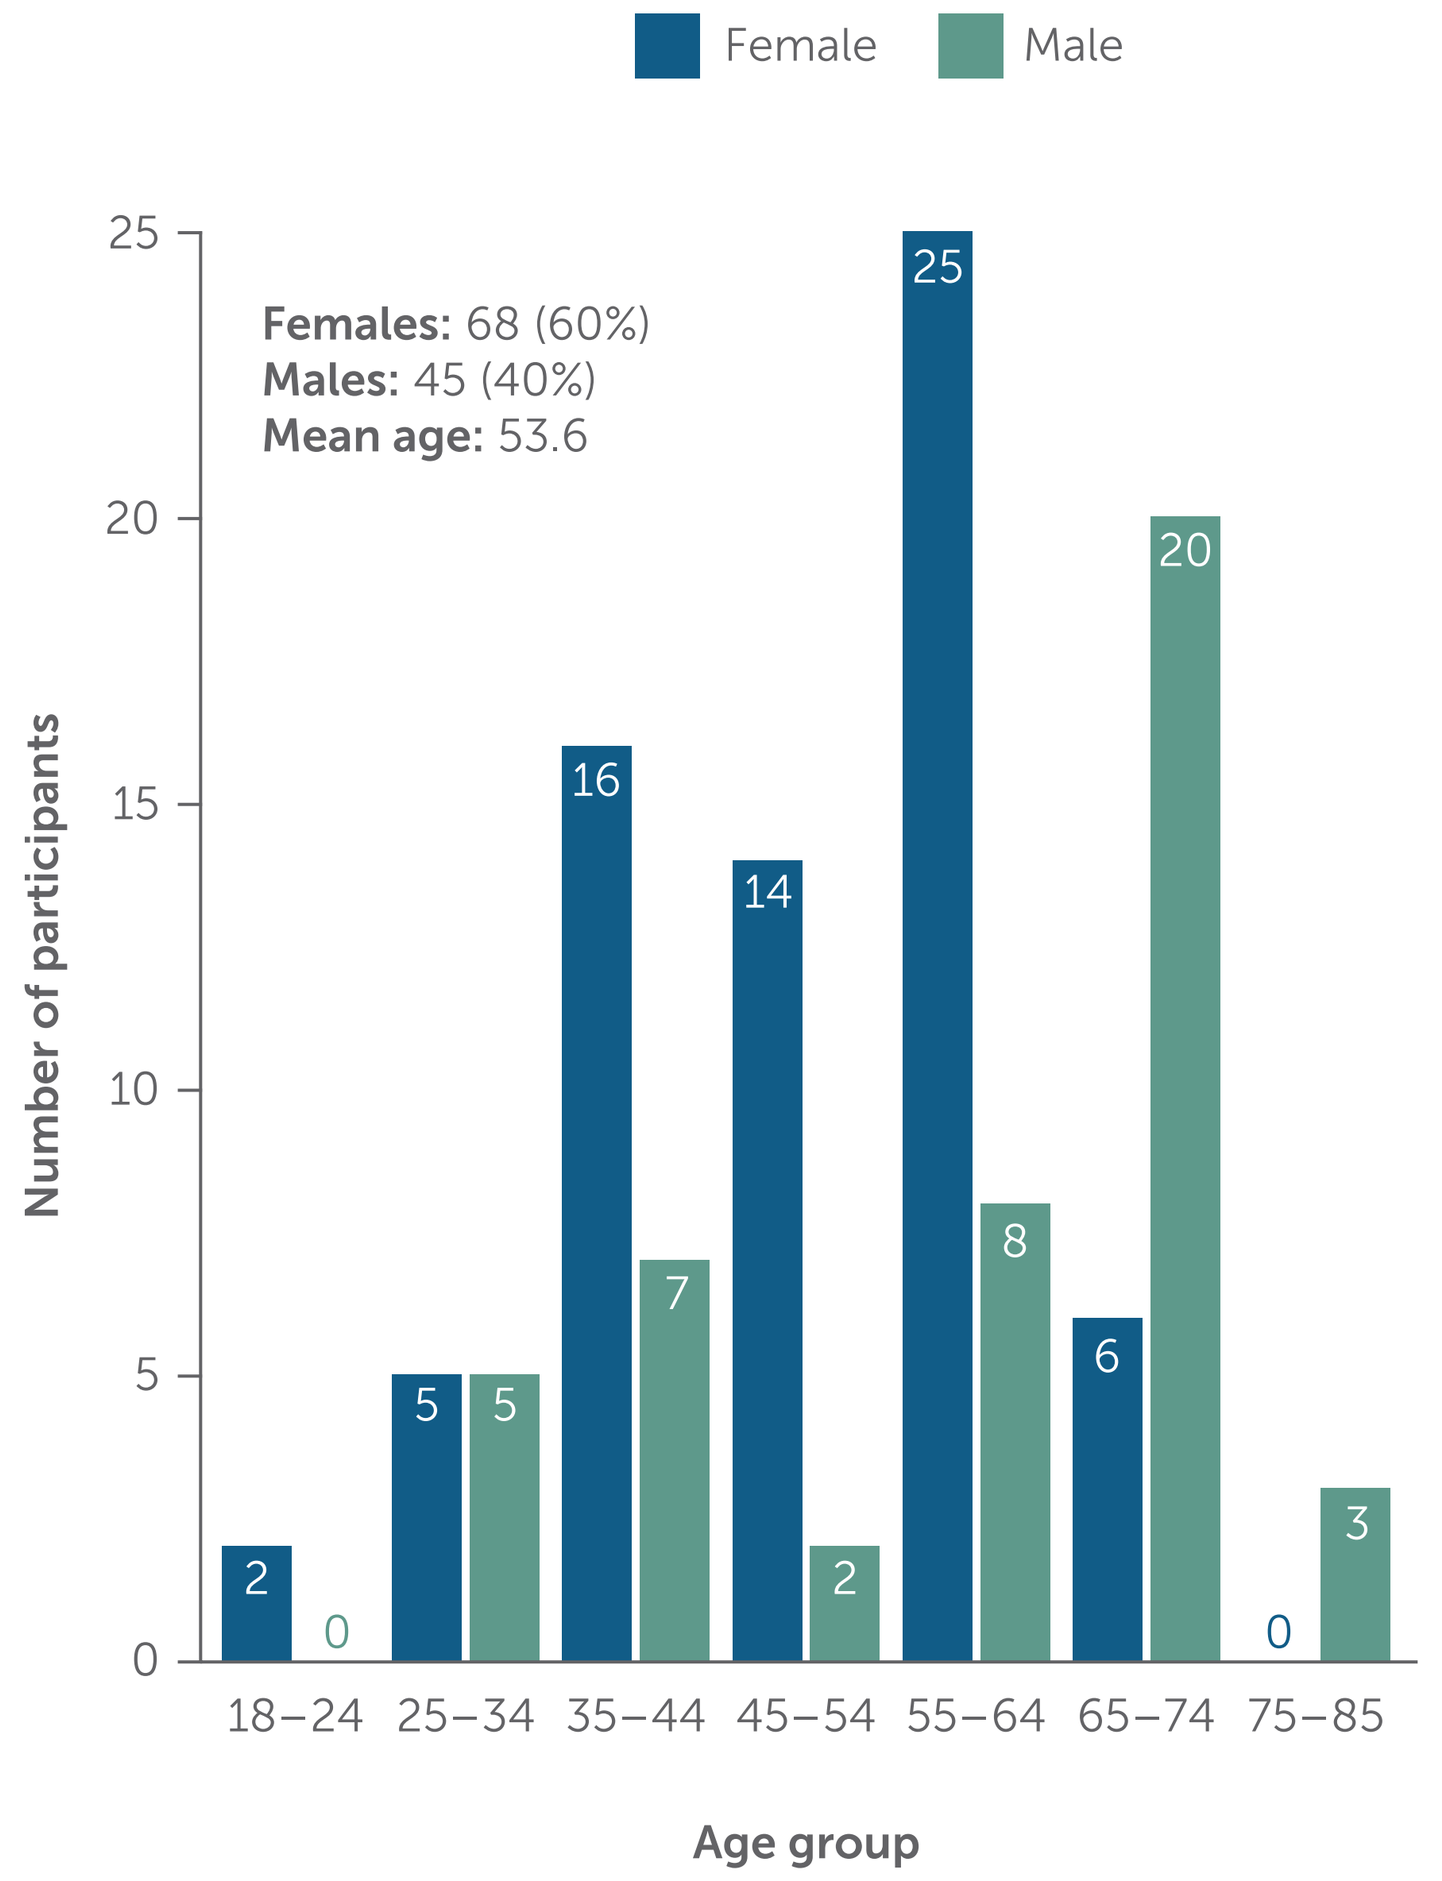
Female
Male
Females: 68 (60%)
Males: 45 (40%)
Mean age: 53.6
Number of participants
18–24 25–34 35–44 45–54 55–64 65–74 75–85
Age group
2 0 5 5 16 7 14 2 25 8 6 20 0 3

B

Race/ethnicity*	Number of participants
White	102
Hispanic, Latin or Spanish origin	3
Black or African American	6
American Indian or Alaska Native	2
Asian	3
Other	2

*Participants may select more than one option.

C

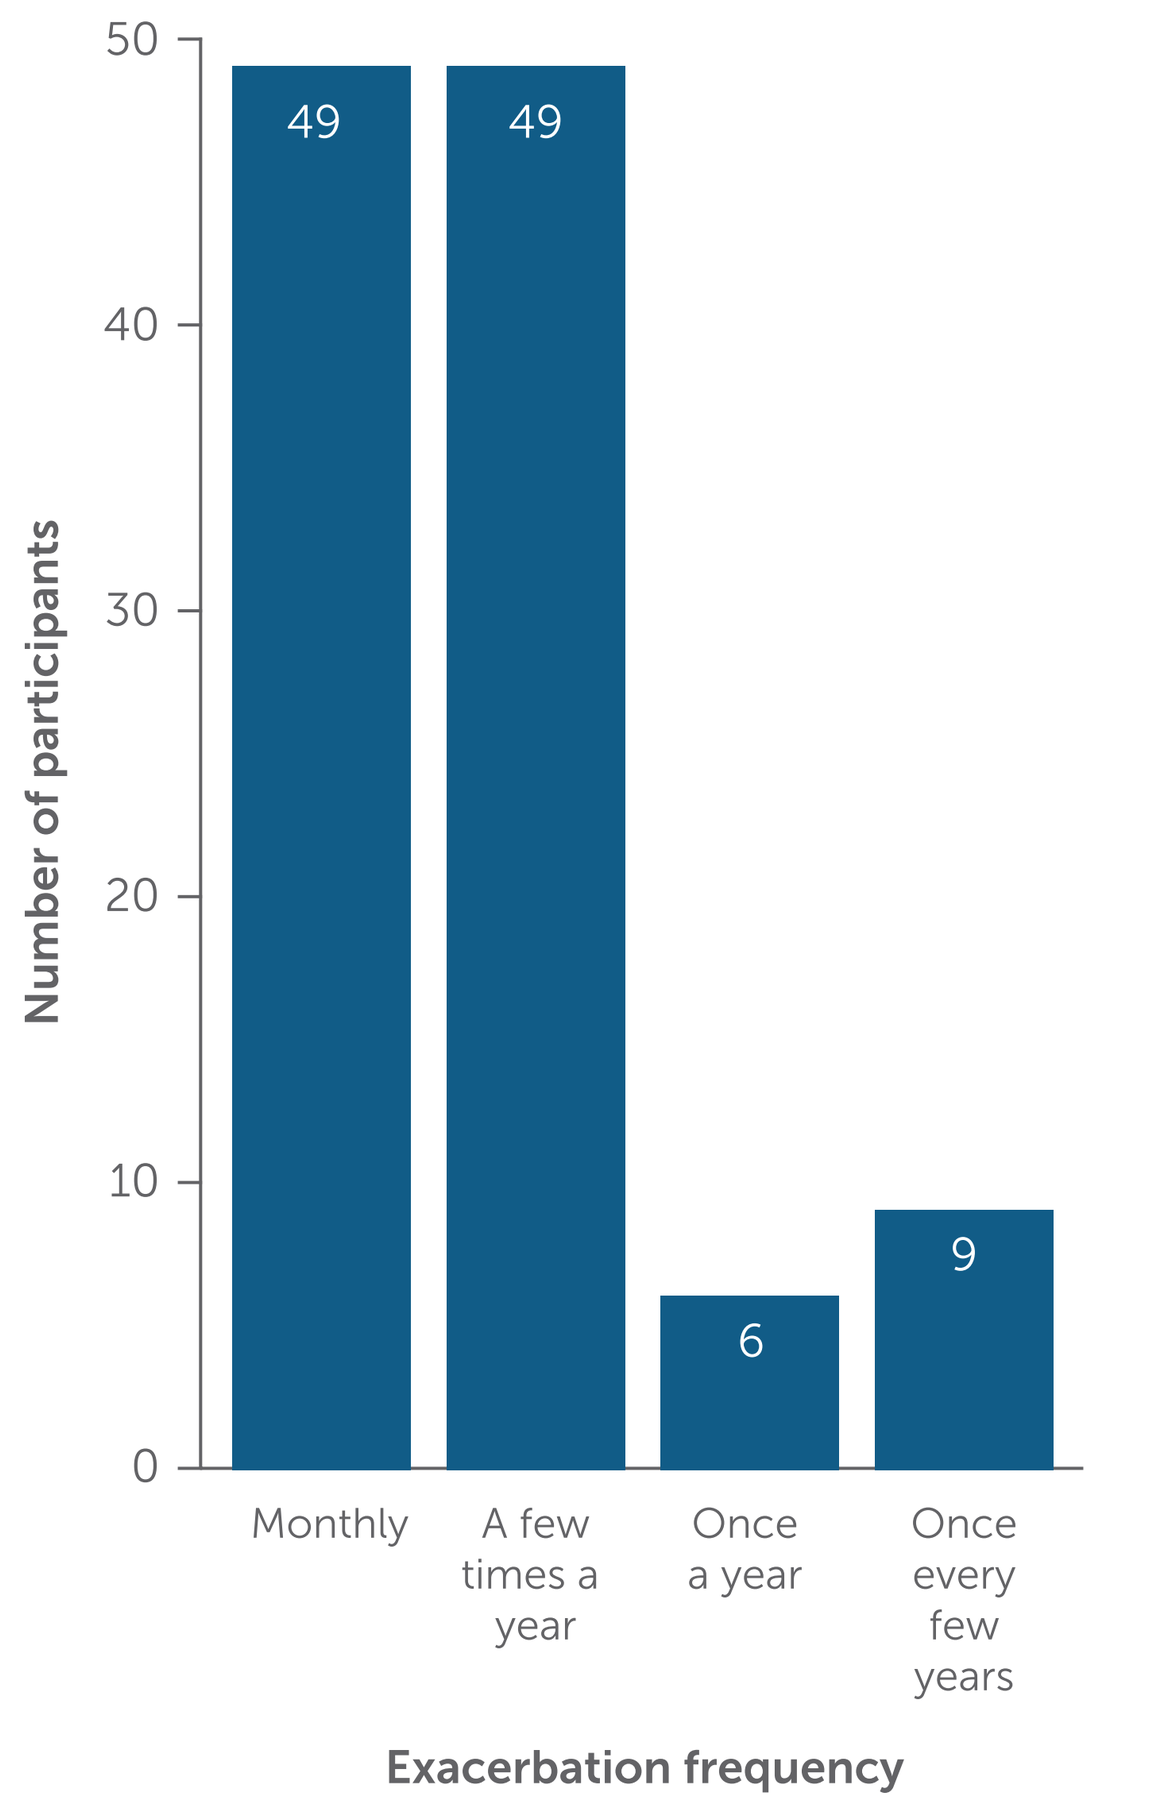
Number of participants
49 49 6 9
Monthly
A few times a year
Once a year
Once every few years
Exacerbation frequency

D

Group 0: No active medications reported
Group 1: Pyridostigmine
Group 2: Pyridostigmine + glucocorticoid
Group 3: Steroid sparing, chronic immunosuppression (azathioprine, mycophenolate mofetil, tacrolimus, cyclosporine, methotrexate)
Group 4: Treatment for refractory disease (eculizumab, rituximab, cyclophosphamide, maintenance IVIG, maintenance plasmapheresis)

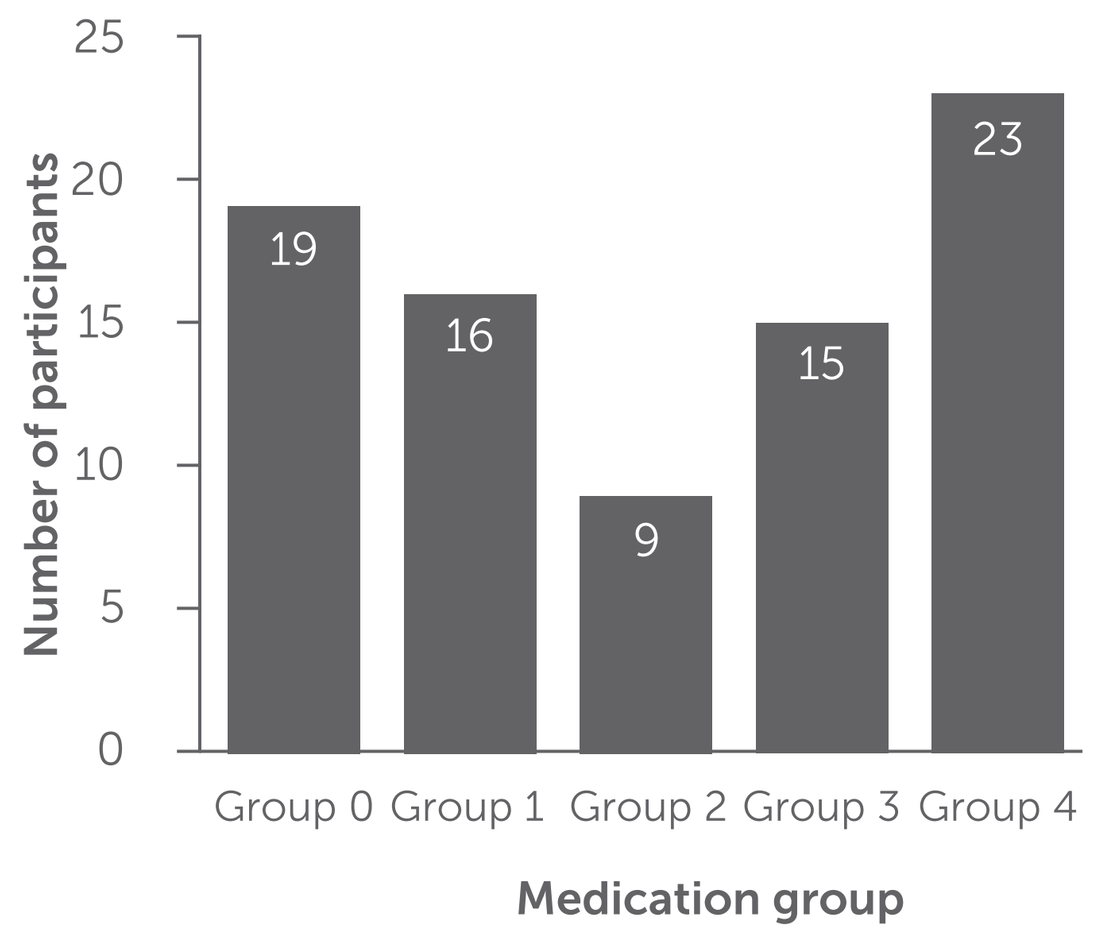
Number of participants
19 16 9 15 23
Group 0 Group 1 Group 2 Group 3 Group 4
Medication group

Figure 2

A

Self-reported MG-ADL scores in daily check-ins: Exacerbation vs. non-exacerbation days (n=82)

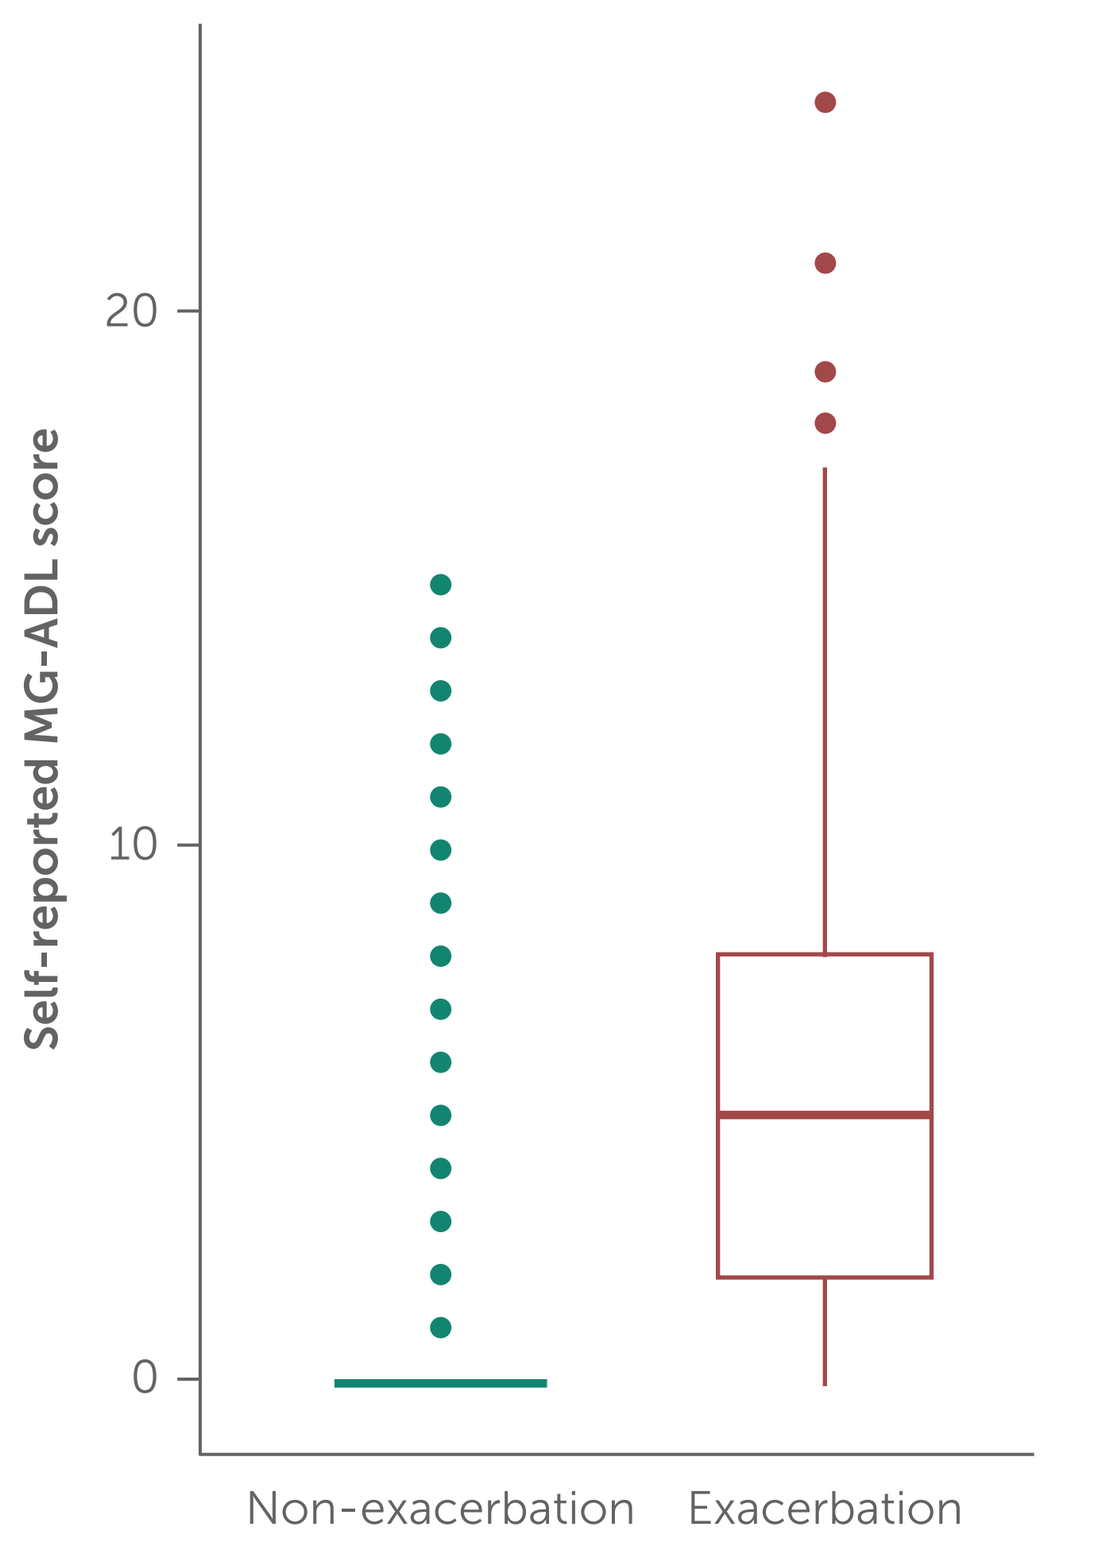
Self-reported MG-ADL score
Non-exacerbation
Exacerbation

B

Average MG-ADL scores for participants with self-reported exacerbations (n=45)

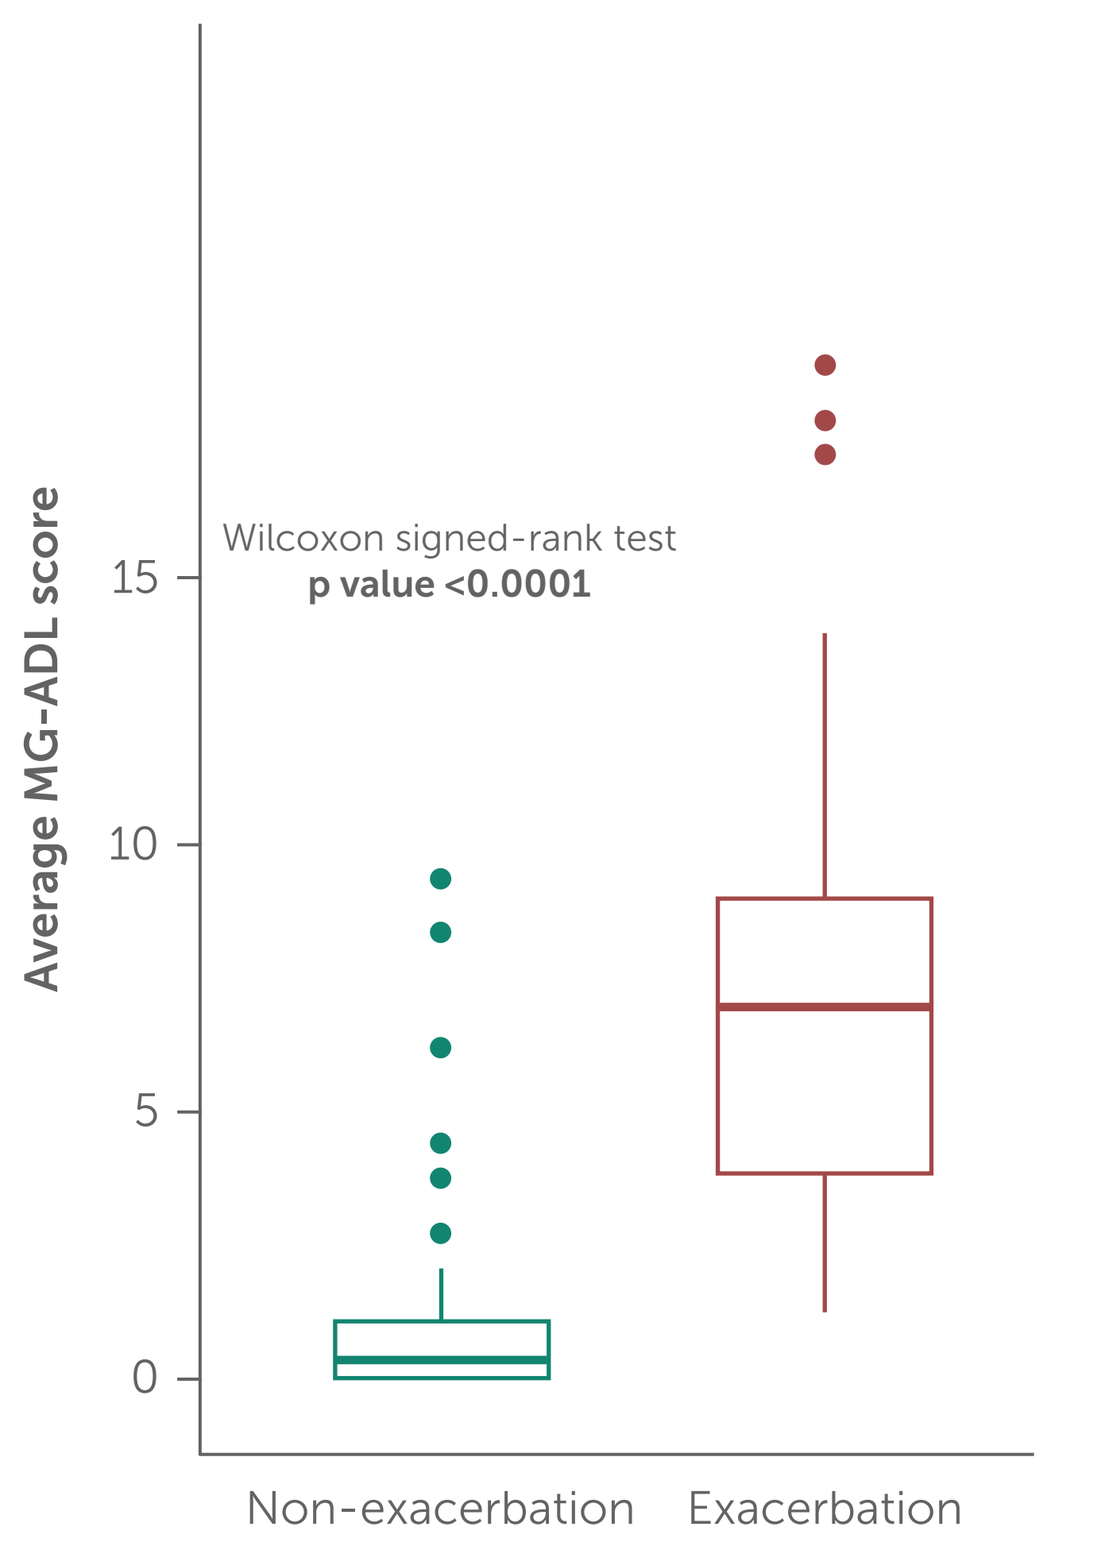
Average MG-ADL score
Wilcoxon signed-rank test
p value <0.0001
Non-exacerbation
Exacerbation

Figure 3

A

Daily step counts vs. self-reported MG-ADL score:
All participants with daily step counts (n=29)

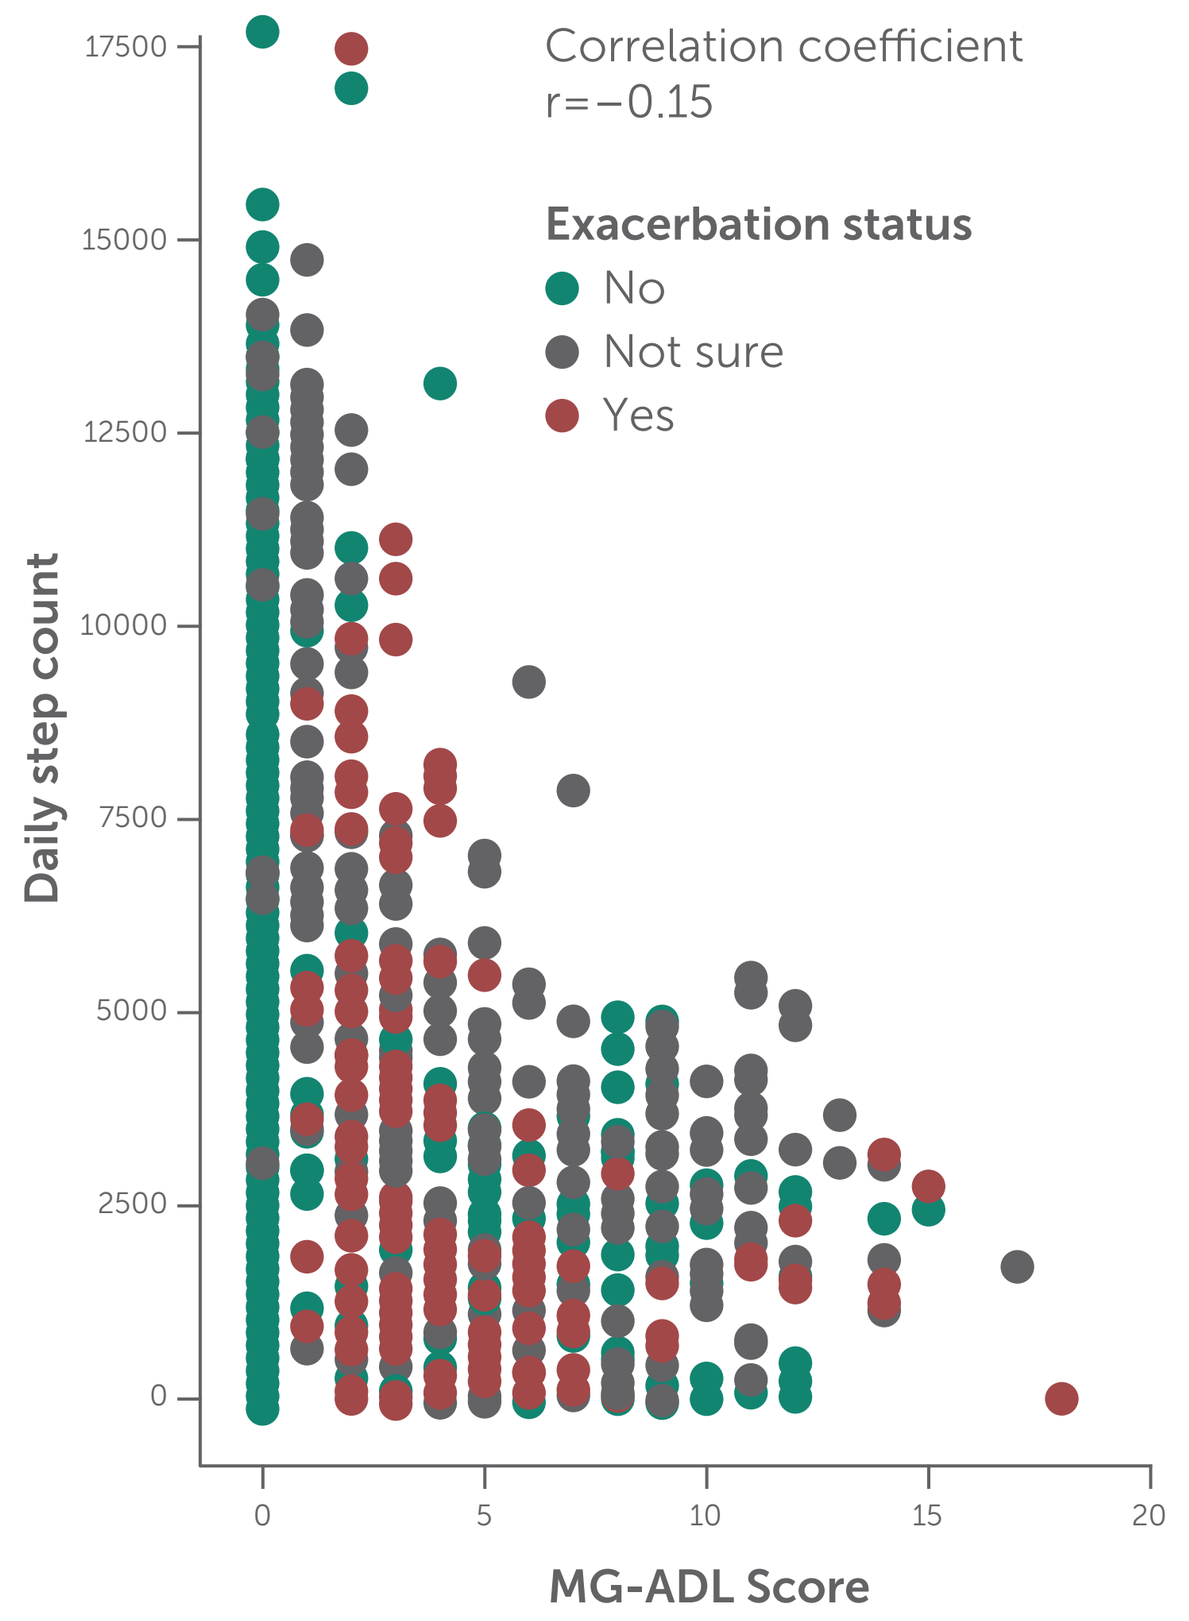
Correlation coefficient
r=−0.15
Exacerbation status
No
Not sure
Yes
Daily step count
MG-ADL Score

B

Variance in participant-generated daily step counts:
Participants with exacerbations (n=14)

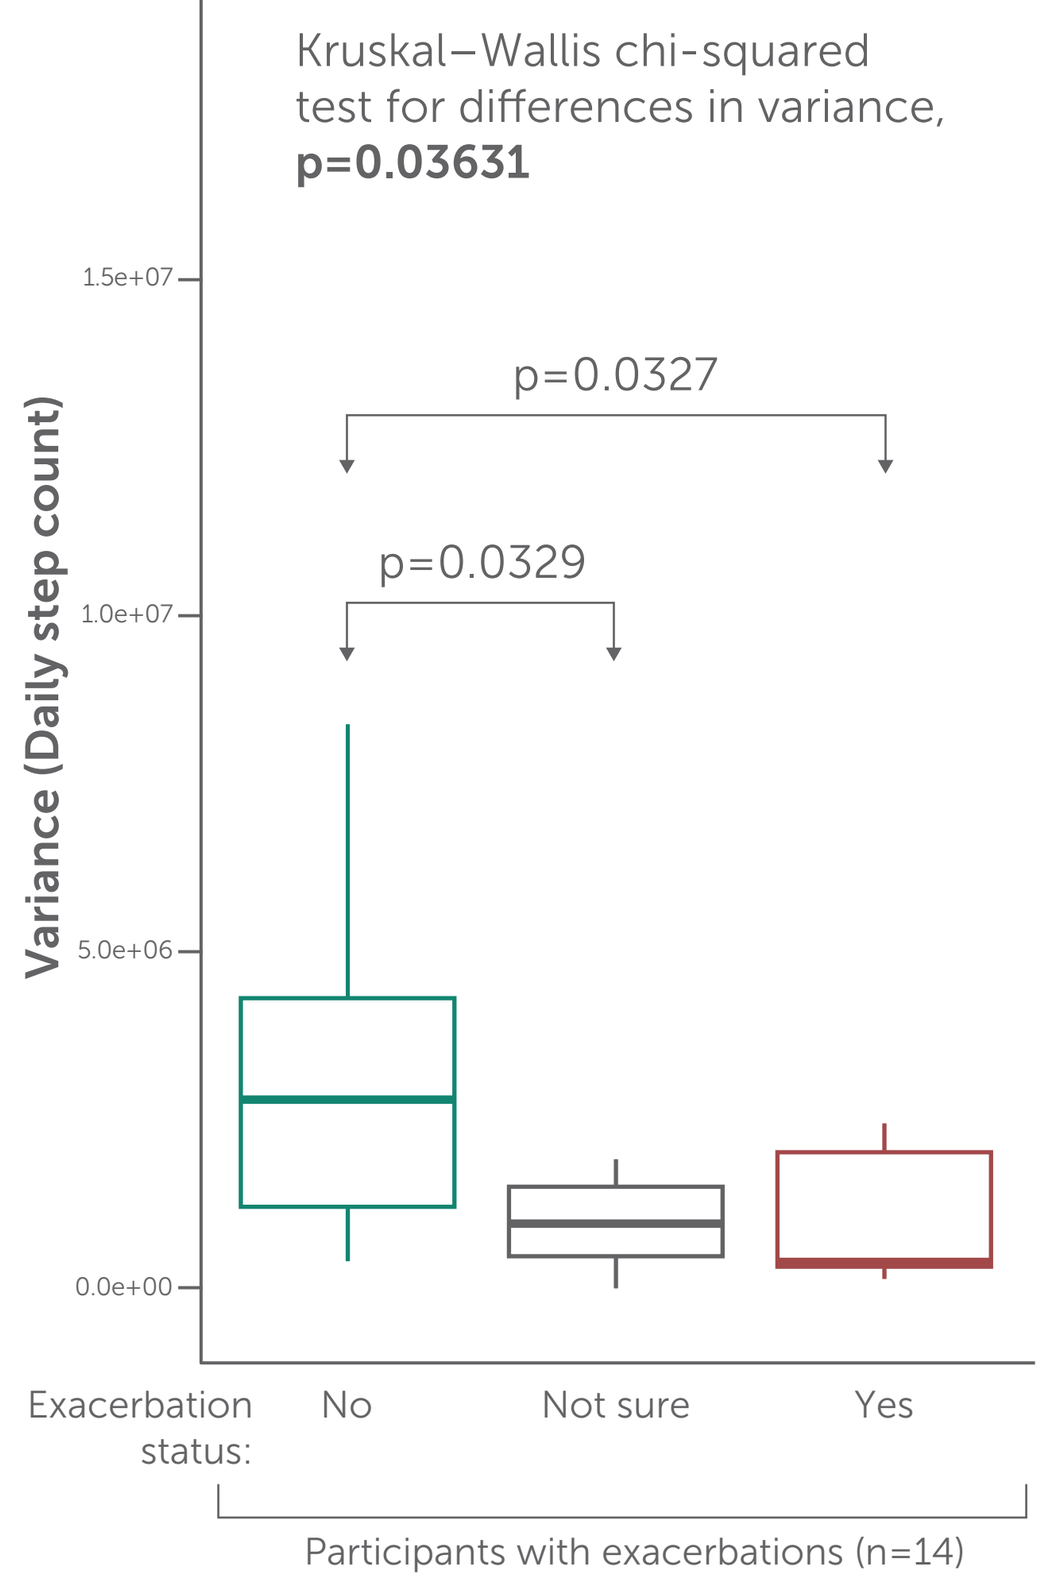
Kruskal–Wallis chi-squared test for differences in variance,
p=0.03631
p=0.0327
p=0.0329
Variance (Daily step count)
Exacerbation status:
No
Not sure
Yes
Participants with exacerbations (n=14)

Conclusions

- This study demonstrated that decentralized, smartphone-based methods to collect real-world data from patients with MG are feasible and may provide enhanced visibility into the significant burden of this disease, which may guide clinical management and future therapeutic development.
- Self-reported MG-ADL scores and passively generated daily step counts may be useful features to differentiate between exacerbation and non-exacerbation periods in MG, and to predict oncoming exacerbations using participant-generated data streams.
- Unsupervised machine-learning methods were able to define unique clusters of exacerbation subtypes with specific symptom representation.
 - These preliminary symptom signatures require further validation, but suggest that digital phenotyping, characterized by increased multidimensionality and frequency of the data collection, holds promise for furthering our understanding of clinically significant exacerbations.

Figure 4

A

Symptom clustering by K-means:
Participants with self-reported exacerbations (n=45)

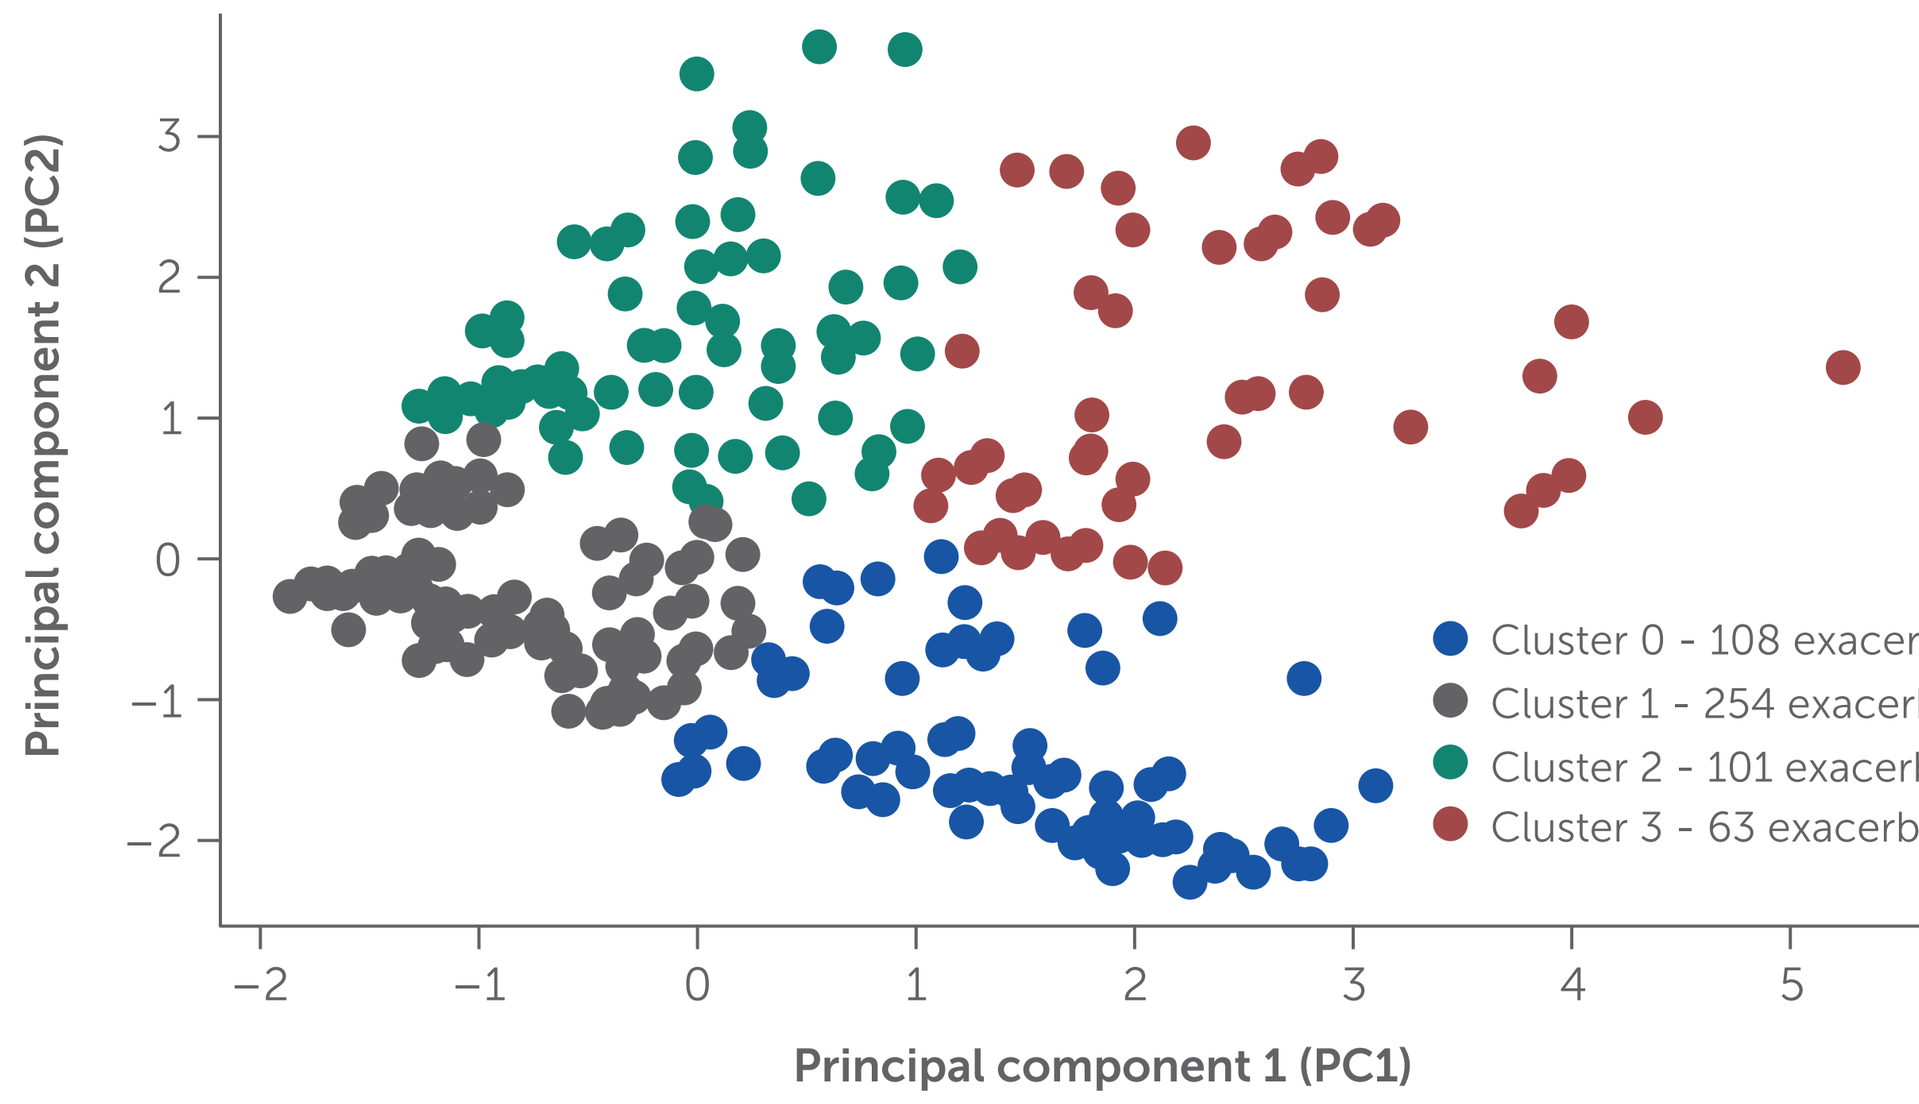
Principal component 2 (PC2)
Principal component 1 (PC1)
Cluster 0 - 108 exacerbations
Cluster 1 - 254 exacerbations
Cluster 2 - 101 exacerbations
Cluster 3 - 63 exacerbations

The clustering represents a total of 526 exacerbation days reported by 45 participants.

B

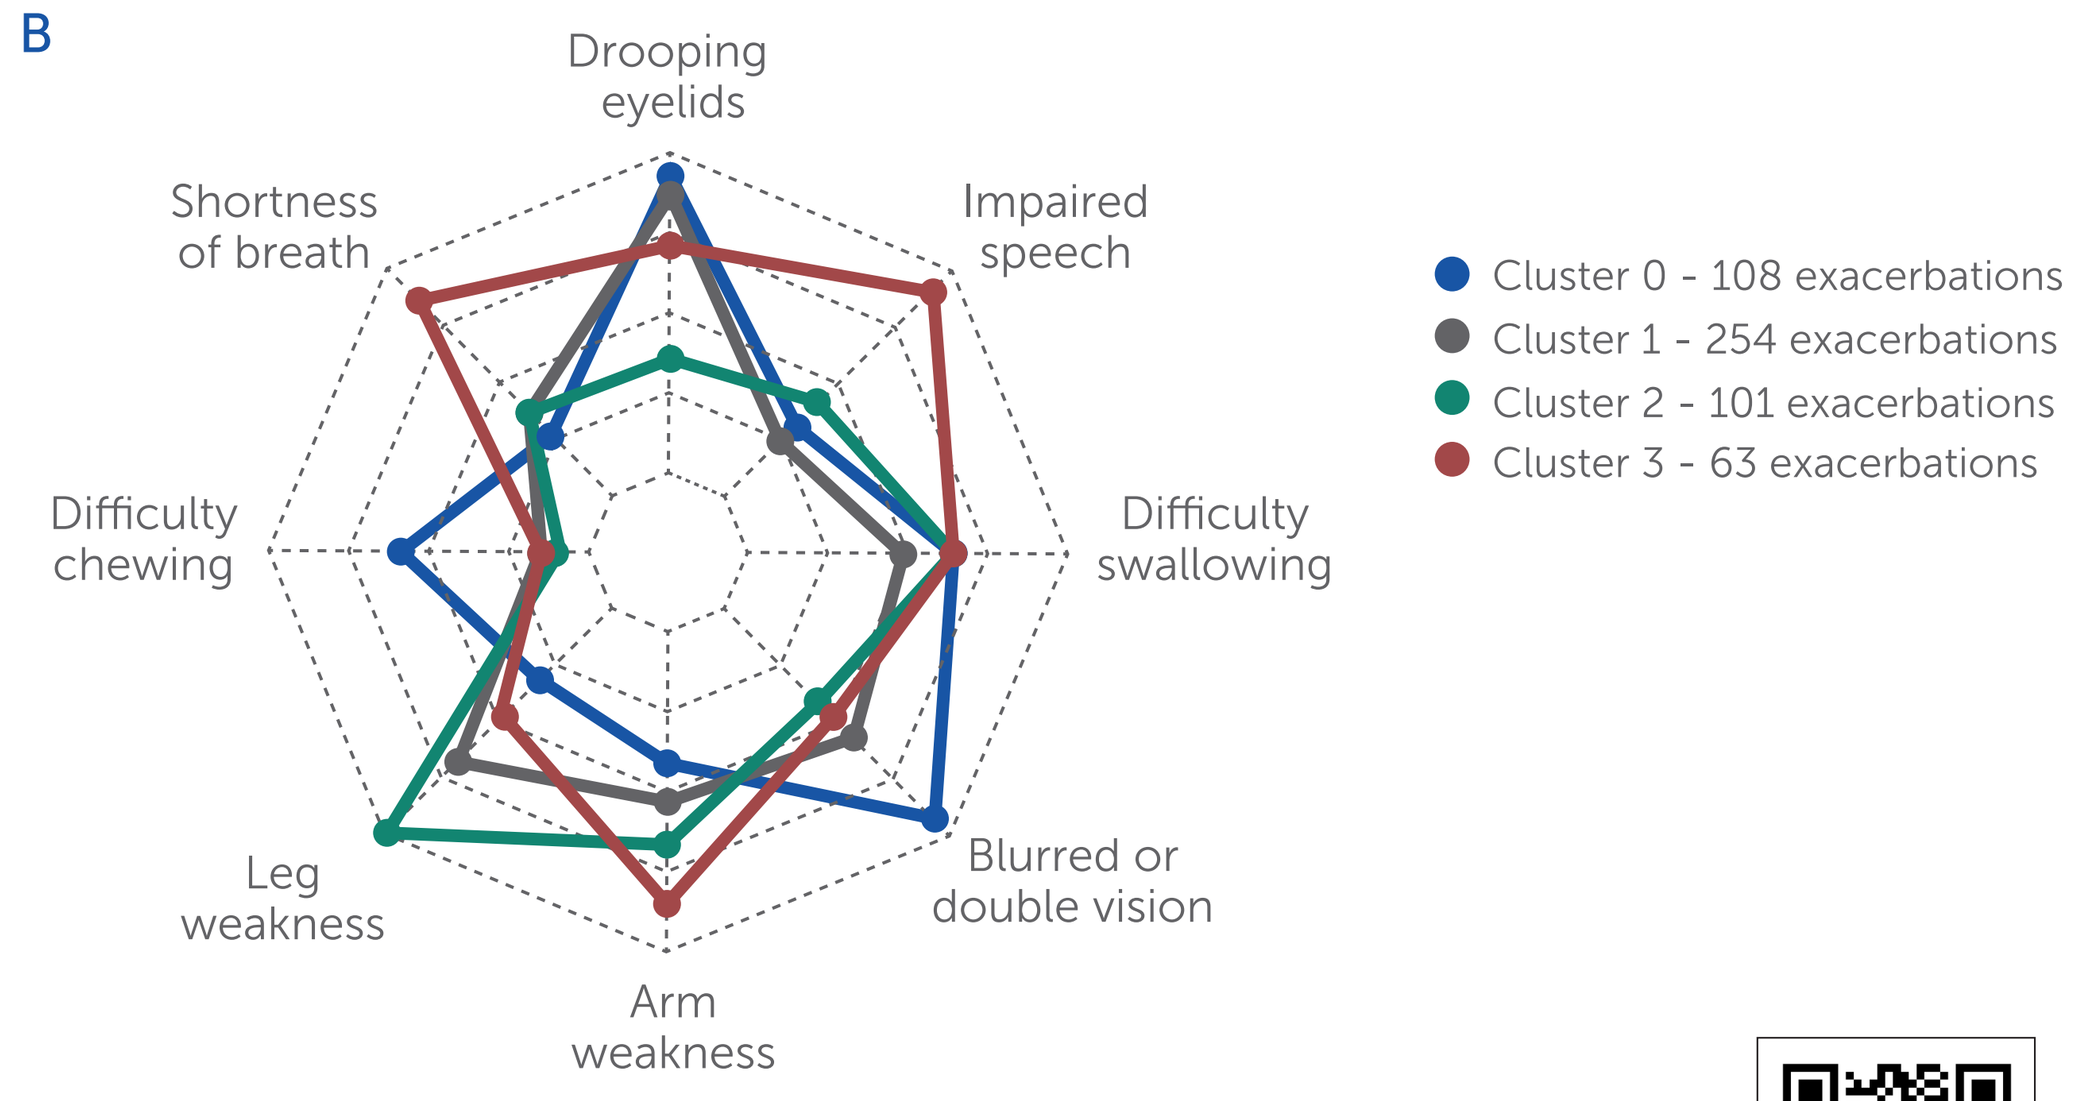
Drooping eyelids
Impaired speech
Difficulty swallowing
Blurred or double vision
Arm weakness
Leg weakness
Difficulty chewing
Shortness of breath
Cluster 0 - 108 exacerbations
Cluster 1 - 254 exacerbations
Cluster 2 - 101 exacerbations
Cluster 3 - 63 exacerbations

Abbreviations: MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; PCA, principal component analysis.

References: 1. Grob D, et al. Lifetime course of myasthenia gravis. Muscle Nerve. 2008;37:141–149. 2. Lopez-Rubio E, et al. Unsupervised learning by cluster quality optimization. Information Sciences. 2018;436:31–55.

Author disclosures: Harriet Dickinson, Emily Kunka and Jean-Christophe Steels are employees and shareholders of UCB Pharma. Sandra Steyaert, Meelis Lootus, Chethan Sarabu, Zeenia Framroze, Nirav R. Shah and Francesca Rinaldo are employees and shareholders of Sharecare Inc. Meelis Lootus is a shareholder of Hammer of the Gods, Inc, neurobotX LTD and ML Technologies LTD.

Acknowledgments: The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study. This study was funded by UCB Pharma in collaboration with Sharecare. The authors acknowledge Veronica Porkess, PhD, of UCB Pharma, Slough, UK, for publication coordination.

Use this QR code to access a copy of this poster